A Multidrug Resistant Engineered CAR T Cell for Allogeneic Combination Immunotherapy

Julien Valton[1], Alexandre Juillerat[1], Anne-Sophie Gautron[2], Philippe Duchateau[2] and Laurent Poirot[2].

[1]Cellectis Inc 430 E 29th street New York, NY 10016, [2]Cellectis SA, 8 rue de la croix Jarry 75013 Paris, France

Abstract

The adoptive transfer of CAR T cell represents a highly promising strategy to fight against multiple cancers. The clinical outcome of such therapies is intimately linked to the ability of effector cells to engraft, proliferate and specifically kill tumor cells within patients. When allogeneic CAR T cell infusion is considered, host versus graft and graft versus host reactions must be avoided to elicit successful antitumor activity in vivo. We propose to address these requirements through the development of multidrug resistant TCRαβ-deficient CAR T cells (CAR T cells KO D/T). We demonstrate that CAR T cells KO D/T are fully TCR deficient, display efficient antitumor activity and proliferate in the presence of purine nucleotide analogues (PNA), currently used in clinic as preconditioning lymphodepleting regimens. We envision that CAR T cells KO D/T could be generated from third party healthy donors and used in any patients as antitumor allogeneic immunotherapy without generating TCRαβ-dependent GvH reaction. Their drug resistance properties could enable them to resist to simultaneous infusion of lymphodepleting regimens to inhibit the host immune system and control their rate of ablation via HvG reaction.

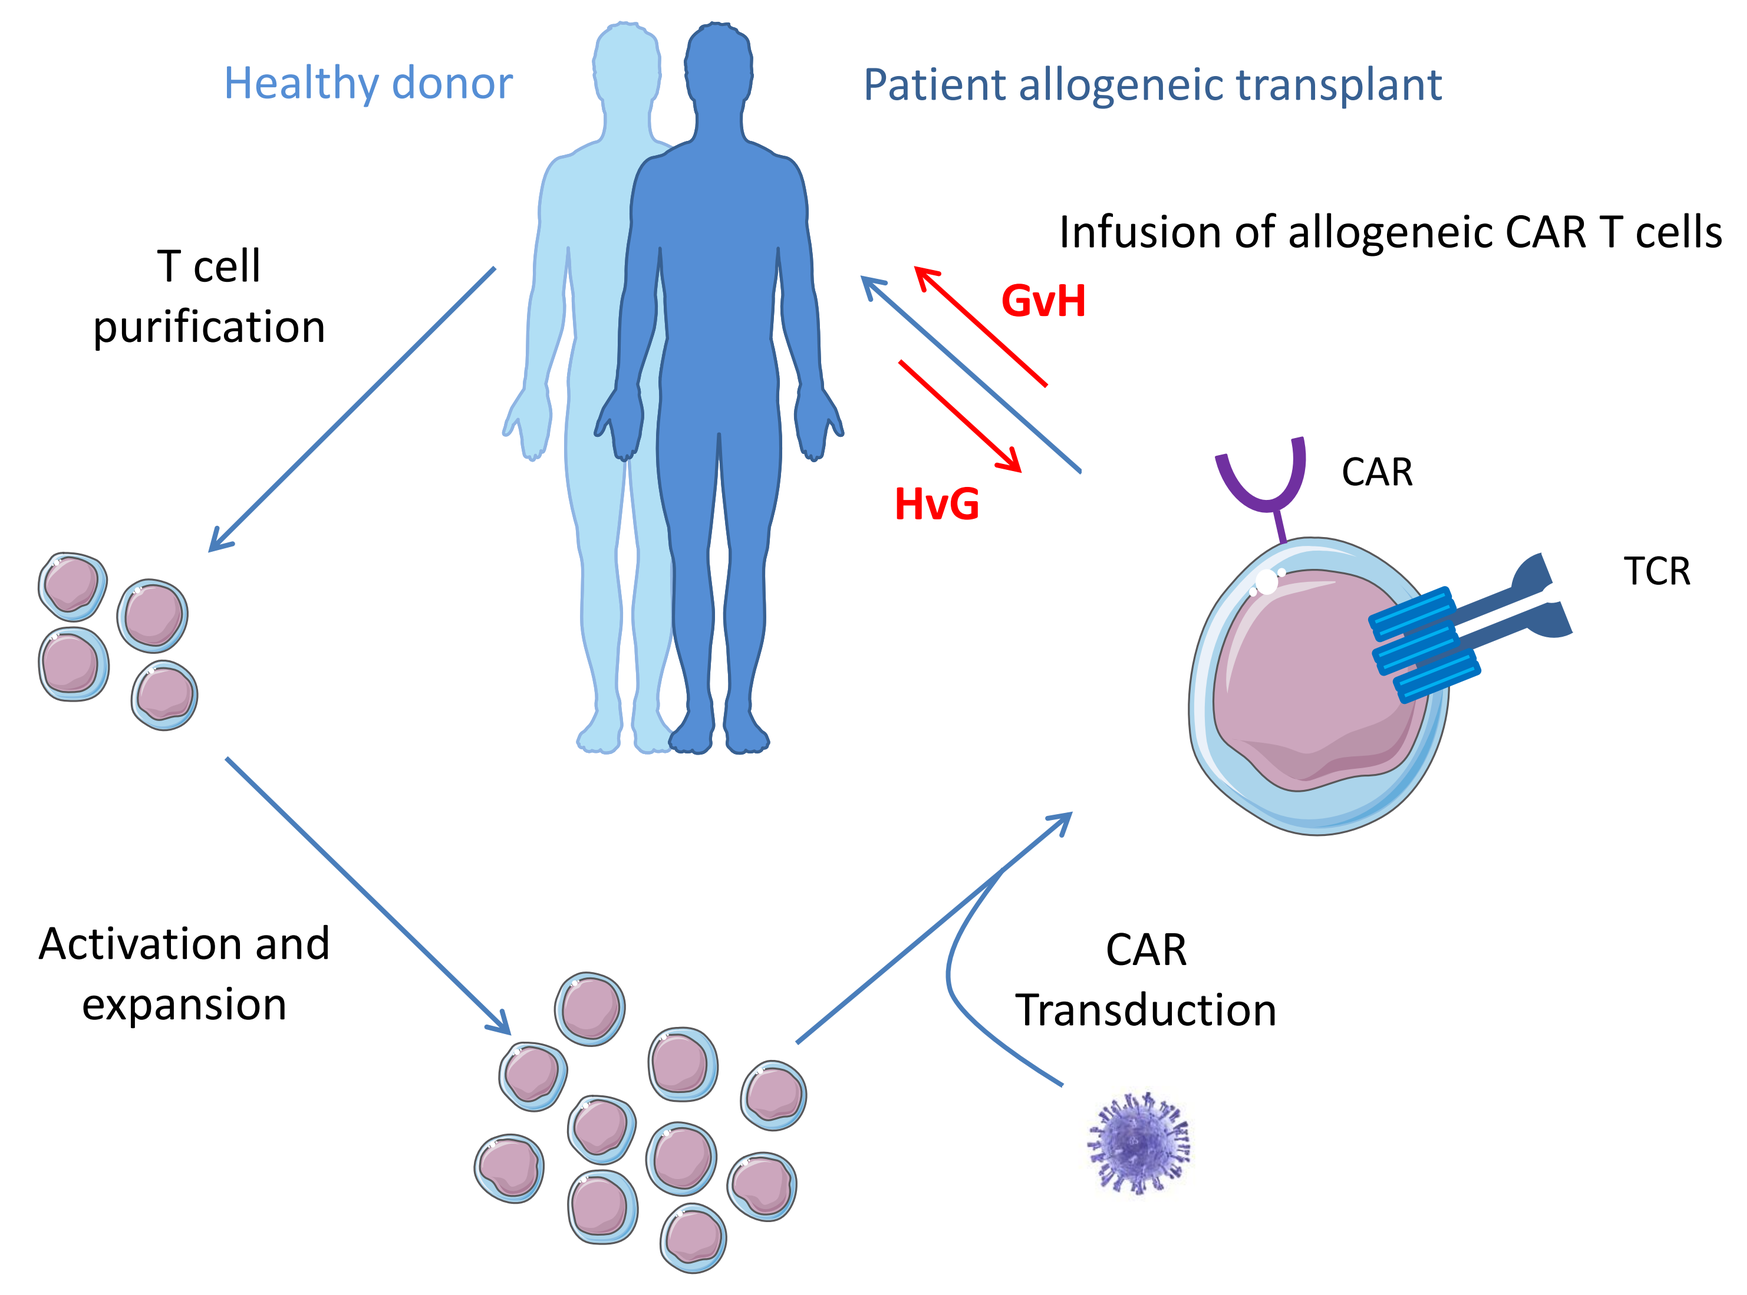
Healthy donor
Patient allogeneic transplant
T cell purification
Infusion of allogeneic CAR T cells
GvH
HvG
CAR
TCR
Activation and expansion
CAR Transduction

Figure 1. Principle of allogeneic CAR T cell immunotherapy illustrating graft versus host (GvH) and host versus graft (HvG) reactions that may occur after allogeneic transfer of CAR T cells

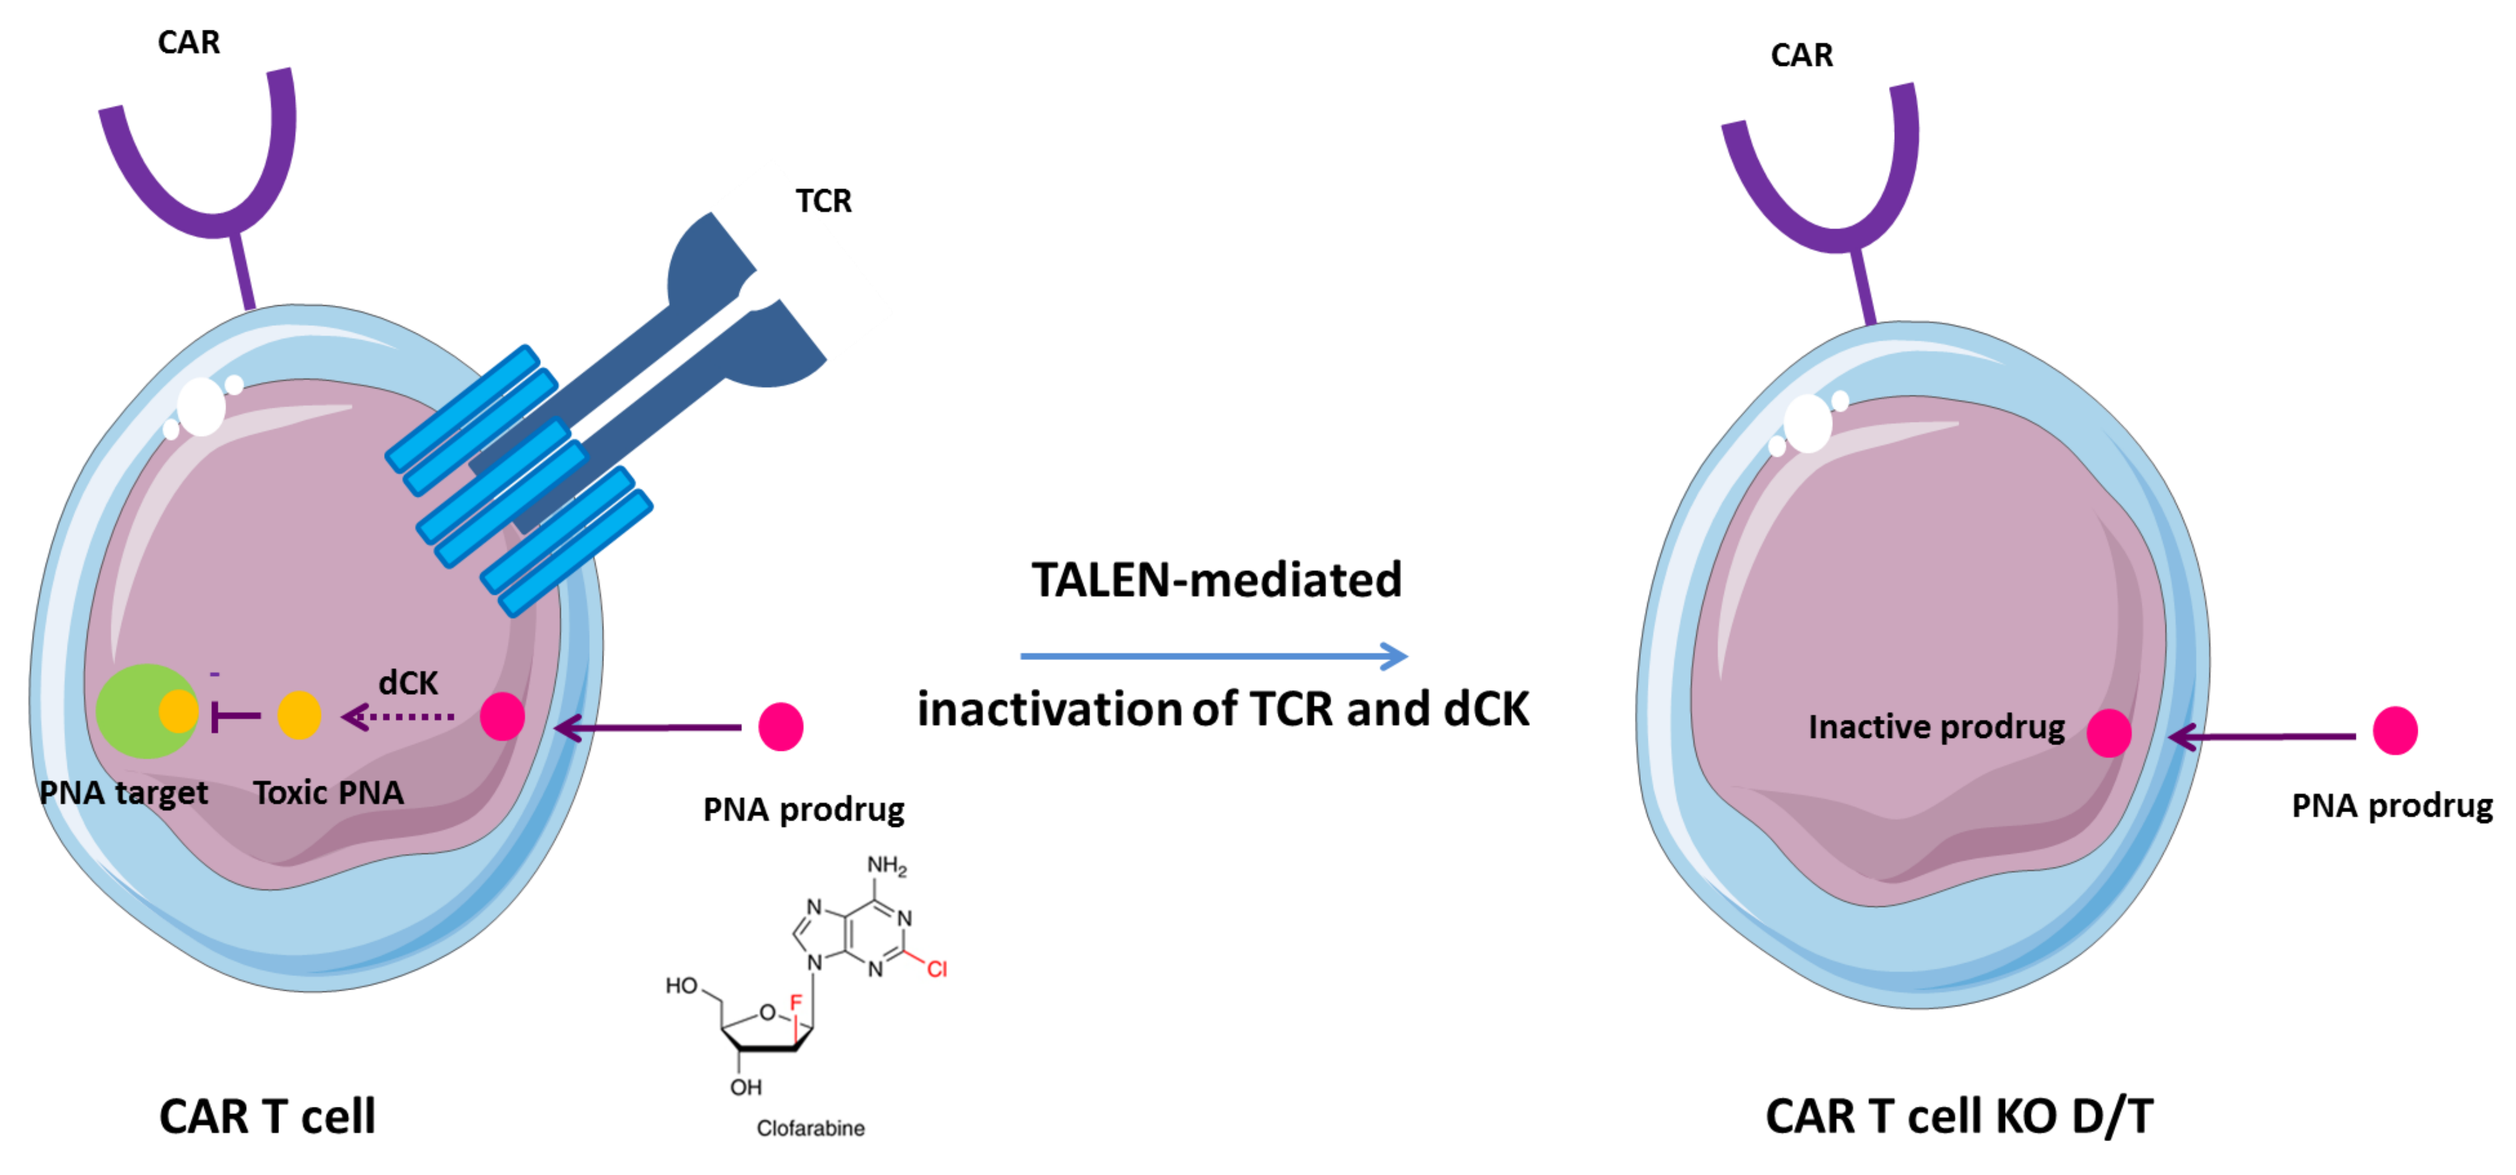
CAR
TCR
dCK
PNA target
Toxic PNA
PNA prodrug
Clofarabine
CAR T cell
TALEN-mediated
inactivation of TCR and dCK
Inactive prodrug
PNA prodrug
CAR T cell KO D/T

Figure 2. Genome engineering strategy to avoid HvG and GvH reactions. This strategy consists in the inactivation of TRAC and dCK (deoxycytidine kinase) genes respectively responsible for TCRαβ surface expression and purine nucleotide analogues (PNA) toxicity. Efficient depletion of TCR at the surface of CAR T cells is expect to prevent them from attacking host tissues via the TCR-dependent GvH reaction. dCK deficiency could enable them to resist to simultaneous infusion of lymphodepleting dose of PNA to inhibit the host immune system and control their rate of ablation via HvG reaction.

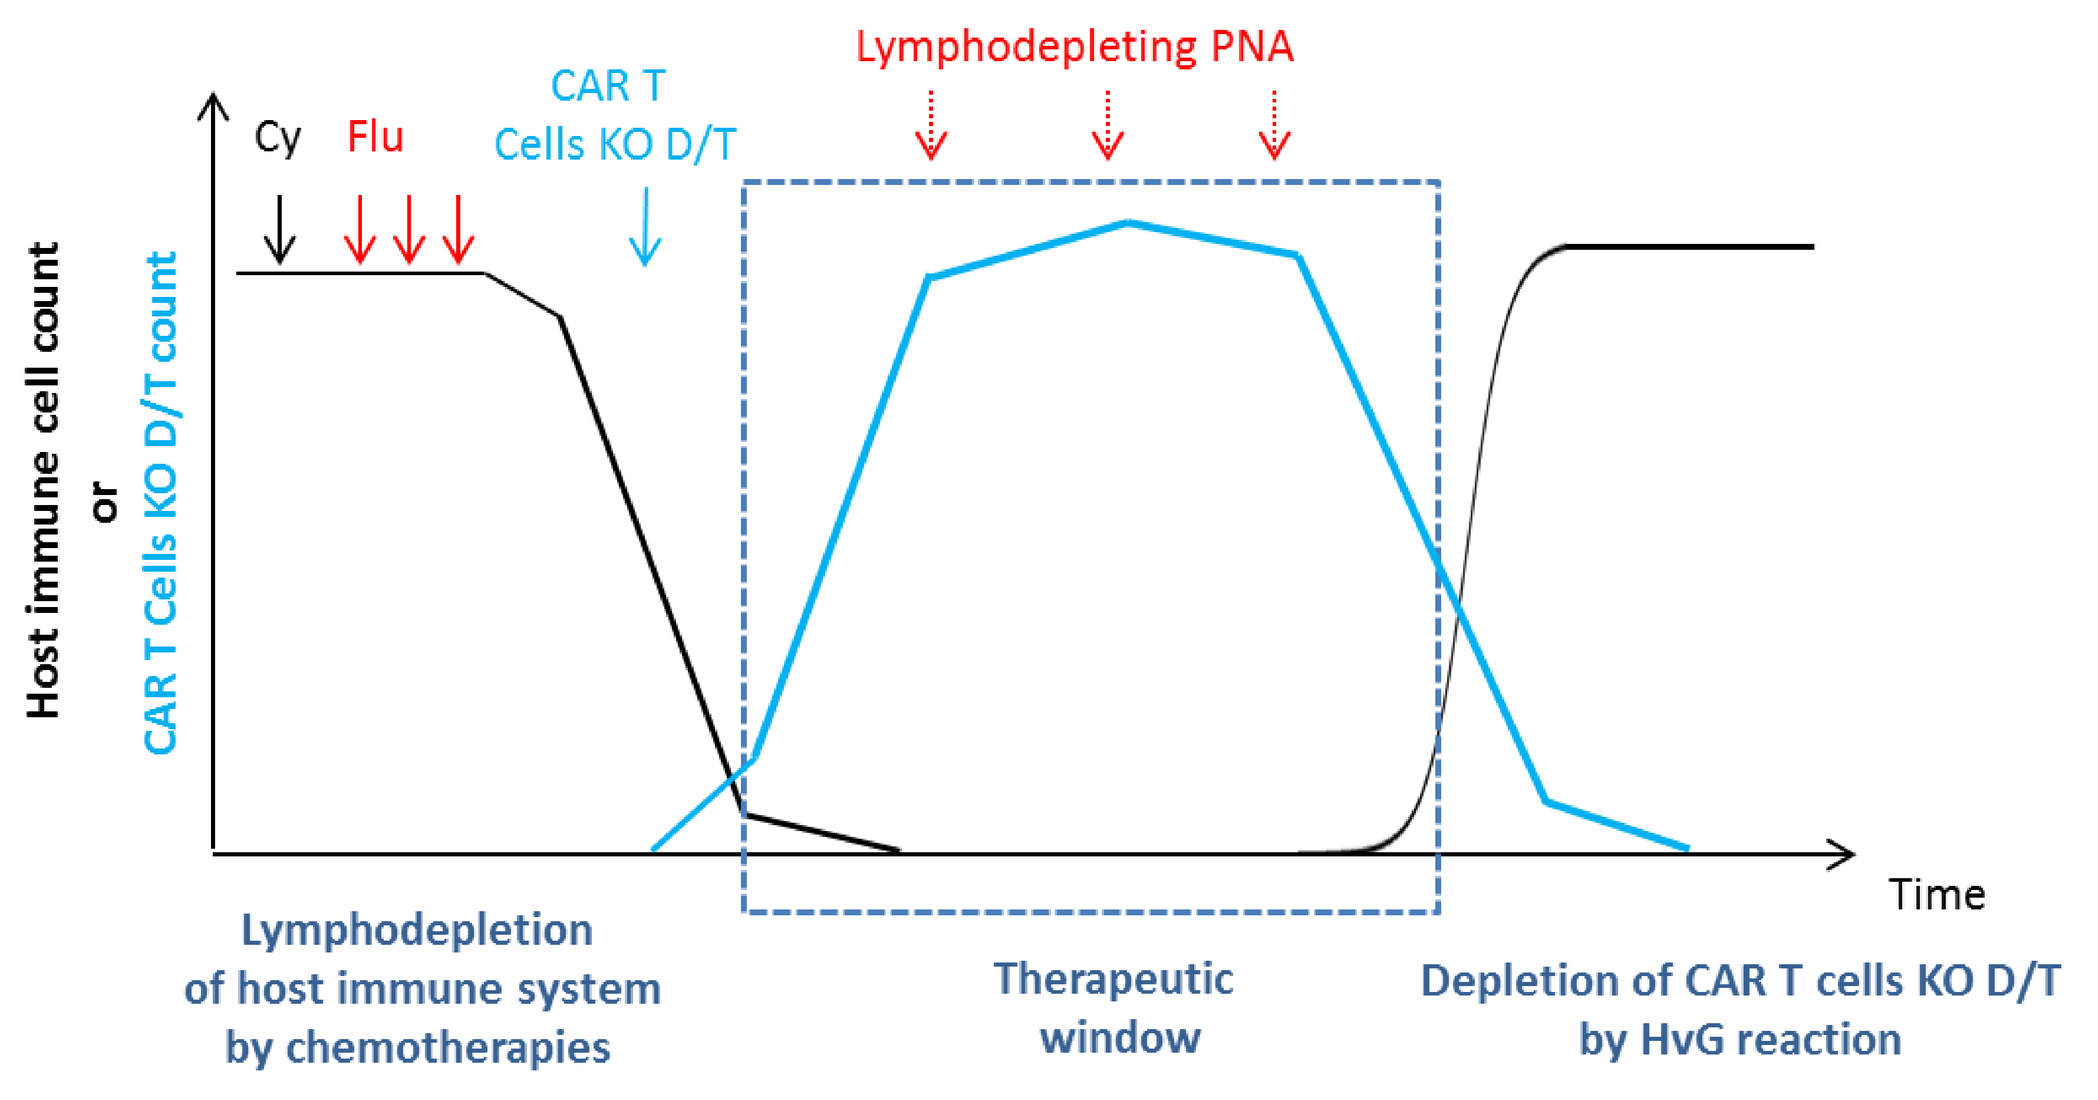
Host immune cell count
or
CAR T Cells KO D/T count
Cy
Flu
CAR T Cells KO D/T
Lymphodepleting PNA
Time
Lymphodepletion of host immune system by chemotherapies
Therapeutic window
Depletion of CAR T cells KO D/T by HvG reaction

Figure 3. Schematic representation of the allogeneic adoptive transfer therapeutic window.

TALEN-mediated TRAC/dCK gene processing is efficient in primary T cells

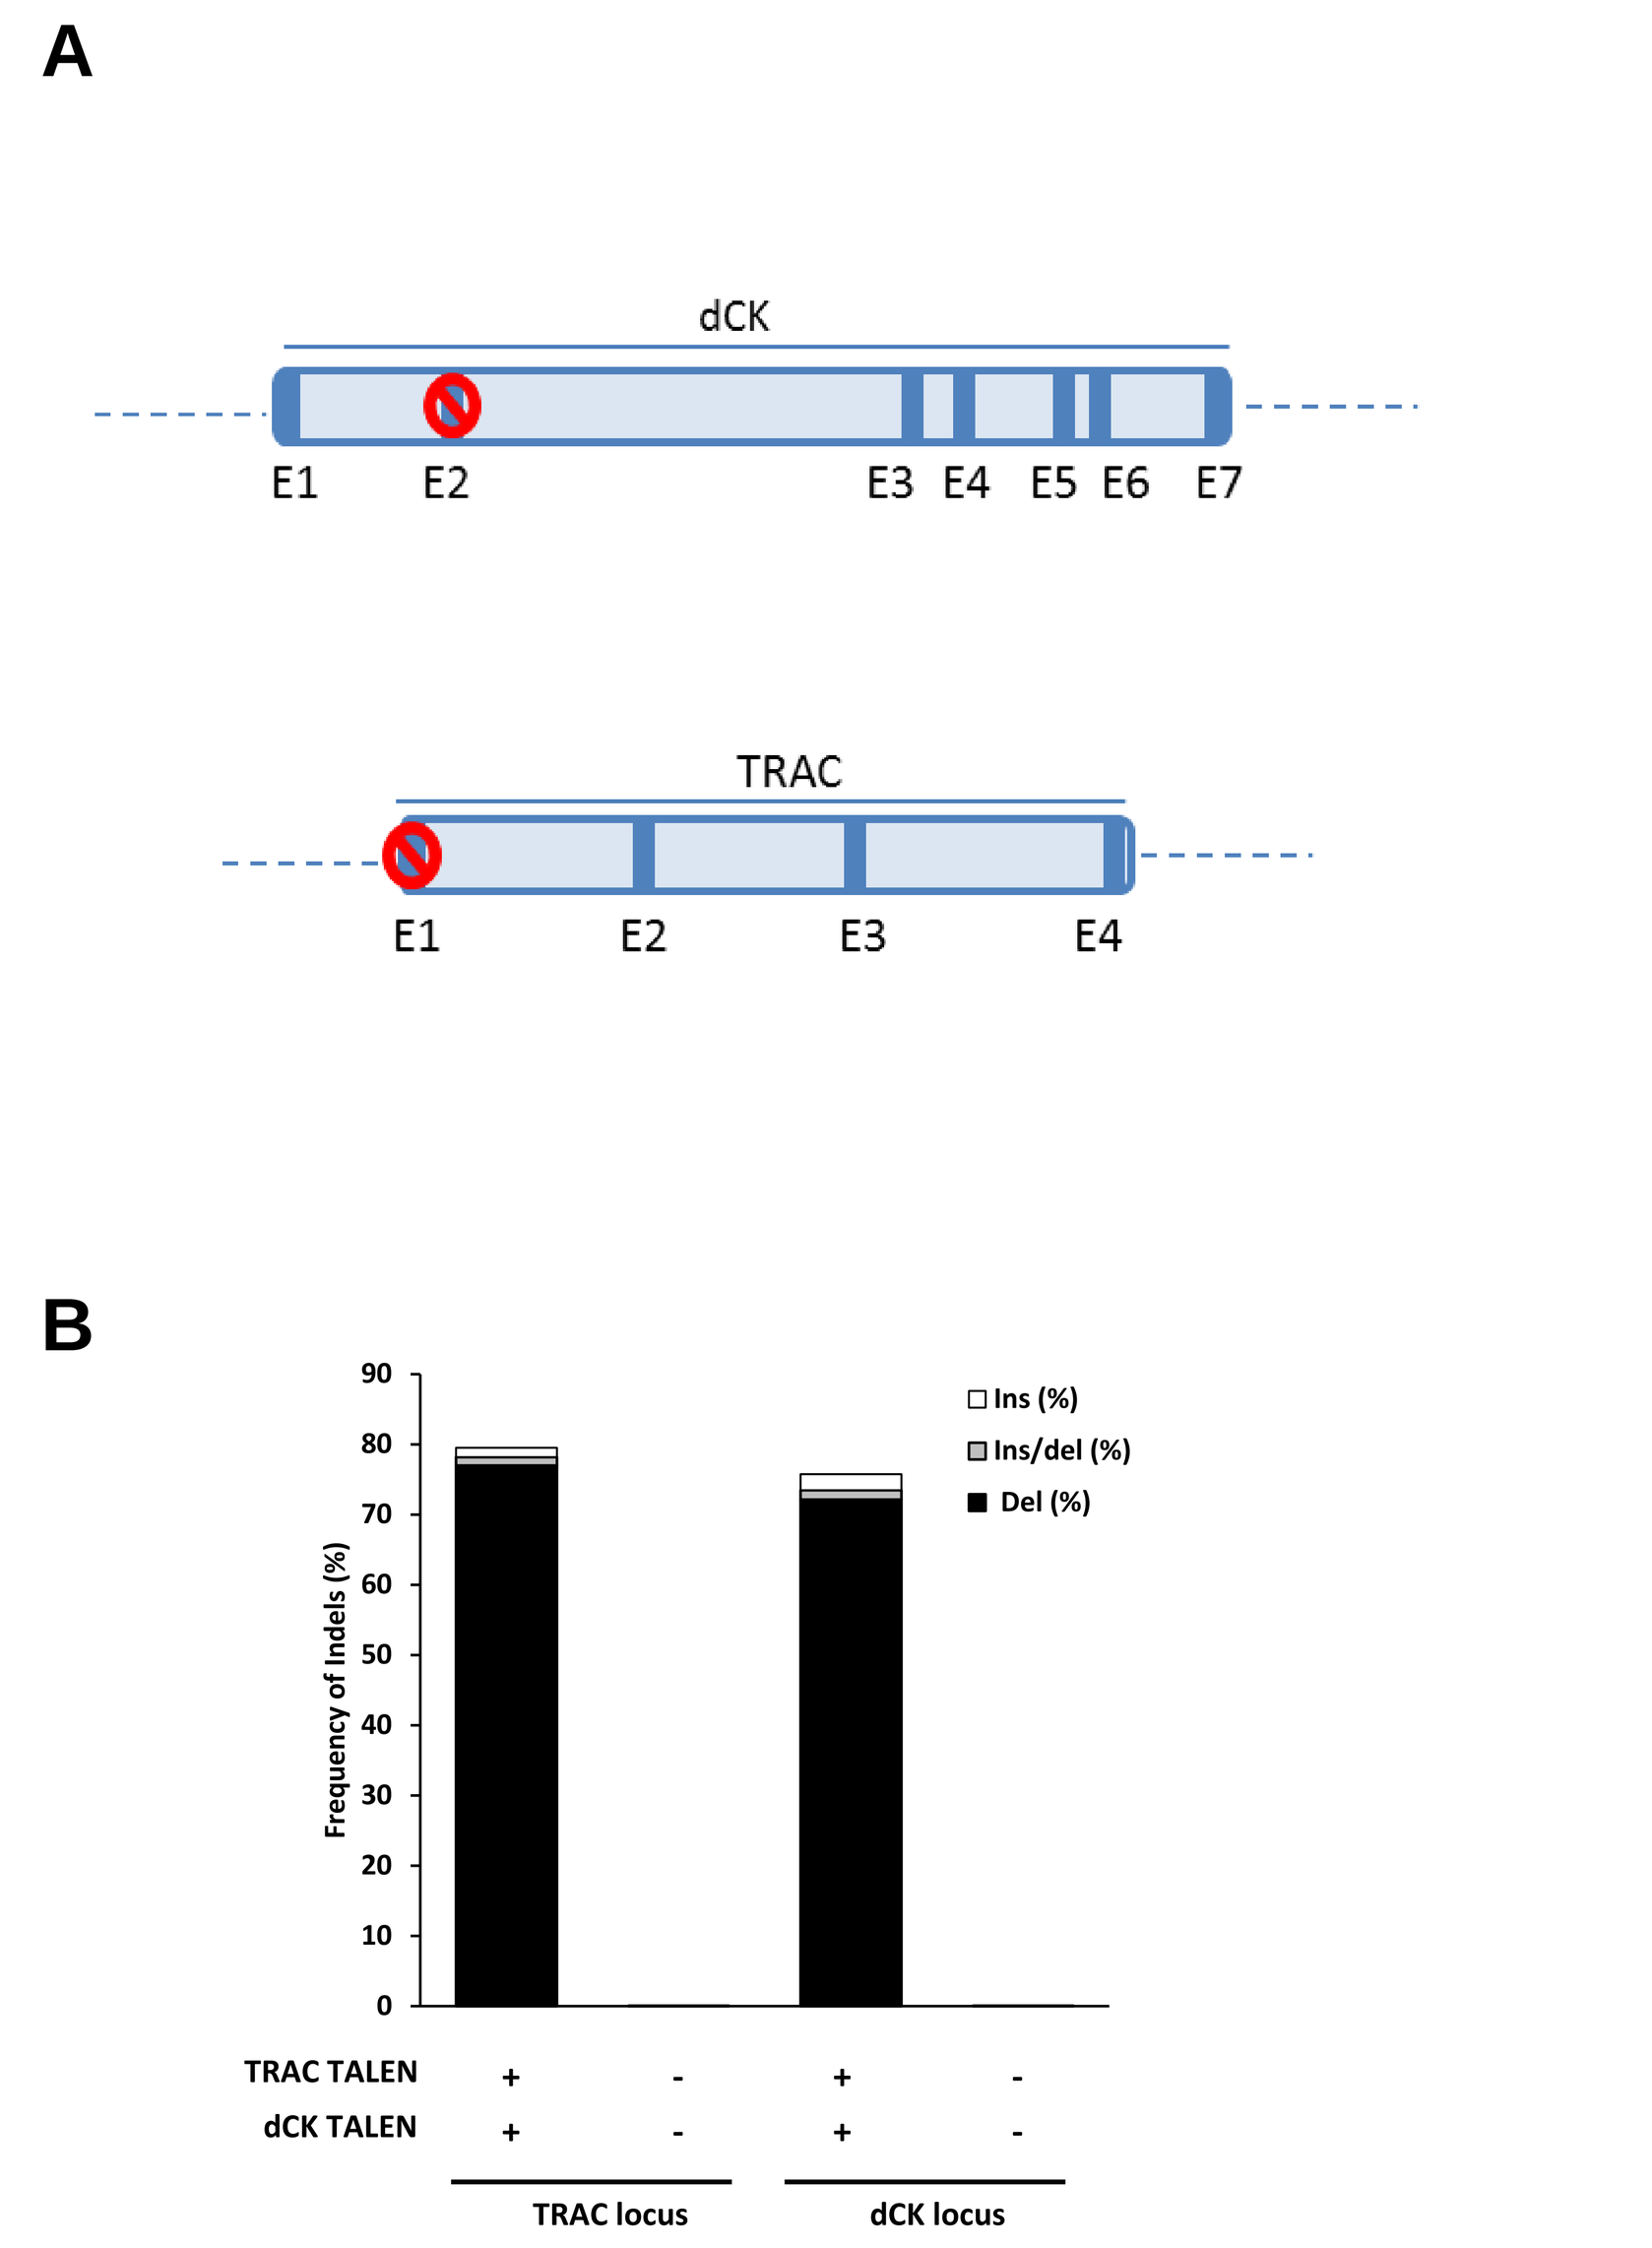
A
dCK
E1 E2 E3 E4 E5 E6 E7
TRAC
E1 E2 E3 E4
B
Frequency of Indels (%)
Ins (%)
Ins/del (%)
Del (%)
TRAC TALEN
dCK TALEN
TRAC locus
dCK locus

Figure 4. TALEN treatment of primary T cells enables simultaneous and highly efficient processing of TRAC and dCK genes. (a) Overall gene architectures of dCK and TRAC open reading frames. Exons (dark blue) and introns (light blue) as well as the location of TALEN-mediated gene processing (red mark) are indicated. **(b)** Genotypic characterization of TALEN-mediated inactivation of dCK and TRAC genes by high throughput DNA sequencing. T cells treated with dCK and TRAC TALEN were allowed to grow for 6 days, their genomic DNA was extracted, amplified using TRAC or dCK specific PCR amplicons and analysed by high throughput DNA sequencing. The frequencies of indels generated at TRAC and dCK locus are indicated.

TALEN-mediated TRAC gene processing disrupts TCRαβ expression at the surface of primary T cells

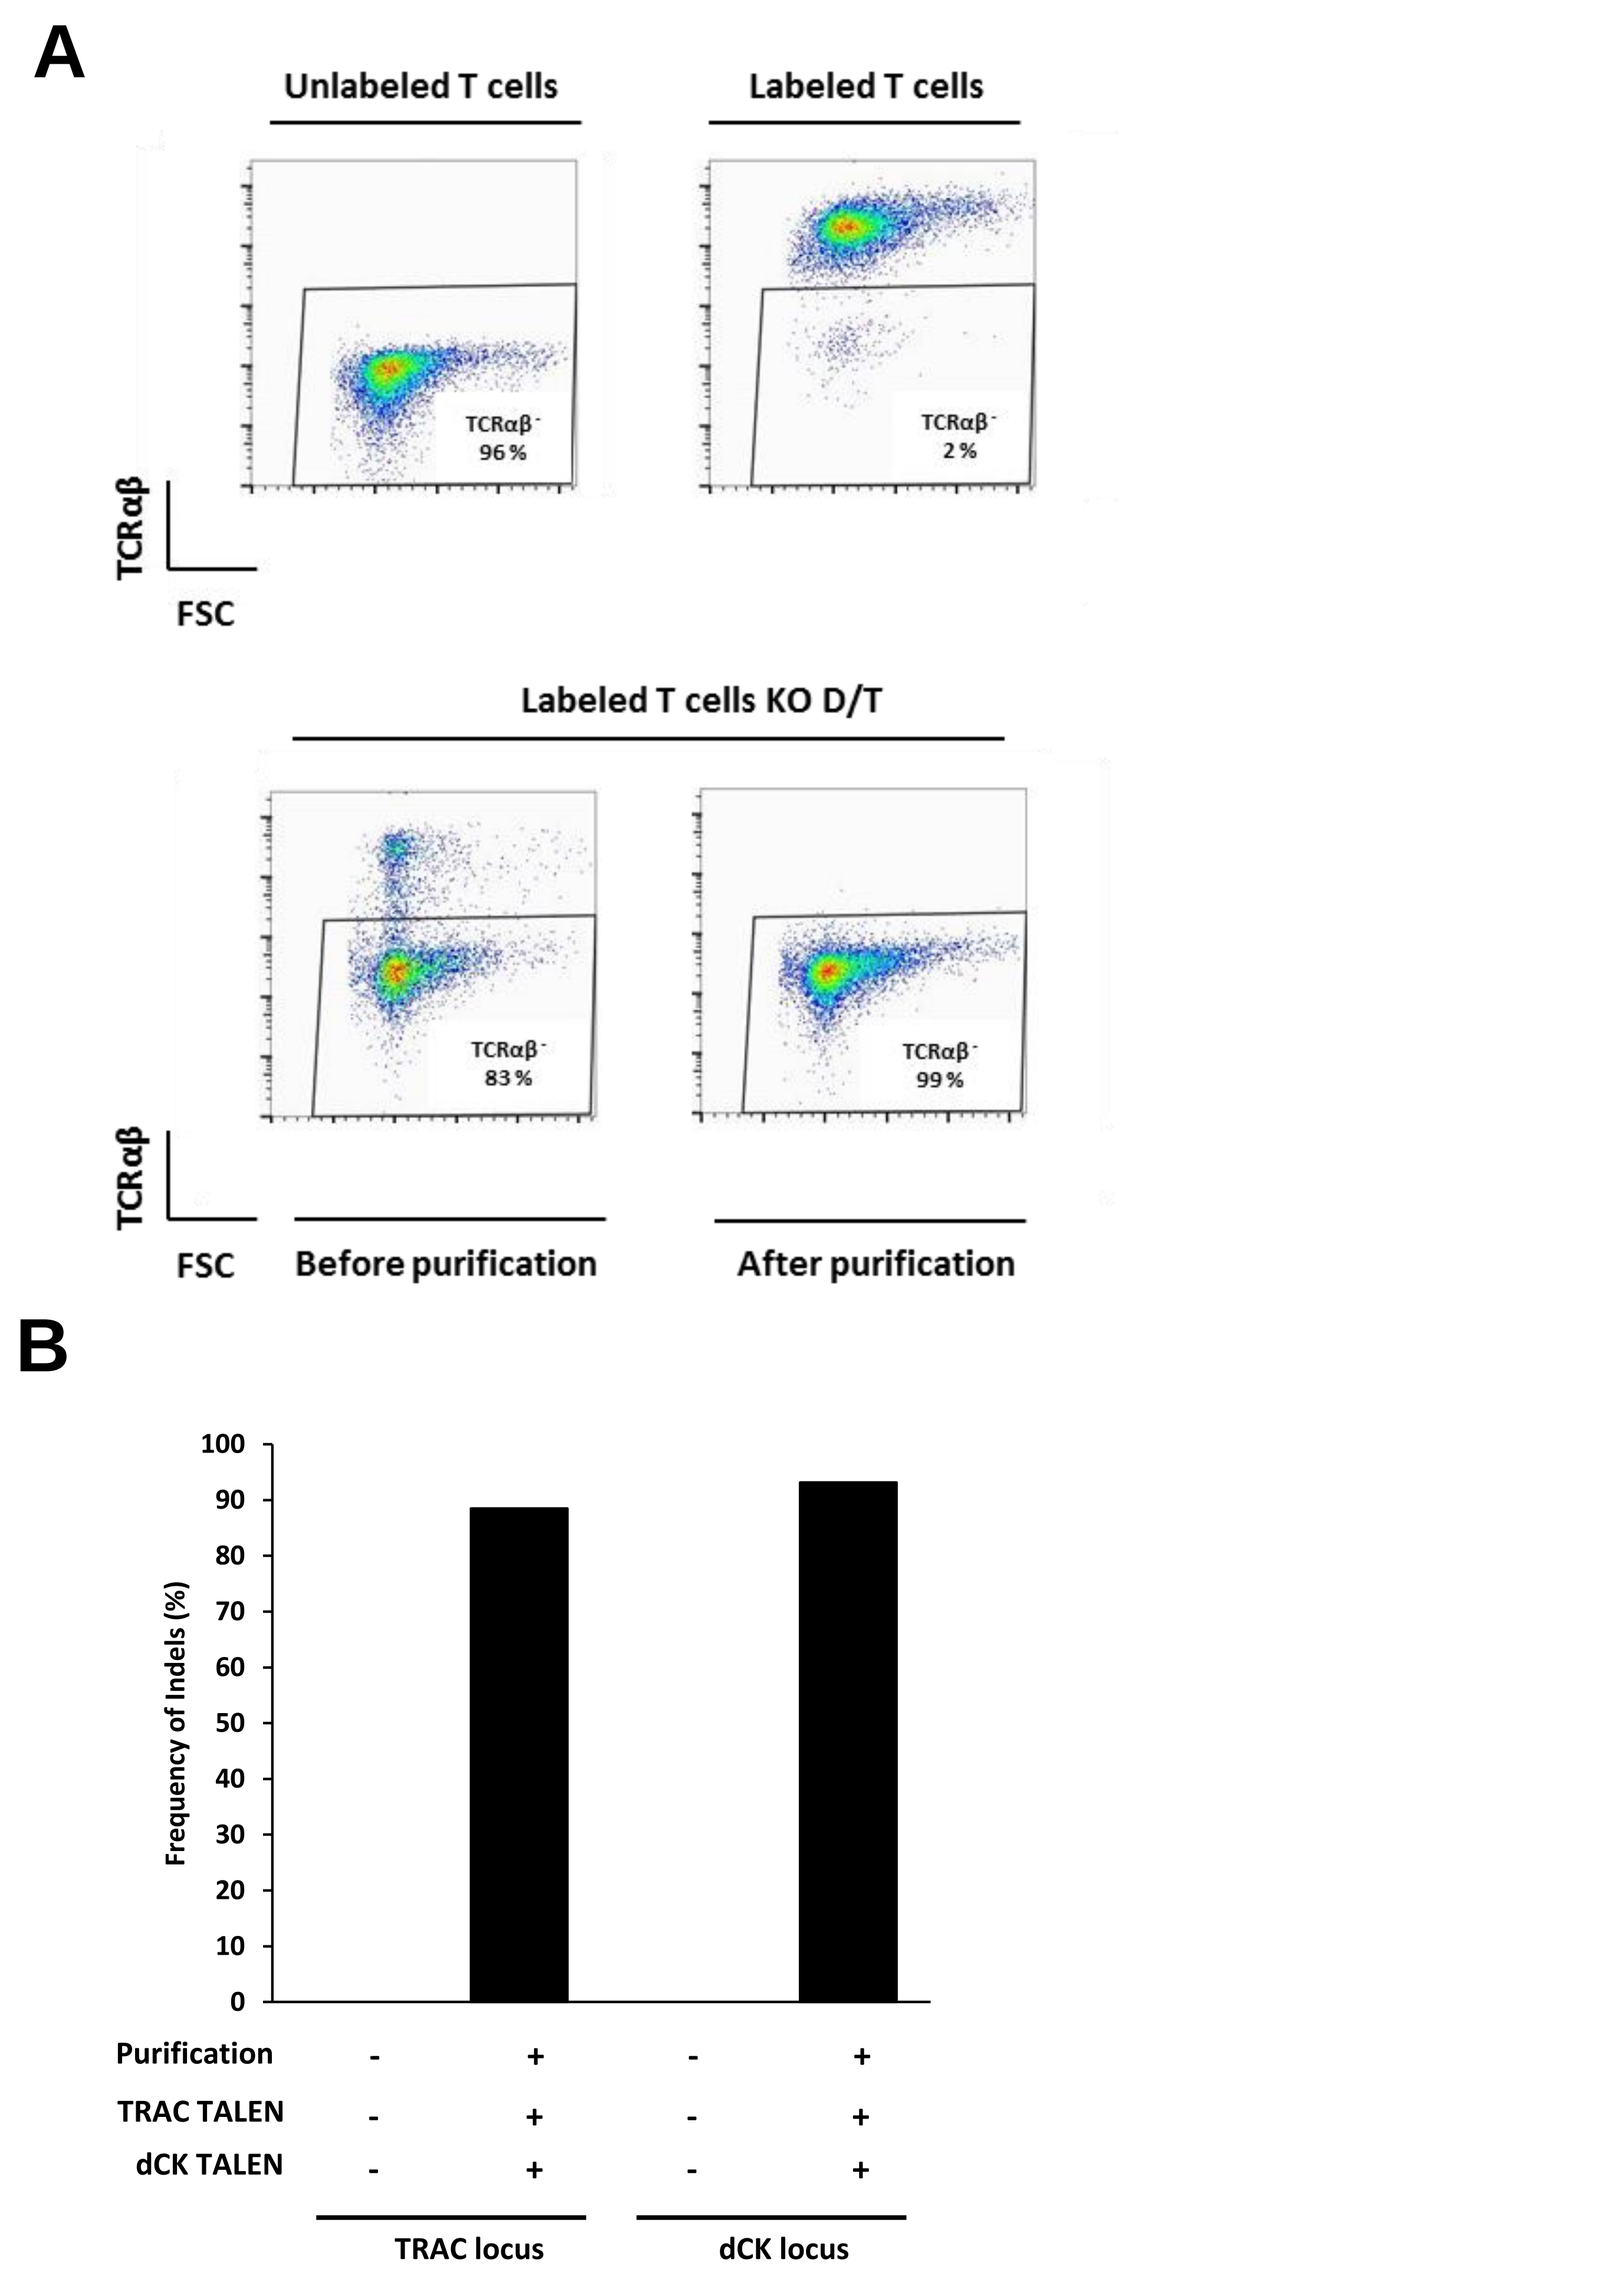
A
Unlabeled T cells
Labeled T cells
TCRαβ- 96 %
TCRαβ- 2 %
TCRαβ
FSC
Labeled T cells KO D/T
TCRαβ- 83 %
TCRαβ- 99 %
Before purification
After purification
B
Frequency of Indels (%)
Purification
TRAC TALEN
dCK TALEN
TRAC locus
dCK locus

Figure 5. Multiplex TALEN treatment of primary T cells disrupts TCR membrane expression. (a) Purification of TRAC KO T cells from the bulk population of cell transfected with mRNA encoding TRAC and dCK TALEN. Labeling control experiment performed with T cells in the presence or in the absence of anti TCR mAb (upper panel) and monitoring of TCRαβ-deficient cells gathered before and after TRAC KO T cells purification using anti-CD3 magnetic beads and MACS LD-column (lower panel). **(b)** Genotypic characterization of TALEN-mediated inactivation of TRAC and dCK genes by high throughput DNA sequencing before and after TCR negative cells purification.

TALEN-treated CAR T cells are resistant to multiple PNAs and are able to proliferate in the presence of their clinically relevant dose

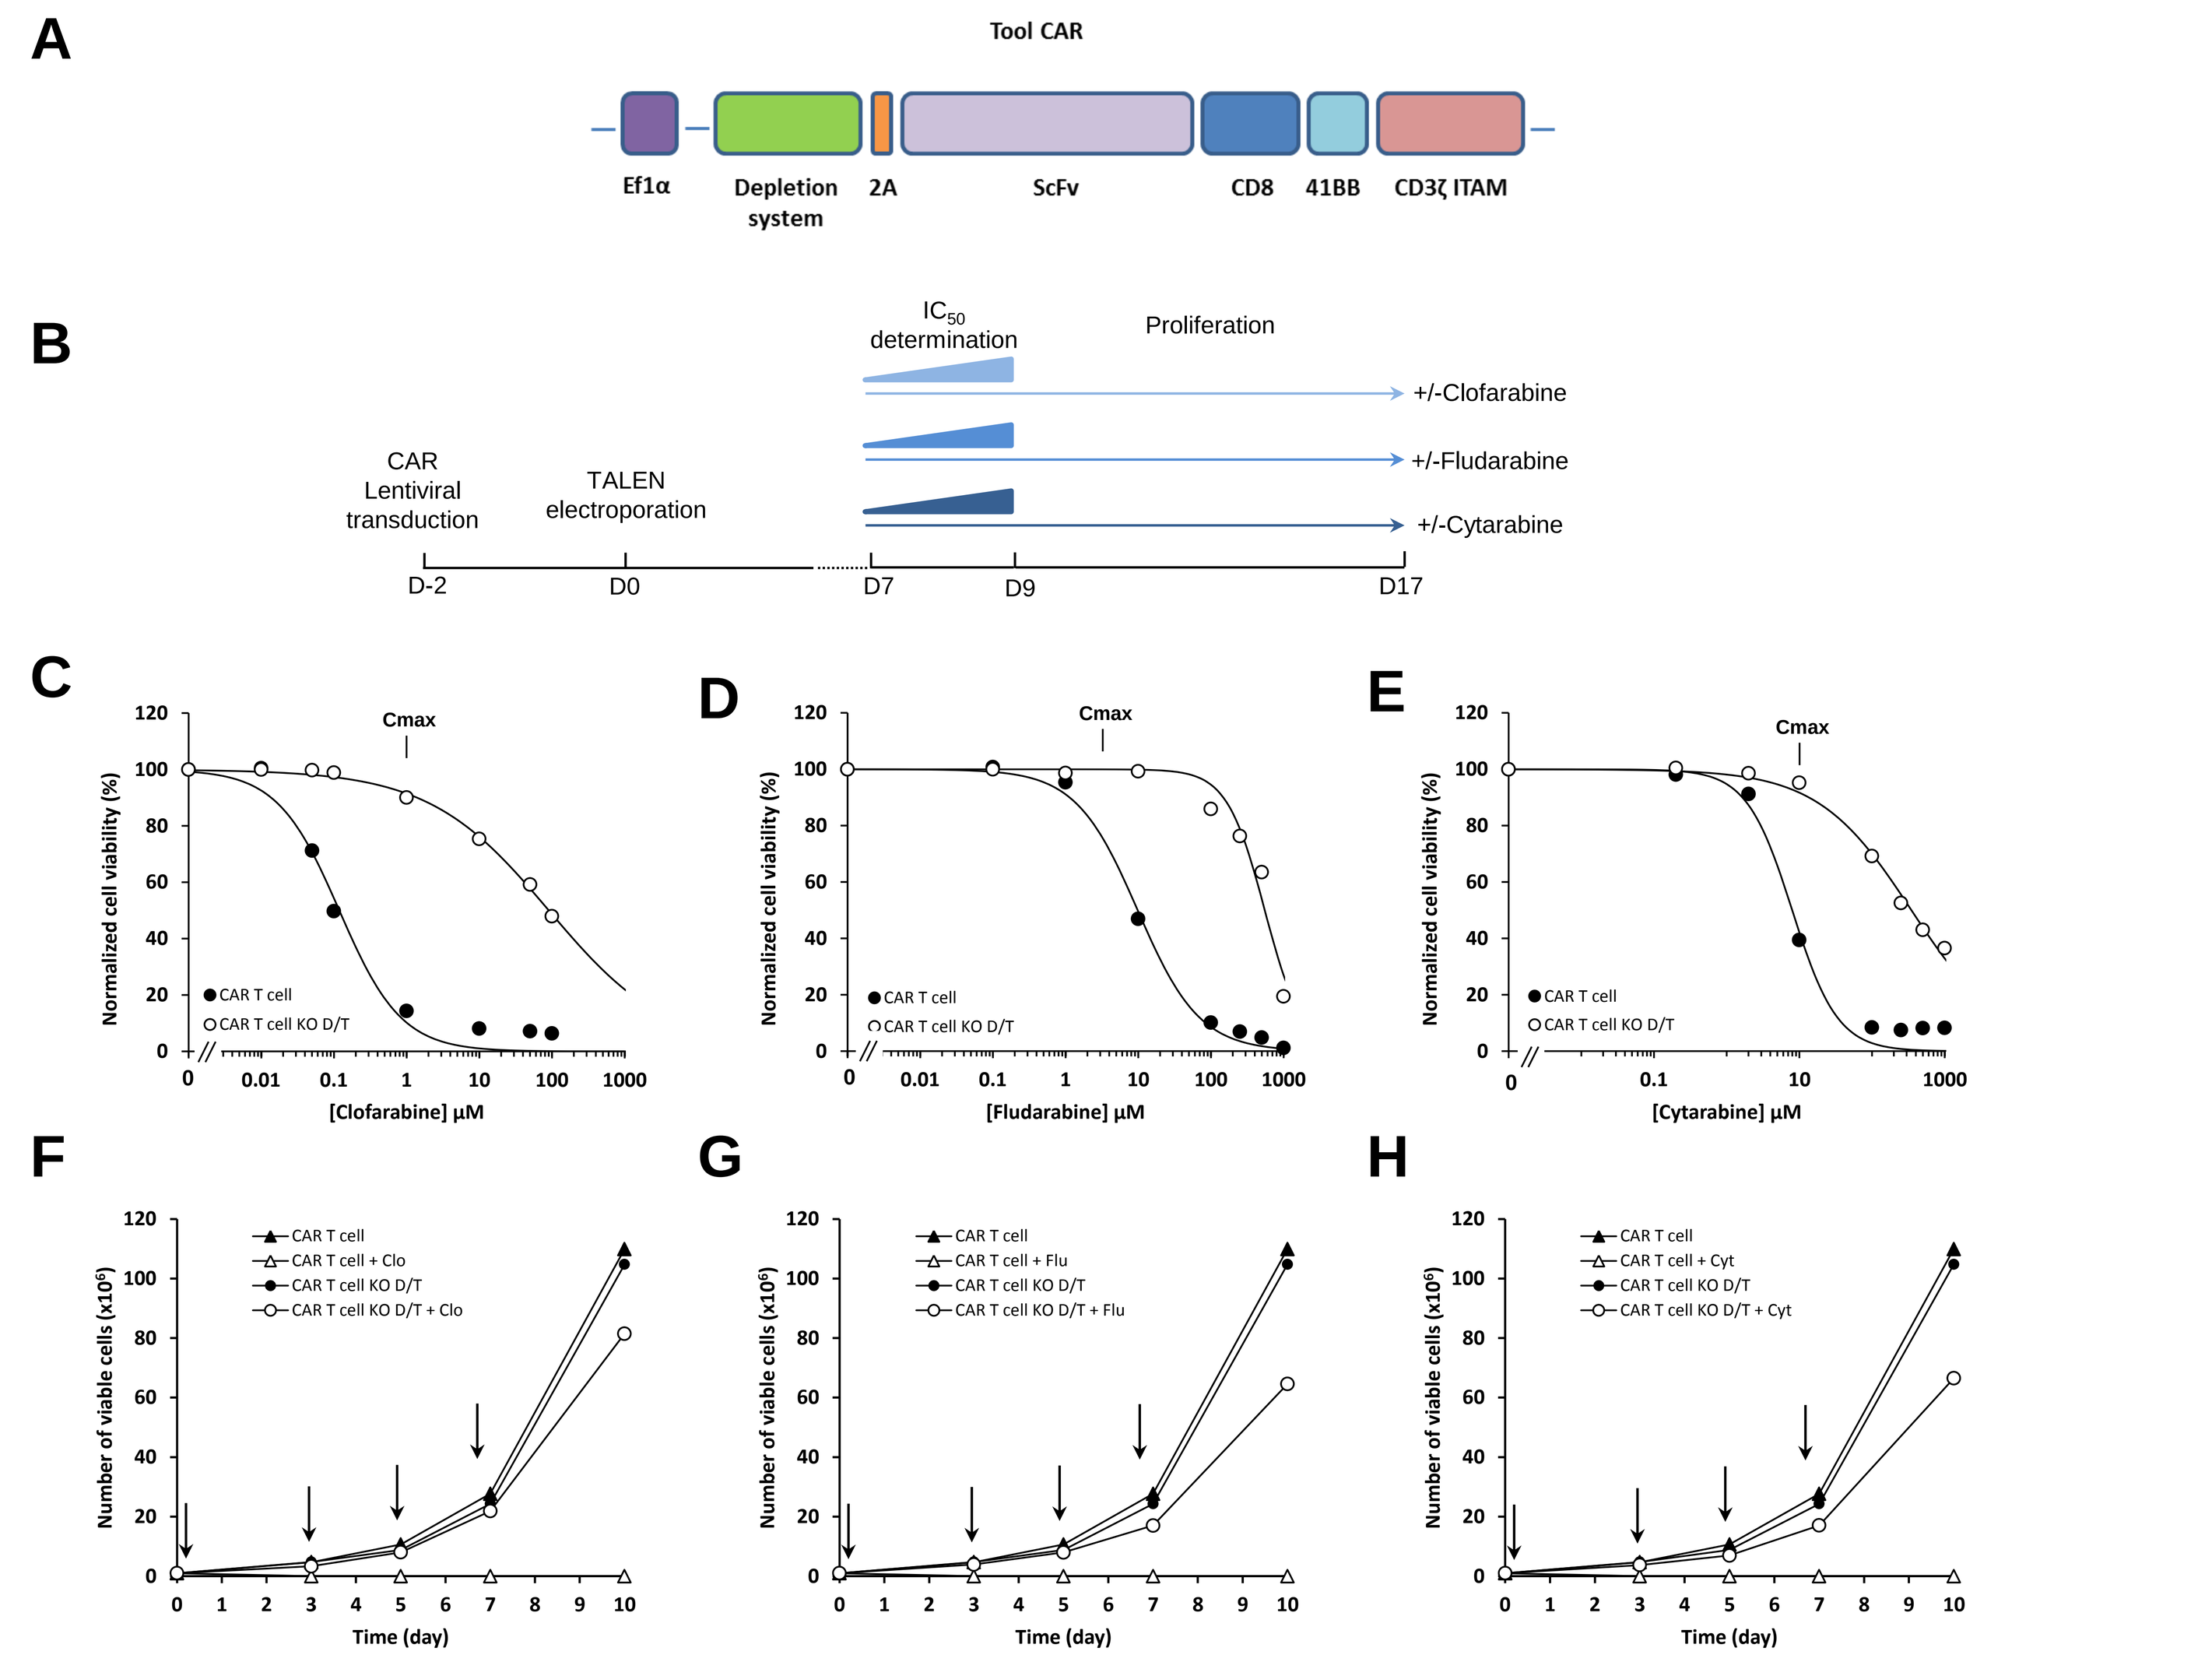
A
Tool CAR
Ef1α
Depletion system
2A
ScFv
CD8
41BB
CD3ζ ITAM
B
IC50 determination
Proliferation
+/-Clofarabine
+/-Fludarabine
+/-Cytarabine
CAR Lentiviral transduction
TALEN electroporation
D-2
D0
D7
D9
D17
C
D
E
Normalized cell viability (%)
Cmax
CAR T cell
CAR T cell KO D/T
[Clofarabine] µM
[Fludarabine] µM
[Cytarabine] µM
F
G
H
Number of viable cells (x10⁶)
CAR T cell + Clo
CAR T cell KO D/T + Clo
CAR T cell + Flu
CAR T cell KO D/T + Flu
CAR T cell + Cyt
CAR T cell KO D/T + Cyt
Time (day)

Figure 6. TALEN-treated CAR T cells are resistant to lethal dose of different purine and pyrimidine nucleotide analogues. (a) Scheme of the polycistronic DNA expression cassette encoding the membrane exposed rituximab-dependent depletion system (RQR8), followed by a 2A cis-acting hydrolase element and a tool CAR. **(b)** Scheme of the experimental workflow used to engineer and characterize T cells proliferative capacity and resistance properties toward clofarabine, fludarabine and Cytarabine. **(c, d and e)** Clofarabine, fludarabine and cytarabine IC_{50} determination. Experimental viability values obtained after 2 days of incubation were normalized with respect to the one obtained in the absence of drug. **(f, g and h).** Proliferation capacity of CAR T cells in the presence or in the absence of clofarabine, fludarabine and cytarabine. 10^6 CAR T cells were grown for a total of 10 days in the absence or in the presence of 1, 5 and 10 µM of clofarabine, fludarabine and cytarabine respectively. Vertical arrows indicate each passage step where cells were diluted to 10^6 cell/mL in the presence or in the absence of drug.

TALEN-treated CAR T cells display efficient antitumor activity

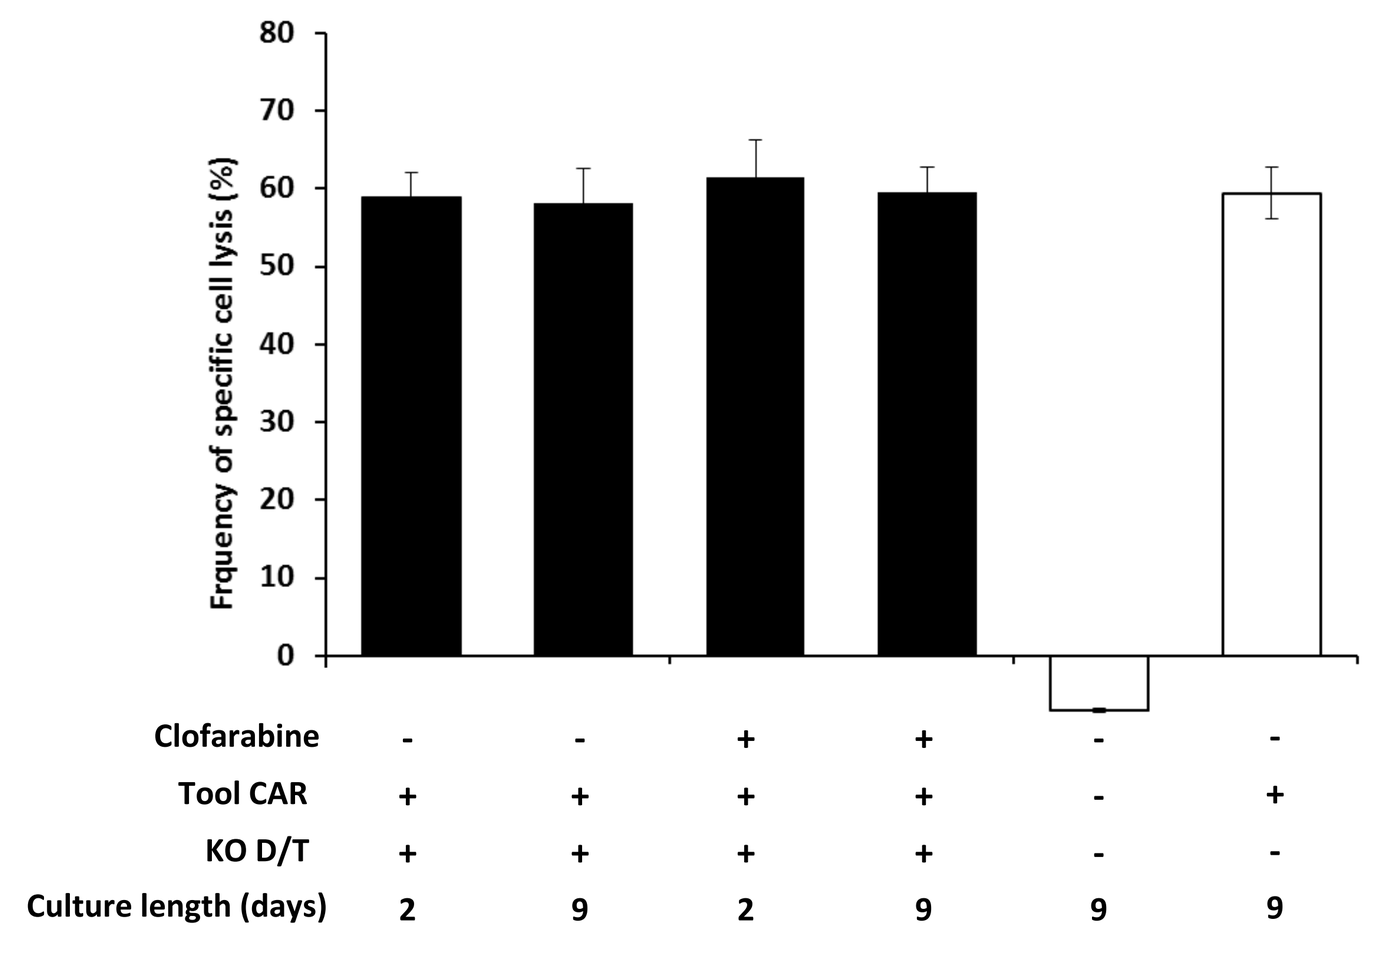
Frquency of specific cell lysis (%)
Clofarabine
Tool CAR
KO D/T
Culture length (days)

Figure 8. TALEN-treated CAR T cells retain their cytolytic activity after 2 to 9 days of culture in the presence of clinically relevant dose of clofarabine. 10^4 Daudi (target cells) and 10^4 K562 (negative control) cells labeled respectively with CellTrace™ CFSE and CellTrace™ violet were co-cultured in 100 µL for 5H at 37°C with 10^5 of TALEN-treated CAR T cells grown for 2 or 9 days in the presence or in the absence of 1 µM clofarabine. Cells were then recovered and their viability was determined by flow cytometry. Viability of Daudi and K562 cells were used to calculate the frequencies of specific cell lysis (lanes 1 to 4). T cells and CAR T cells cultured in the absence of clofarabine for 9 days were respectively used as negative and positive controls of specific cell lysis activity (lanes 5 and 6). Results are displayed as specific cell lysis activities relative to the control experiment performed with CAR T cells. Error bars represent the mean of 2 independent experiments + SD.

TALEN-treated CAR T cells can pair up with PNA chemotherapy for better antitumor activity

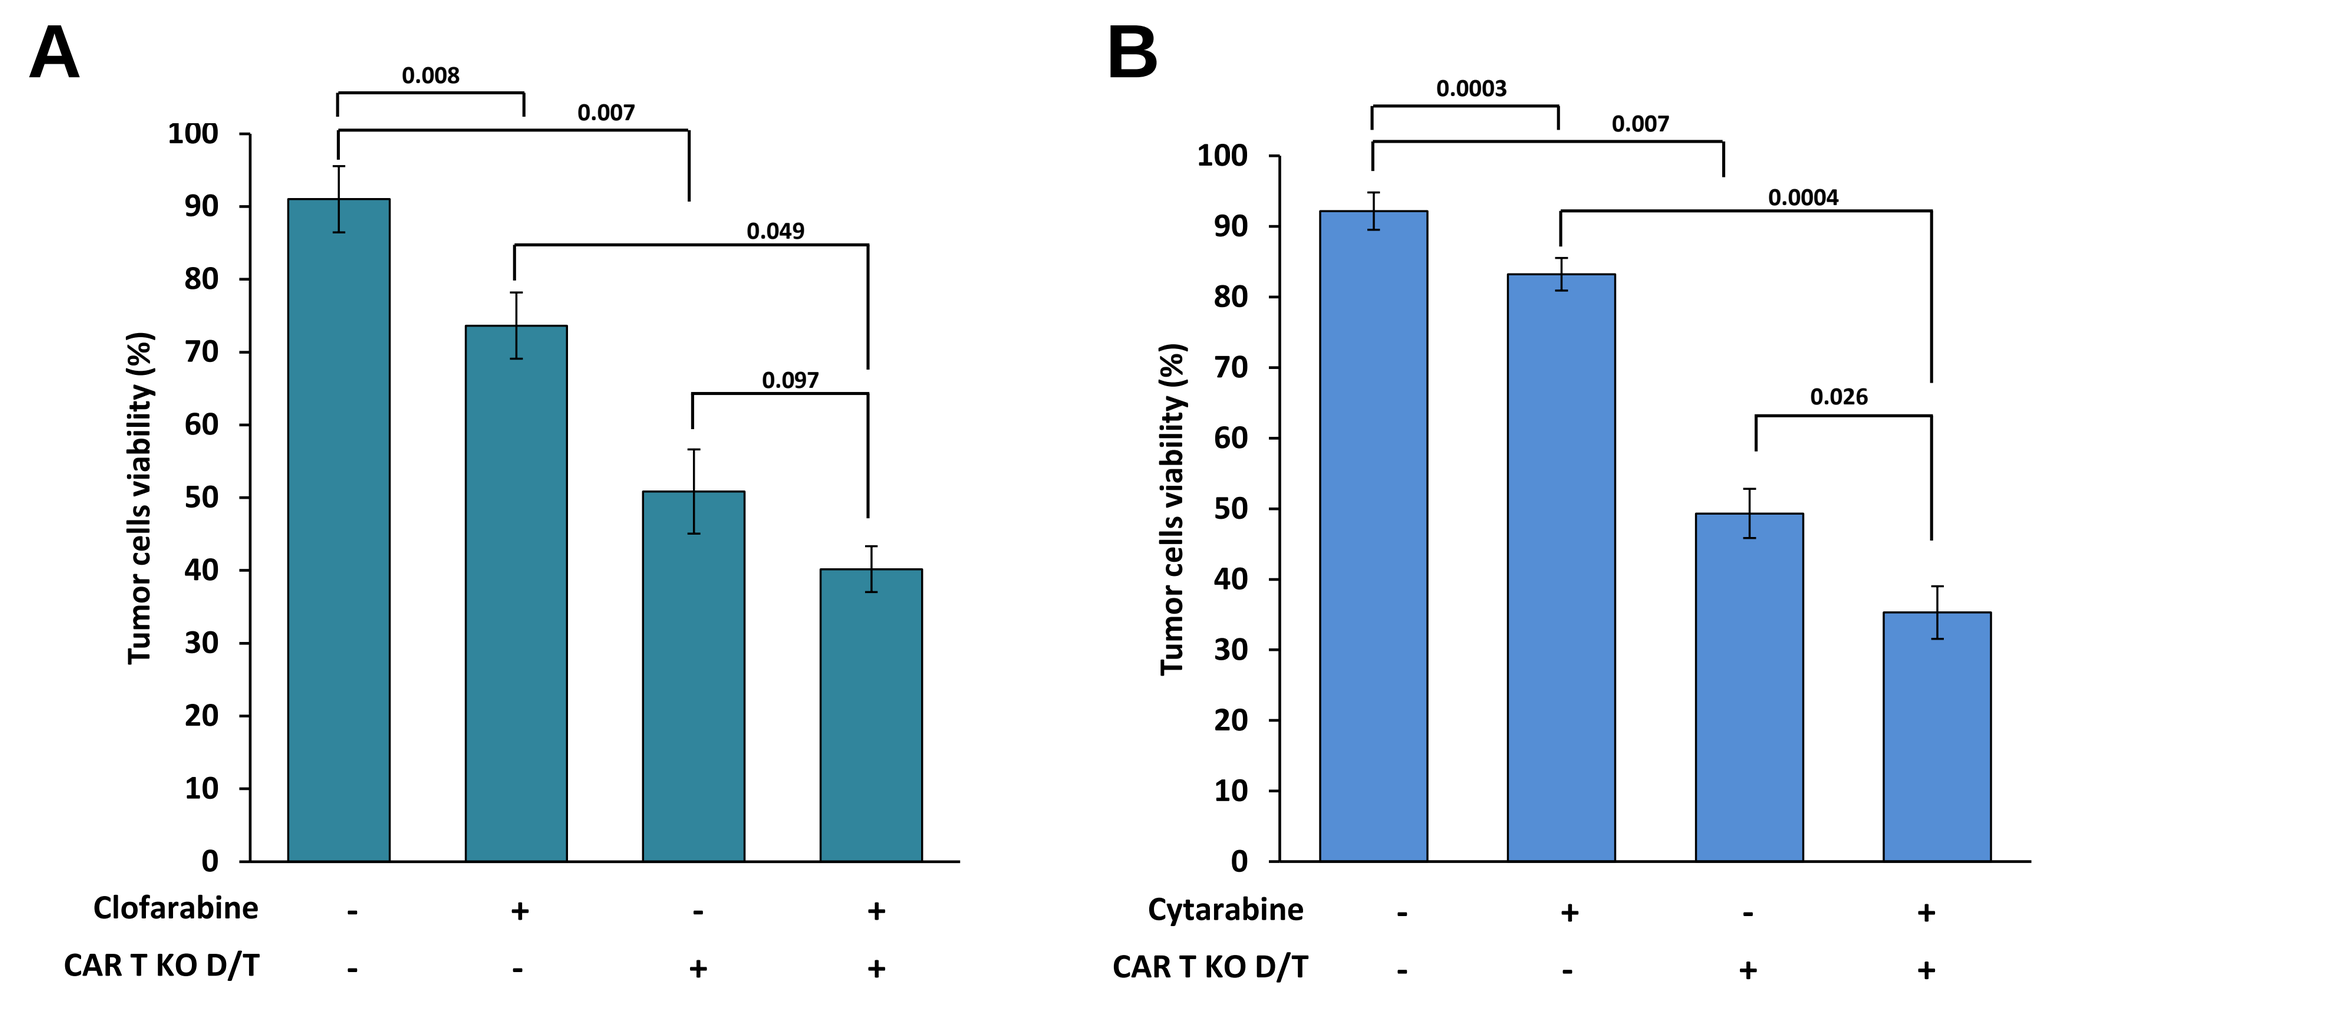
A
B
Tumor cells viability (%)
0.008
0.007
0.049
0.097
0.0003
0.0004
0.026
Clofarabine
Cytarabine
CAR T KO D/T

Figure 9. TALEN-treated CAR T cells can pair up with chemotherapy for better antitumor activity. 10^4 Daudi cells (target cells) preincubated in the presence or in the absence of PNA (1 µM clofarabine or 10 µM cytarabine) for 24H, were labeled with CellTrace™ CFSE and co-cultured in 100 µL for 5H at 37°C in the absence or the presence of 10^5 TALEN-treated CAR T cells (CAR T KO D/T). Cells were then recovered and their viability was determined by flow cytometry. The residual viability of tumor cells (Daudi) obtained in the presence or in the absence of clofarabine **(a)** or cytarabine **(b)** are displayed. p values numbers, calculated according to the procedure described in material and methods, are indicated. Error bars represent the mean of 2 or 3 independent experiments +/- SD performed in the presence of clofarabine or cytarabine respectively.

Conclusions

In summary, we report the development of multidrug resistant and TCRαβ-deficient CAR T cells compatible with allogeneic immunotherapy. Engineered CAR T cells resistance towards PNA chemotherapies enabled them to proliferate under lymphodepleting conditions while retaining their antitumor activity and specificity. Such PNA resistance provides two main advantages. First, it could enable allogeneically transferred CAR T cells to survive lymphodepleting regimens used to maintain the host immune system in check and control their rate of ablation via a HvG reaction. Second, it could allow combination therapy, an approach likely to improve clinical outcomes by potentializing CAR T cell antitumor activity *in vivo*. In addition, due to their lack of surface exposed-TCR, these cells could be theoretically generated from third party healthy donors and used as allogeneic immunotherapy without generating GvH reaction. In conclusion, we believe that the cellular engineering strategy described in this work will provide a basic frame work to generate a universal T cell that will foster large scale utilization of ACT and thus could benefit a broader range of patients.

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis SA and its business. Cellectis SA is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. This communication contains Cellectis' proprietary information. Talen® and Cellectis® are trademarks owned by the Cellectis Group.

Targeted genome modifications for improved adoptive T-Cell immunotherapy

Laurent Poirot, Alexandre Juillerat, Julien Valton, Anne-Sophie Gautron and Philippe Duchateau

CELLECTIS SA, 8 rue de la Croix Jarry, 75013 Paris, France. Email: philippe.duchateau@cellectis.com

#1 Abstract

Chimeric antigen receptor (CAR)-redirected T-cells have given rise to long-term durable remissions and remarkable objective response rates in patients with refractory leukemia, raising hopes that a wider application of CAR technology may lead to a new paradigm in cancer treatment. Similarly, transcription activator-like effector nuclease (TALEN)-mediated gene editing has emerged as powerful strategy to introduce targeted mutations and holds great promise in therapeutics and offers multiple opportunities to improve CAR T-cell therapies. The knockout of the TCR alpha gene eliminates TCR expression and abrogates the donor T-cell's potential for graft-versus-host disease (GvHD) while maintaining a potent anti-tumoral activity. Thus it is possible to manufacture T-cells from third-party healthy donors to generate allogeneic "off-the-shelf" engineered CAR T-cells. Disruption of CD52 or deoxycytidine kinase genes may be a useful approach to makes T-cells compatible with concurrent oncology treatments such as alemtuzumab or Fludarabine while the bypass of key immune checkpoint regulators such PD1/PDL1 by inactivation of T-cells receptors would potentiate the antitumor T-cell response by impairing the interaction of the inhibitory receptor PD-1 on T-cells with PD-L1 expressed on tumor cells. Here, we will present *in vitro* and *in vivo* proof of concepts demonstrating the potential of TALEN-mediated gene editing for adoptive T-cell therapy.

#2 Sharpening the power of T-cell by editing the genome

Autologous CAR Therapy

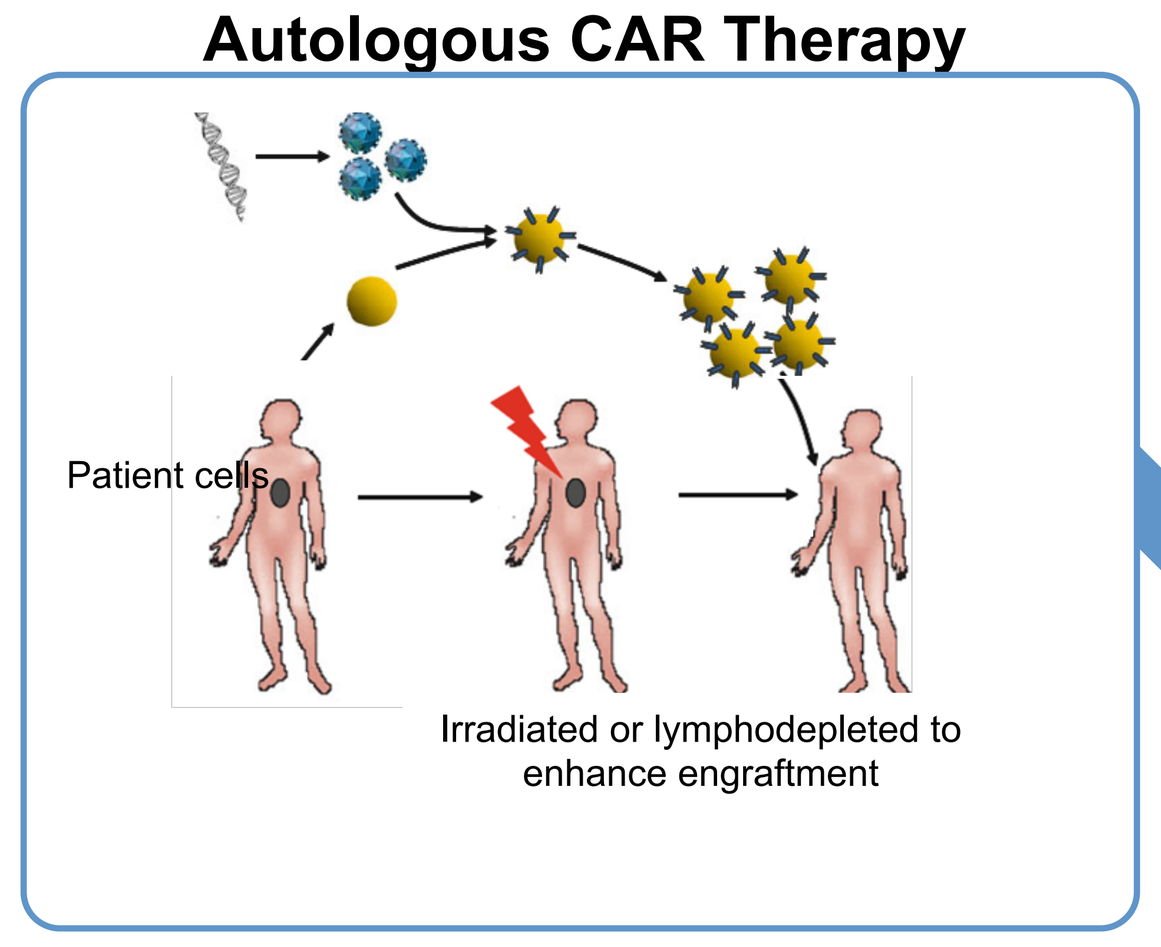
Patient cells
Irradiated or lymphodepleted to enhance engraftment

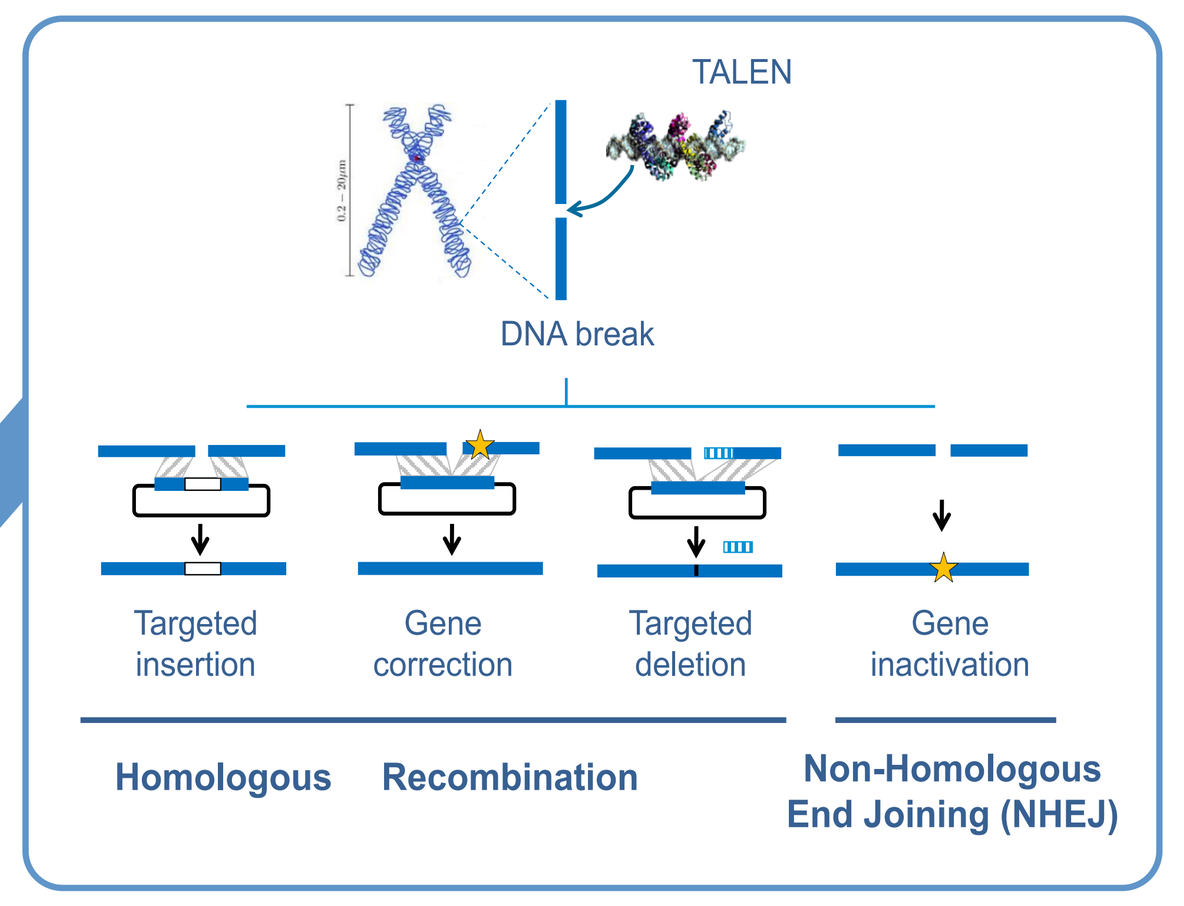
TALEN
DNA break
Targeted insertion
Gene correction
Targeted deletion
Gene inactivation
Homologous
Recombination
Non-Homologous End Joining (NHEJ)

Allogeneic, non alloreactive	**Off-the-shelf product (KO TCR)**
Resistant to chemotherapy	**Compatible with SoC, Use in combination therapies (KO CD52)**
Resistant to lymphodepleting agents	**Enhanced engraftment (KO DCK)**
Resistant to tumor inhibition	**Enhanced efficacy (KO PD1, KO CTLA4,…)**
Suppressed cross T-cell reaction	**Better suited for specific tumors (T-cell auto Ag)**

TALEN enable us to efficiently modify or inactivate any target gene in a highly efficient, specific and precise manner. Thus, We can engineered product candidates to achieve desired clinical attributes. By editing and engineering the genomes of the T-cell, we have been able to establish "smart" CAR T-cells that include the above traits.

#3 Efficient Gene inactivation using TALEN mRNA electroporation

AgilePulse™ Electroporation system

Cuvette or Large volume chamber (12 ml)
$2x10^6$ -10^9 cells

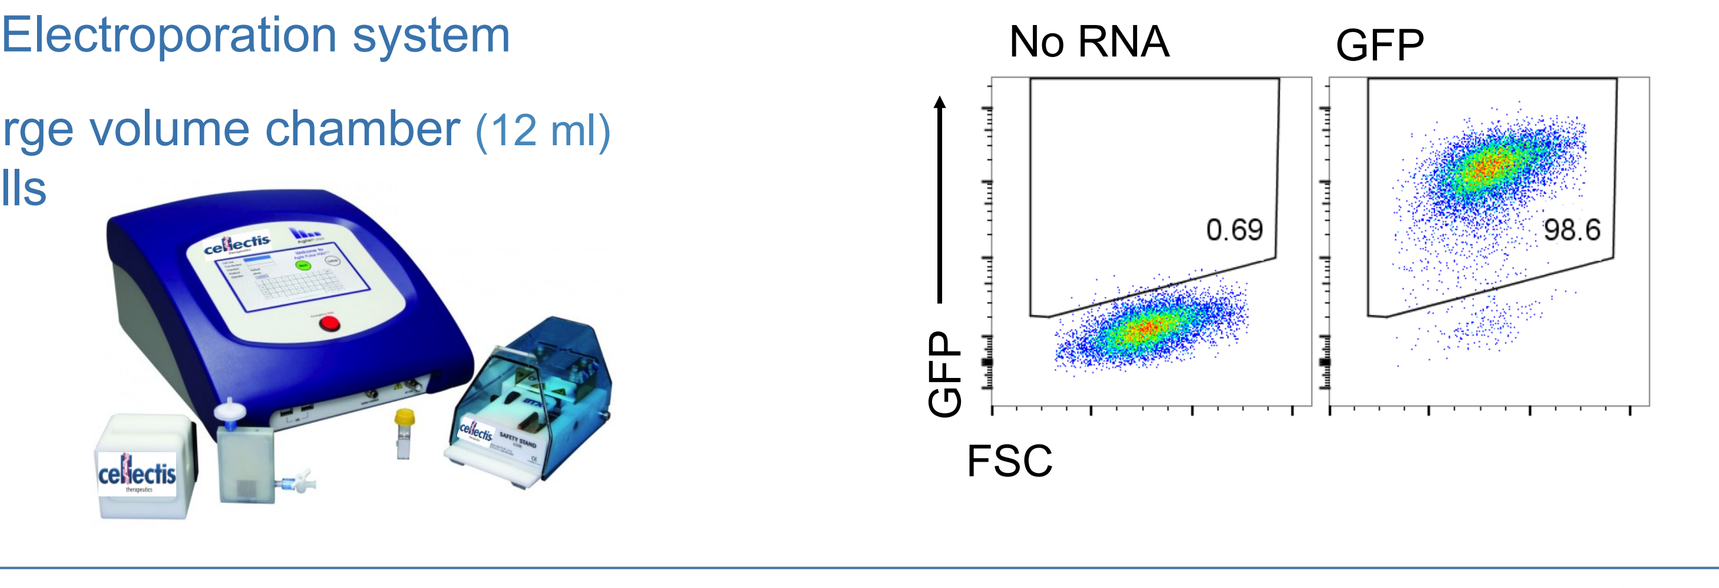
No RNA
GFP
0.69
98.6
GFP
FSC

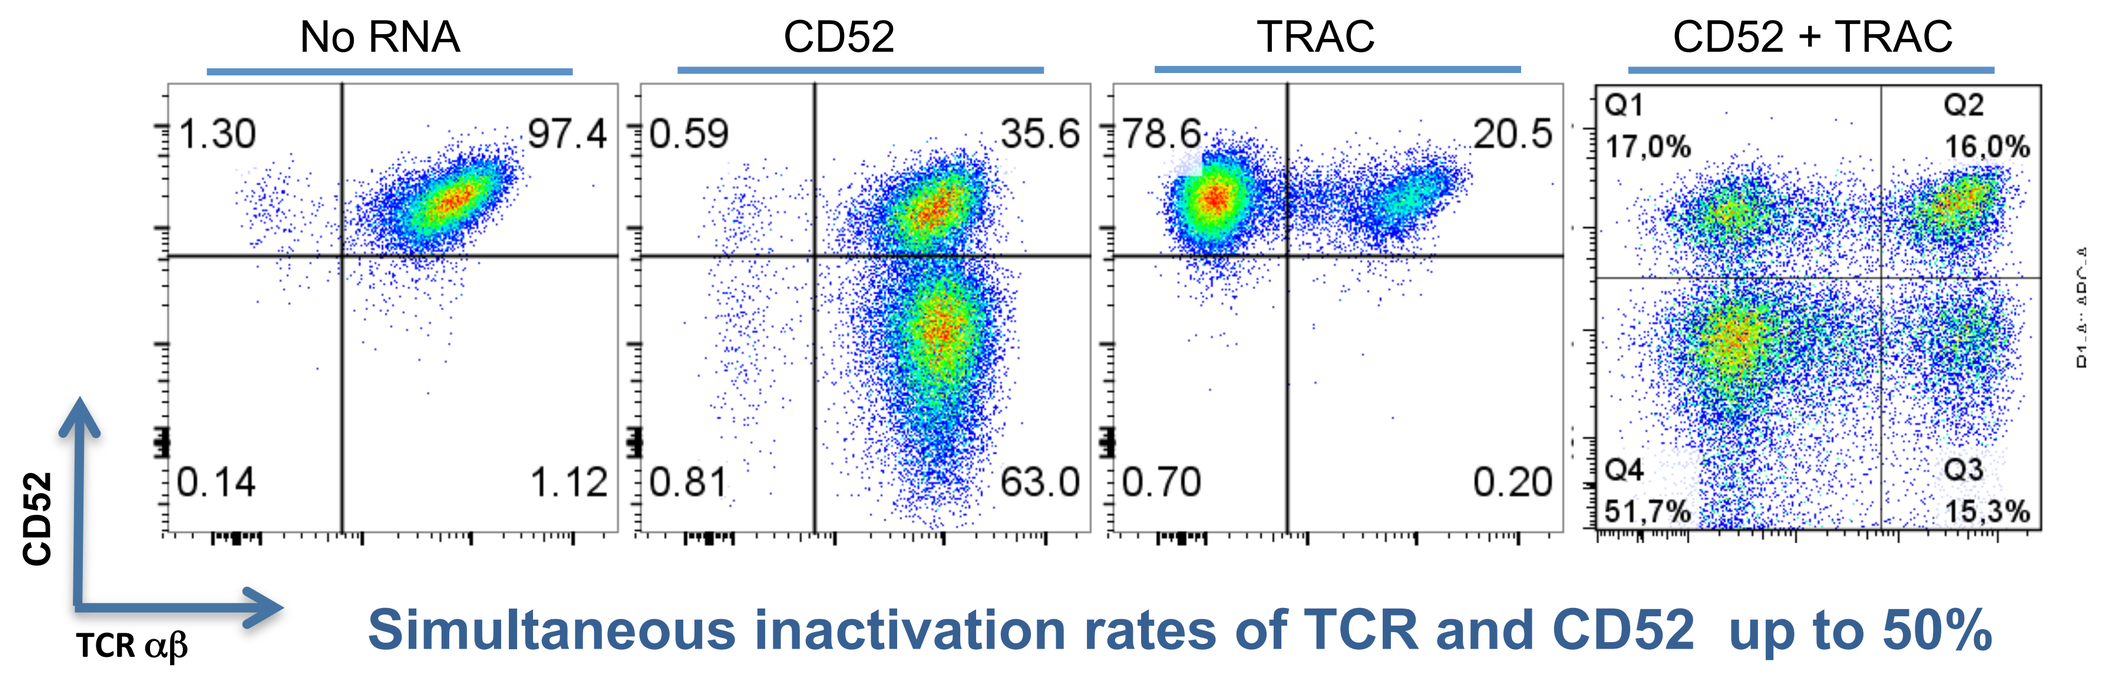
No RNA
CD52
TRAC
CD52 + TRAC
1.30
97.4
0.14
1.12
0.59
35.6
0.81
63.0
78.6
20.5
0.70
0.20
Q1 17,0%
Q2 16,0%
Q4 51,7%
Q3 15,3%
CD52
TCR αβ

Simultaneous inactivation rates of TCR and CD52 up to 50%

- A mean of mutation frequency ~ 80% at several loci in primary T-cells
- T-cell viability upon electroporation >90%

In order to perform gene editing, we use our proprietary PulseAgile electroporation technology to introduce nucleases as mRNA inside the target T-cell to access the cell's DNA. The mRNA molecules are rapidly degraded by the cell, which means that the nuclease is only expressed for a short time, enough to make permanent changes in the cell's targeted gene.

#4 Towards a "universal" adoptive T-cell immunotherapy

Inactivation of TCR in primary T-cells to avoid GvHD

Autologous CAR Therapy | **"Off the shelf" Allogeneic CAR Therapy with enhanced efficacy**

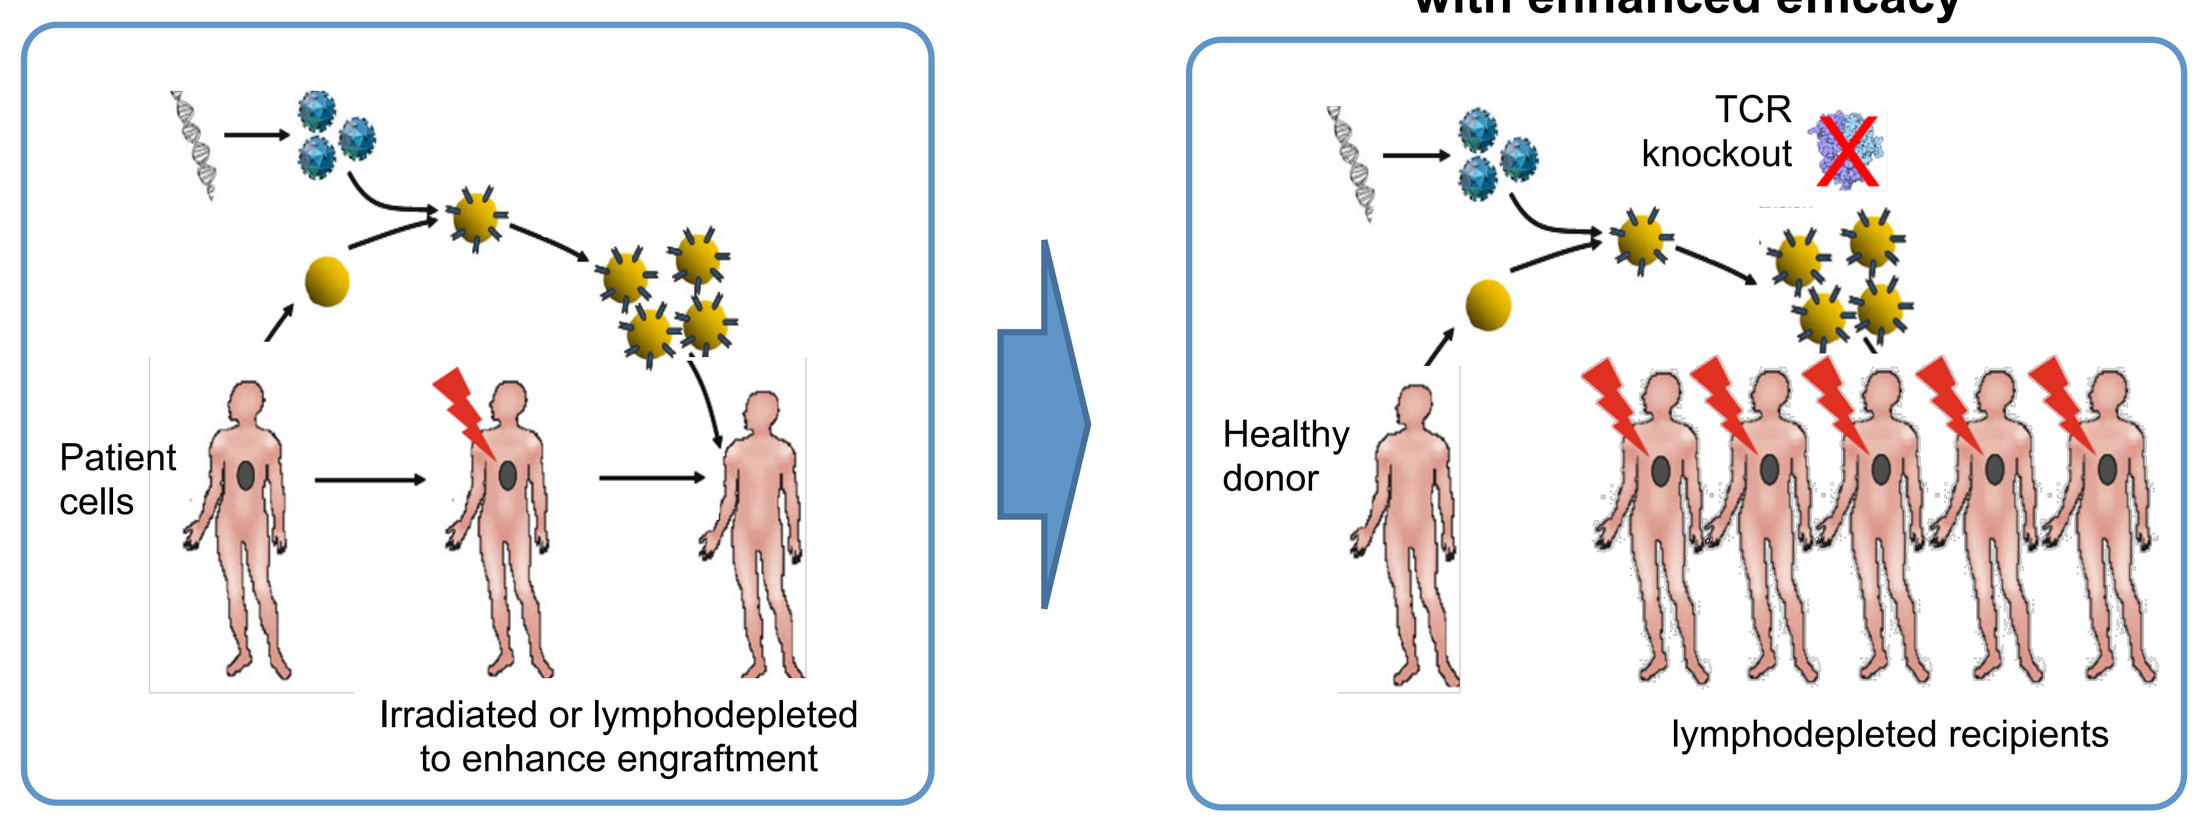
Patient cells
Irradiated or lymphodepleted to enhance engraftment
TCR knockout
Healthy donor
lymphodepleted recipients

Personalized therapy

Autologous CAR T cell therapy demonstrates impressive efficacy but faces major challenges (costs, logistics)

Cells from one donor could be used to treat hundreds of patients

Knocking Out the TCR gene would allow the use of allogenic CAR T-Cells without GvHD

A TALEN was designed to cleave the first exon of the TCRα constant region

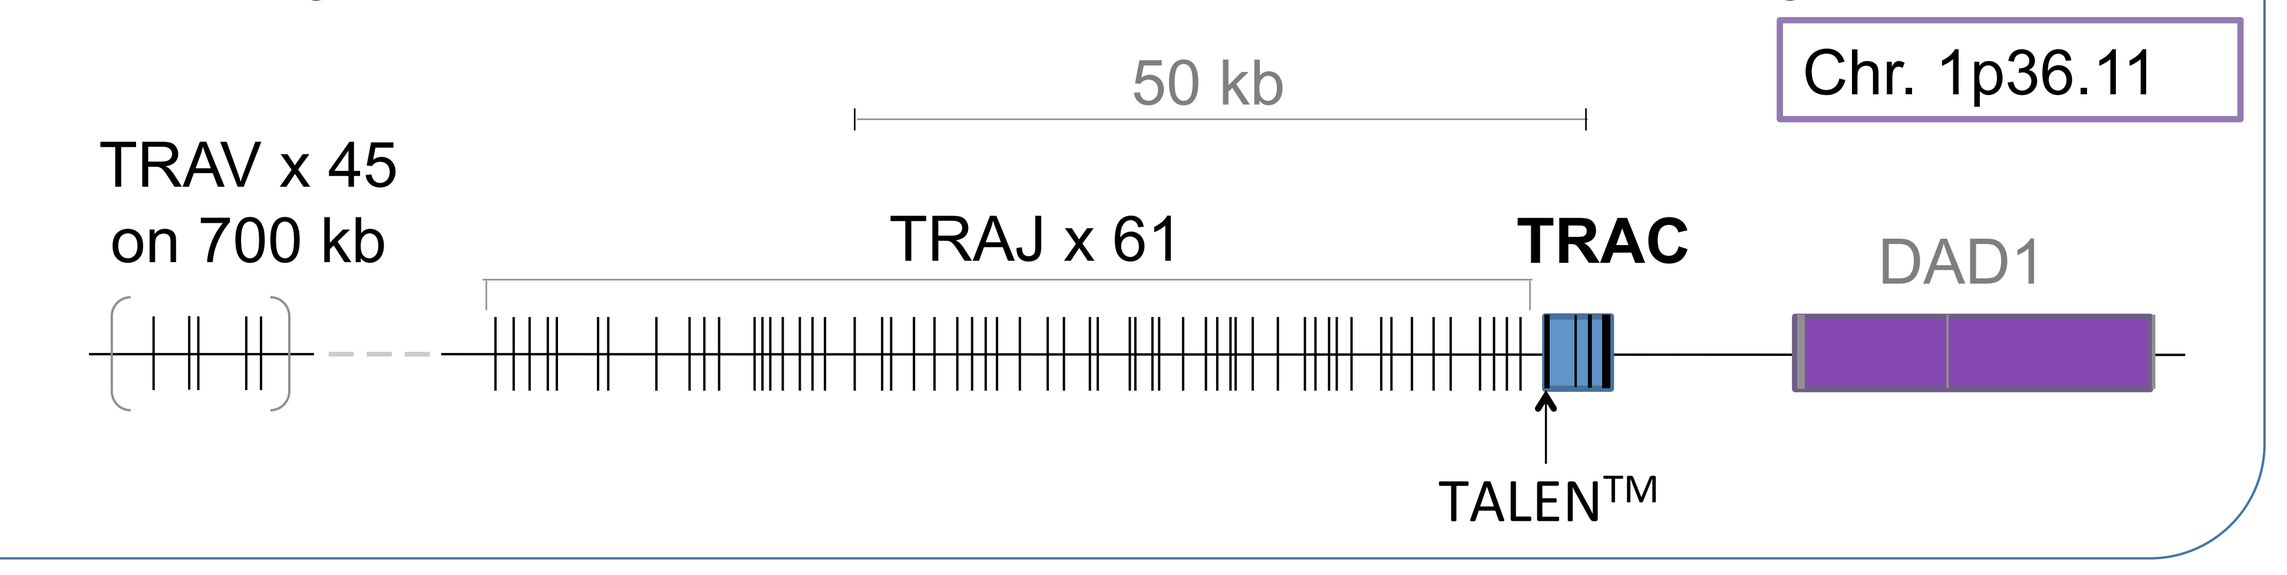
Chr. 1p36.11
50 kb
TRAV x 45
on 700 kb
TRAJ x 61
TRAC
DAD1
TALEN™

#5 Off the shelf « TCRα-deficient T-cells" for adoptive immunotherapy

TCR-deficient cells cannot be activated via TCR

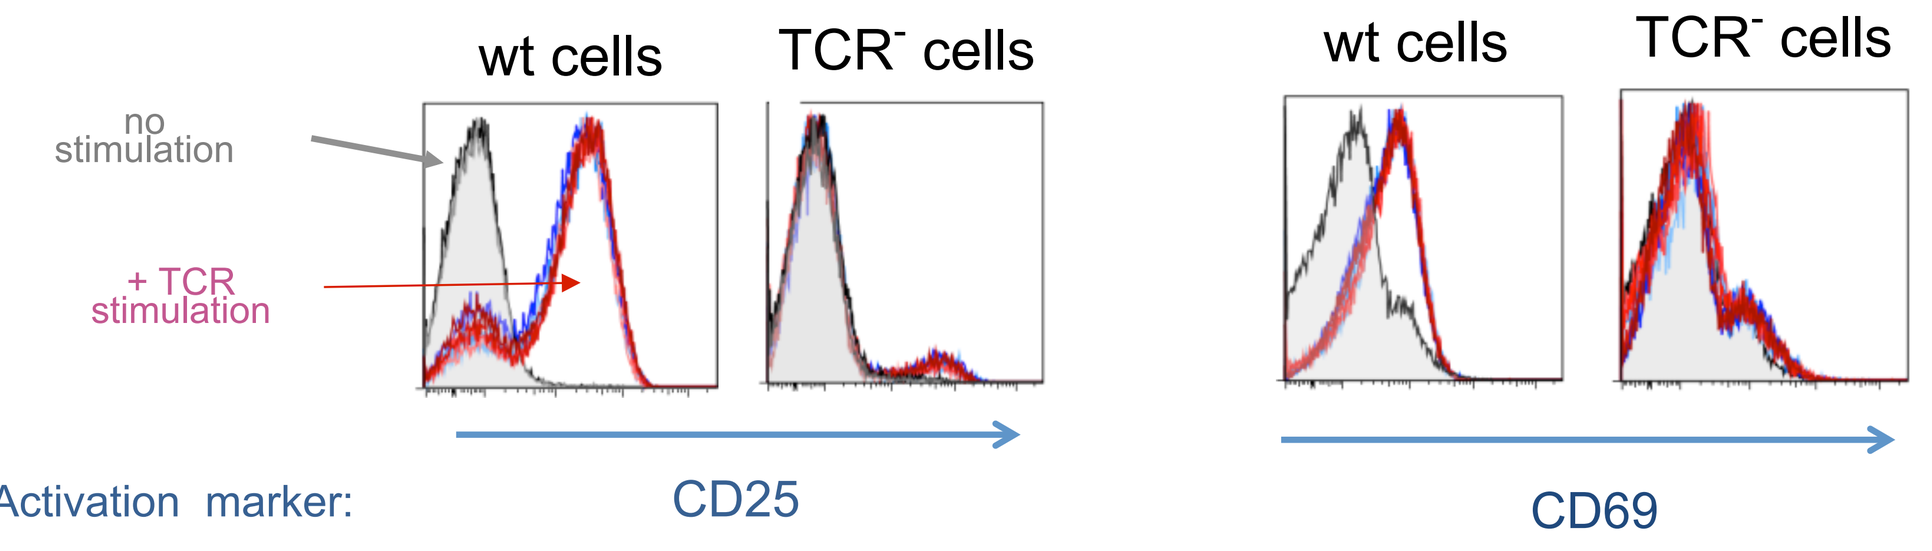
wt cells
TCR⁻ cells
wt cells
TCR⁻ cells
no stimulation
+ TCR stimulation
Activation marker:
CD25
CD69

TCRα-deficient T-cells do not induce GvHD

NOG mice irradiated (2.5 Gy) 1 day before T-cell injection (i.v., $30x10^6$ cells)

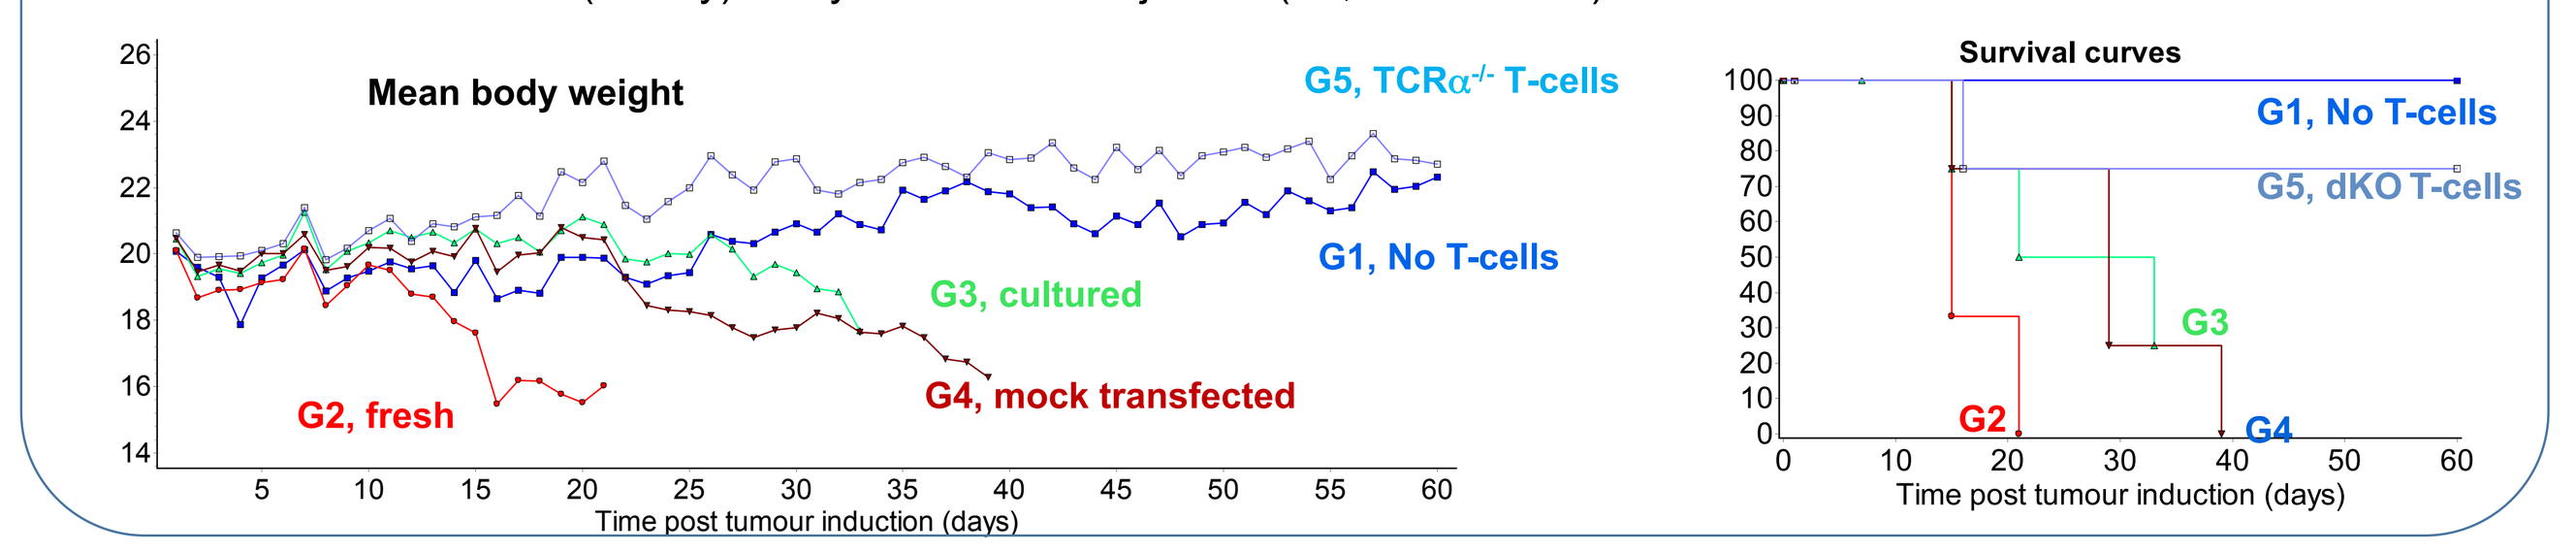
Mean body weight
G5, TCRα-/- T-cells
G1, No T-cells
G3, cultured
G2, fresh
G4, mock transfected
Time post tumour induction (days)
Survival curves
G1, No T-cells
G5, dKO T-cells
G3
G2
G4
Time post tumour induction (days)

TCRα/CD52-deficient CAR T-cells maintain anti-tumor activity in presence of alemtuzumab

Anti-tumor activity in the presence of alemtuzumab therapy using tumor cells partially resistant to alemtuzumab ($CD52^{+/-}$ Raji-Luc cells

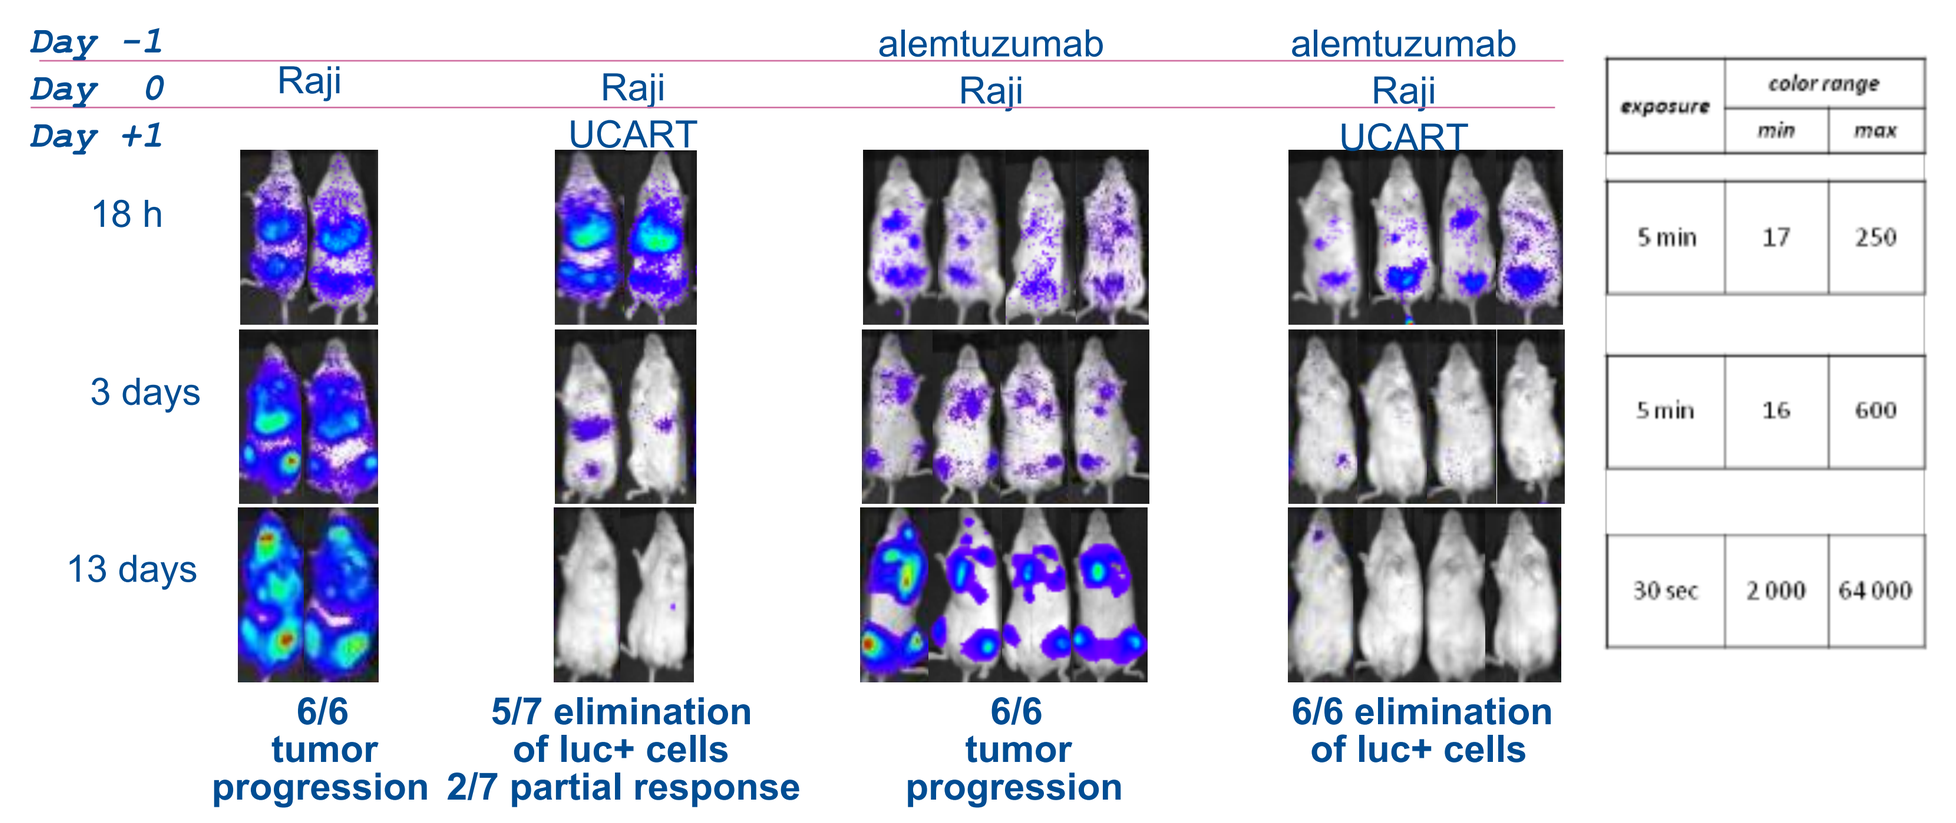
Day -1
Day 0
Day +1
Raji
Raji UCART
alemtuzumab Raji
alemtuzumab Raji UCART
18 h
3 days
13 days
6/6 tumor progression
5/7 elimination of luc+ cells 2/7 partial response
6/6 tumor progression
6/6 elimination of luc+ cells

exposure	color range min	color range max
5 min	17	250
5 min	16	600
30 sec	2 000	64 000

- No clinical symptoms of GvHD were observed in mice injected with $CD52^{+/-}$ TCRα-deficient T-cells. One mouse found dead at day 16 without any sign of GvHD.
- NSG mice treated with alemtuzumab combined with UCART cells or UCART alone display complete elimination of tumor cells at day 13

#6 T-cell engineering allows compatibility with concomitant drugs

dCK-deficient CAR T-cells resist to anticancer chemotherapeutic

(Purine nucleotide analogues Clafarabine, fludarabine, cytarabine)

Improvement of CAR T cell engraftment and Extension of therapeutic window (prevent HvG)

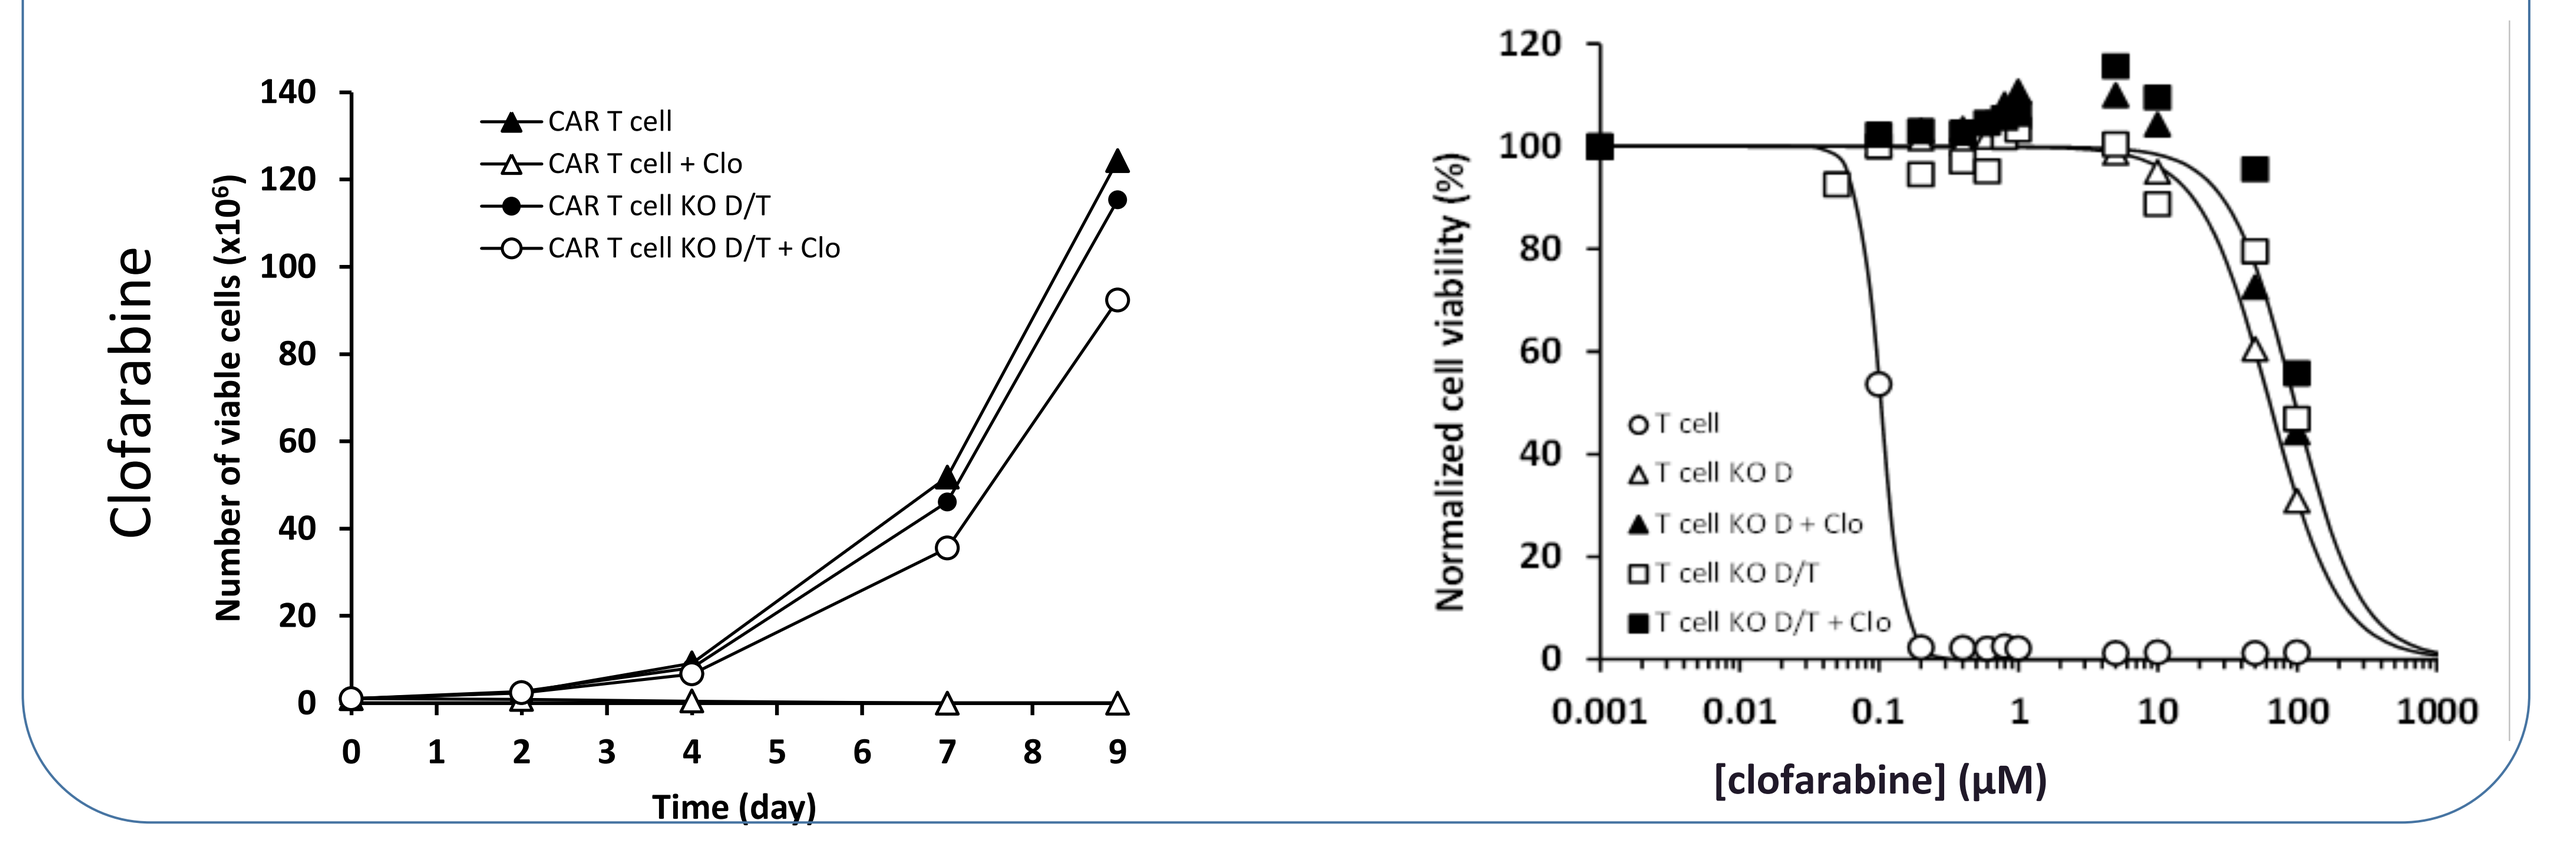
Clofarabine
Number of viable cells (x10⁶)
CAR T cell
CAR T cell + Clo
CAR T cell KO D/T
CAR T cell KO D/T + Clo
Time (day)
Normalized cell viability (%)
T cell
T cell KO D
T cell KO D + Clo
T cell KO D/T
T cell KO D/T + Clo
[clofarabine] (μM)

dCK-deficient CAR T-cells maintain anti-tumor activity in the presence of PNA

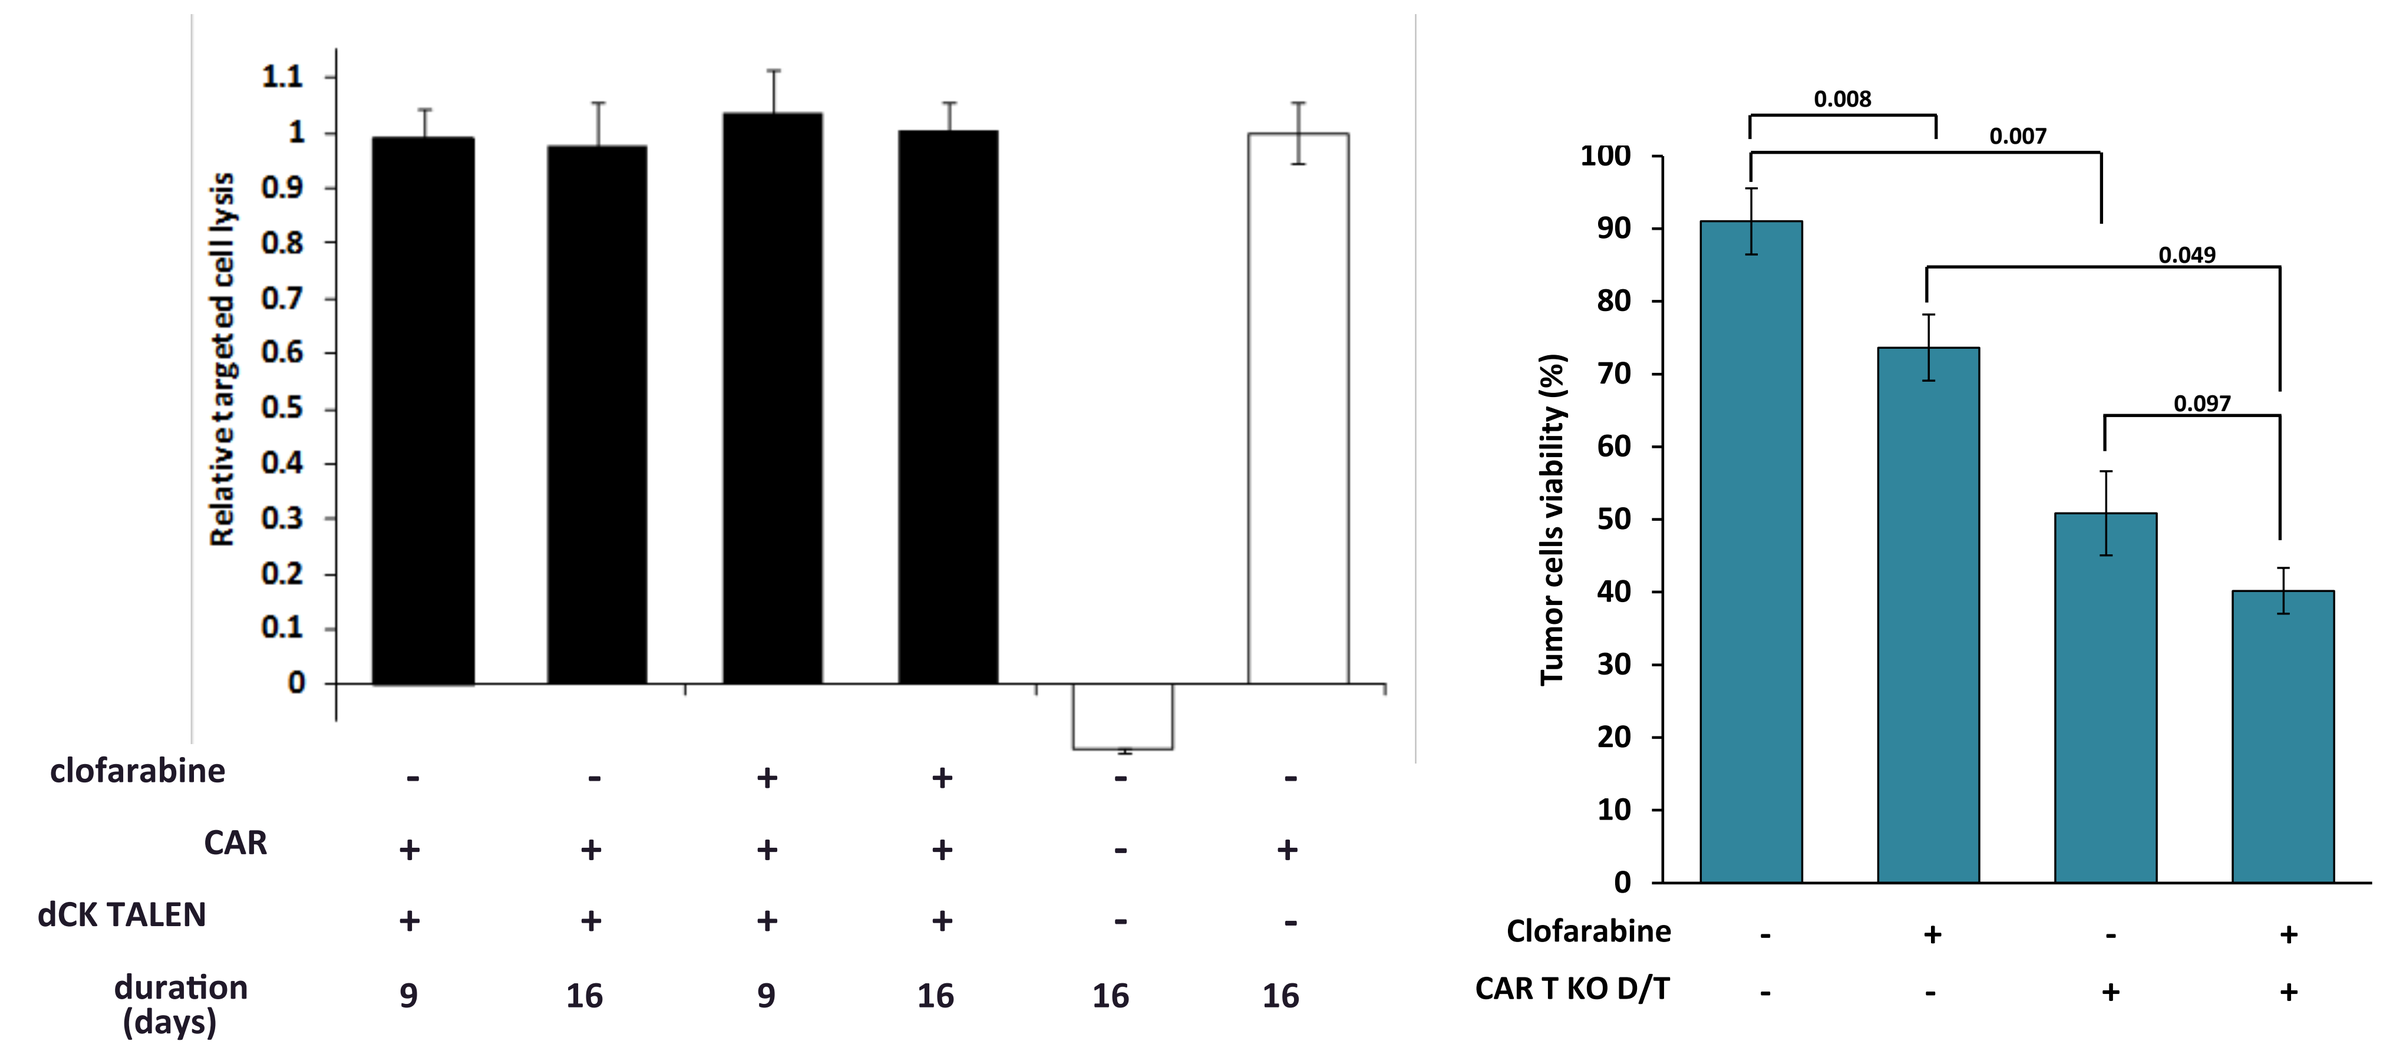
Relative targeted cell lysis
clofarabine
CAR
dCK TALEN
duration (days)
Tumor cells viability (%)
0.008
0.007
0.049
0.097
Clofarabine
CAR T KO D/T

Note: TCR/dCK double KO efficiency up to 75 %

#7 Gene editing for improved antitumor CAR T-cells

Glucocorticoid Receptor-deficient T-cells resist to glucocorticoid treatment

Inactivation of Glucocorticoid Receptor (GR) in primary T cells

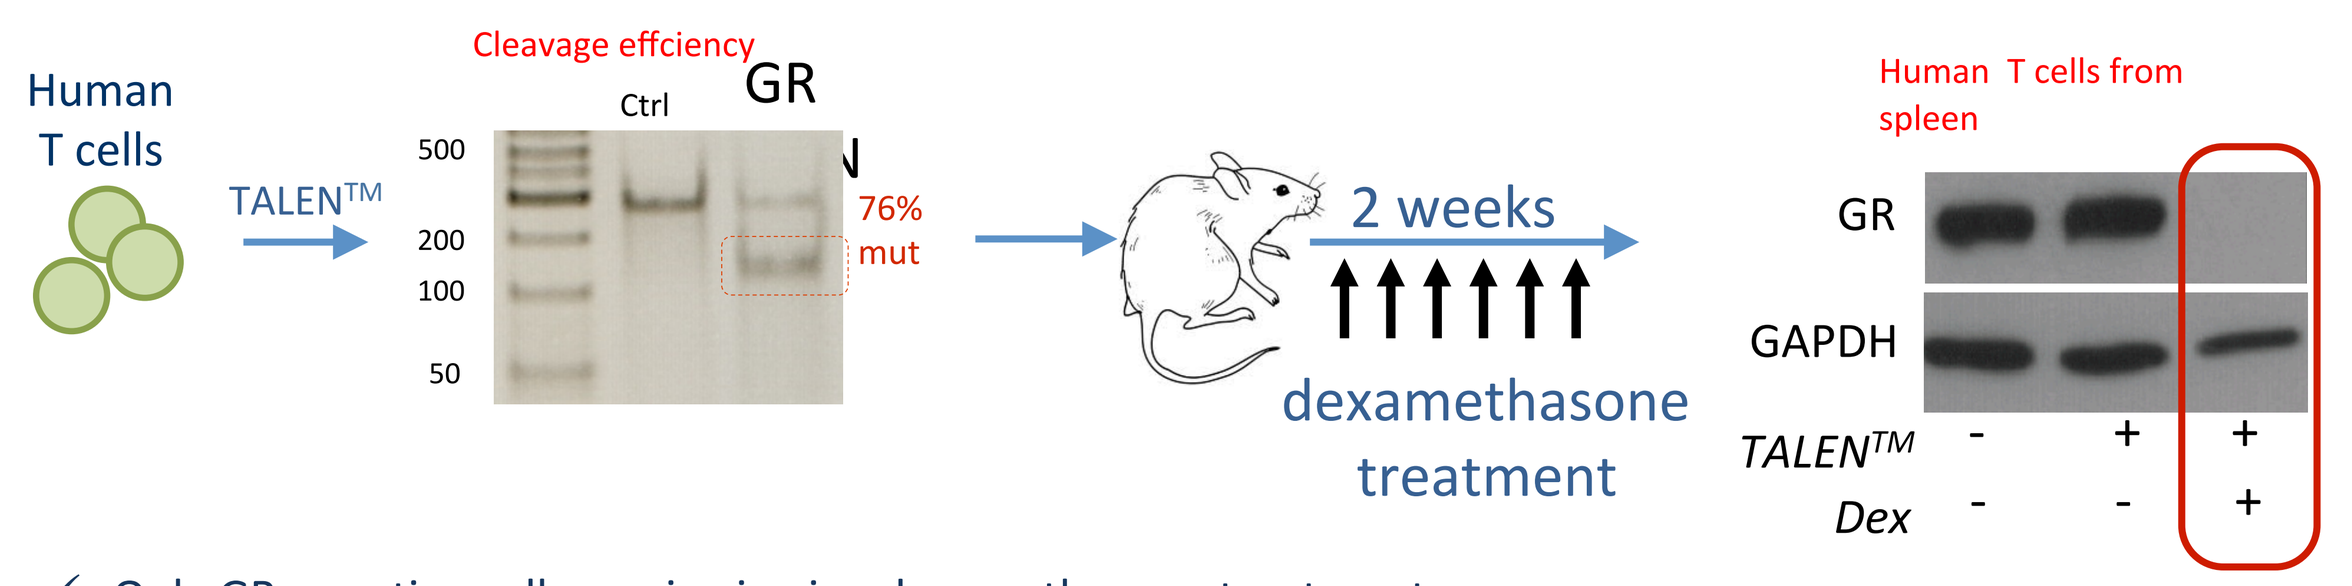
Human T cells
TALEN™
Cleavage effciency
Ctrl
GR
76% mut
2 weeks
dexamethasone treatment
Human T cells from spleen
GR
GAPDH
TALEN™
Dex

- ✓ Only GR-negative cells survive in vivo dexamethasone treatment.
- ✓ Glucocorticoid resistance opens therapeutic solutions : Prevention of allograft rejection, Control of inflammation in Glioblastoma, other brain tumors and metastasis, CMV reactivation after Graft vs Host disease

PD-1-deficient CAR T-cells have improved antitumor CAR T-cells

Inactivation of PD-1 in primary T cells

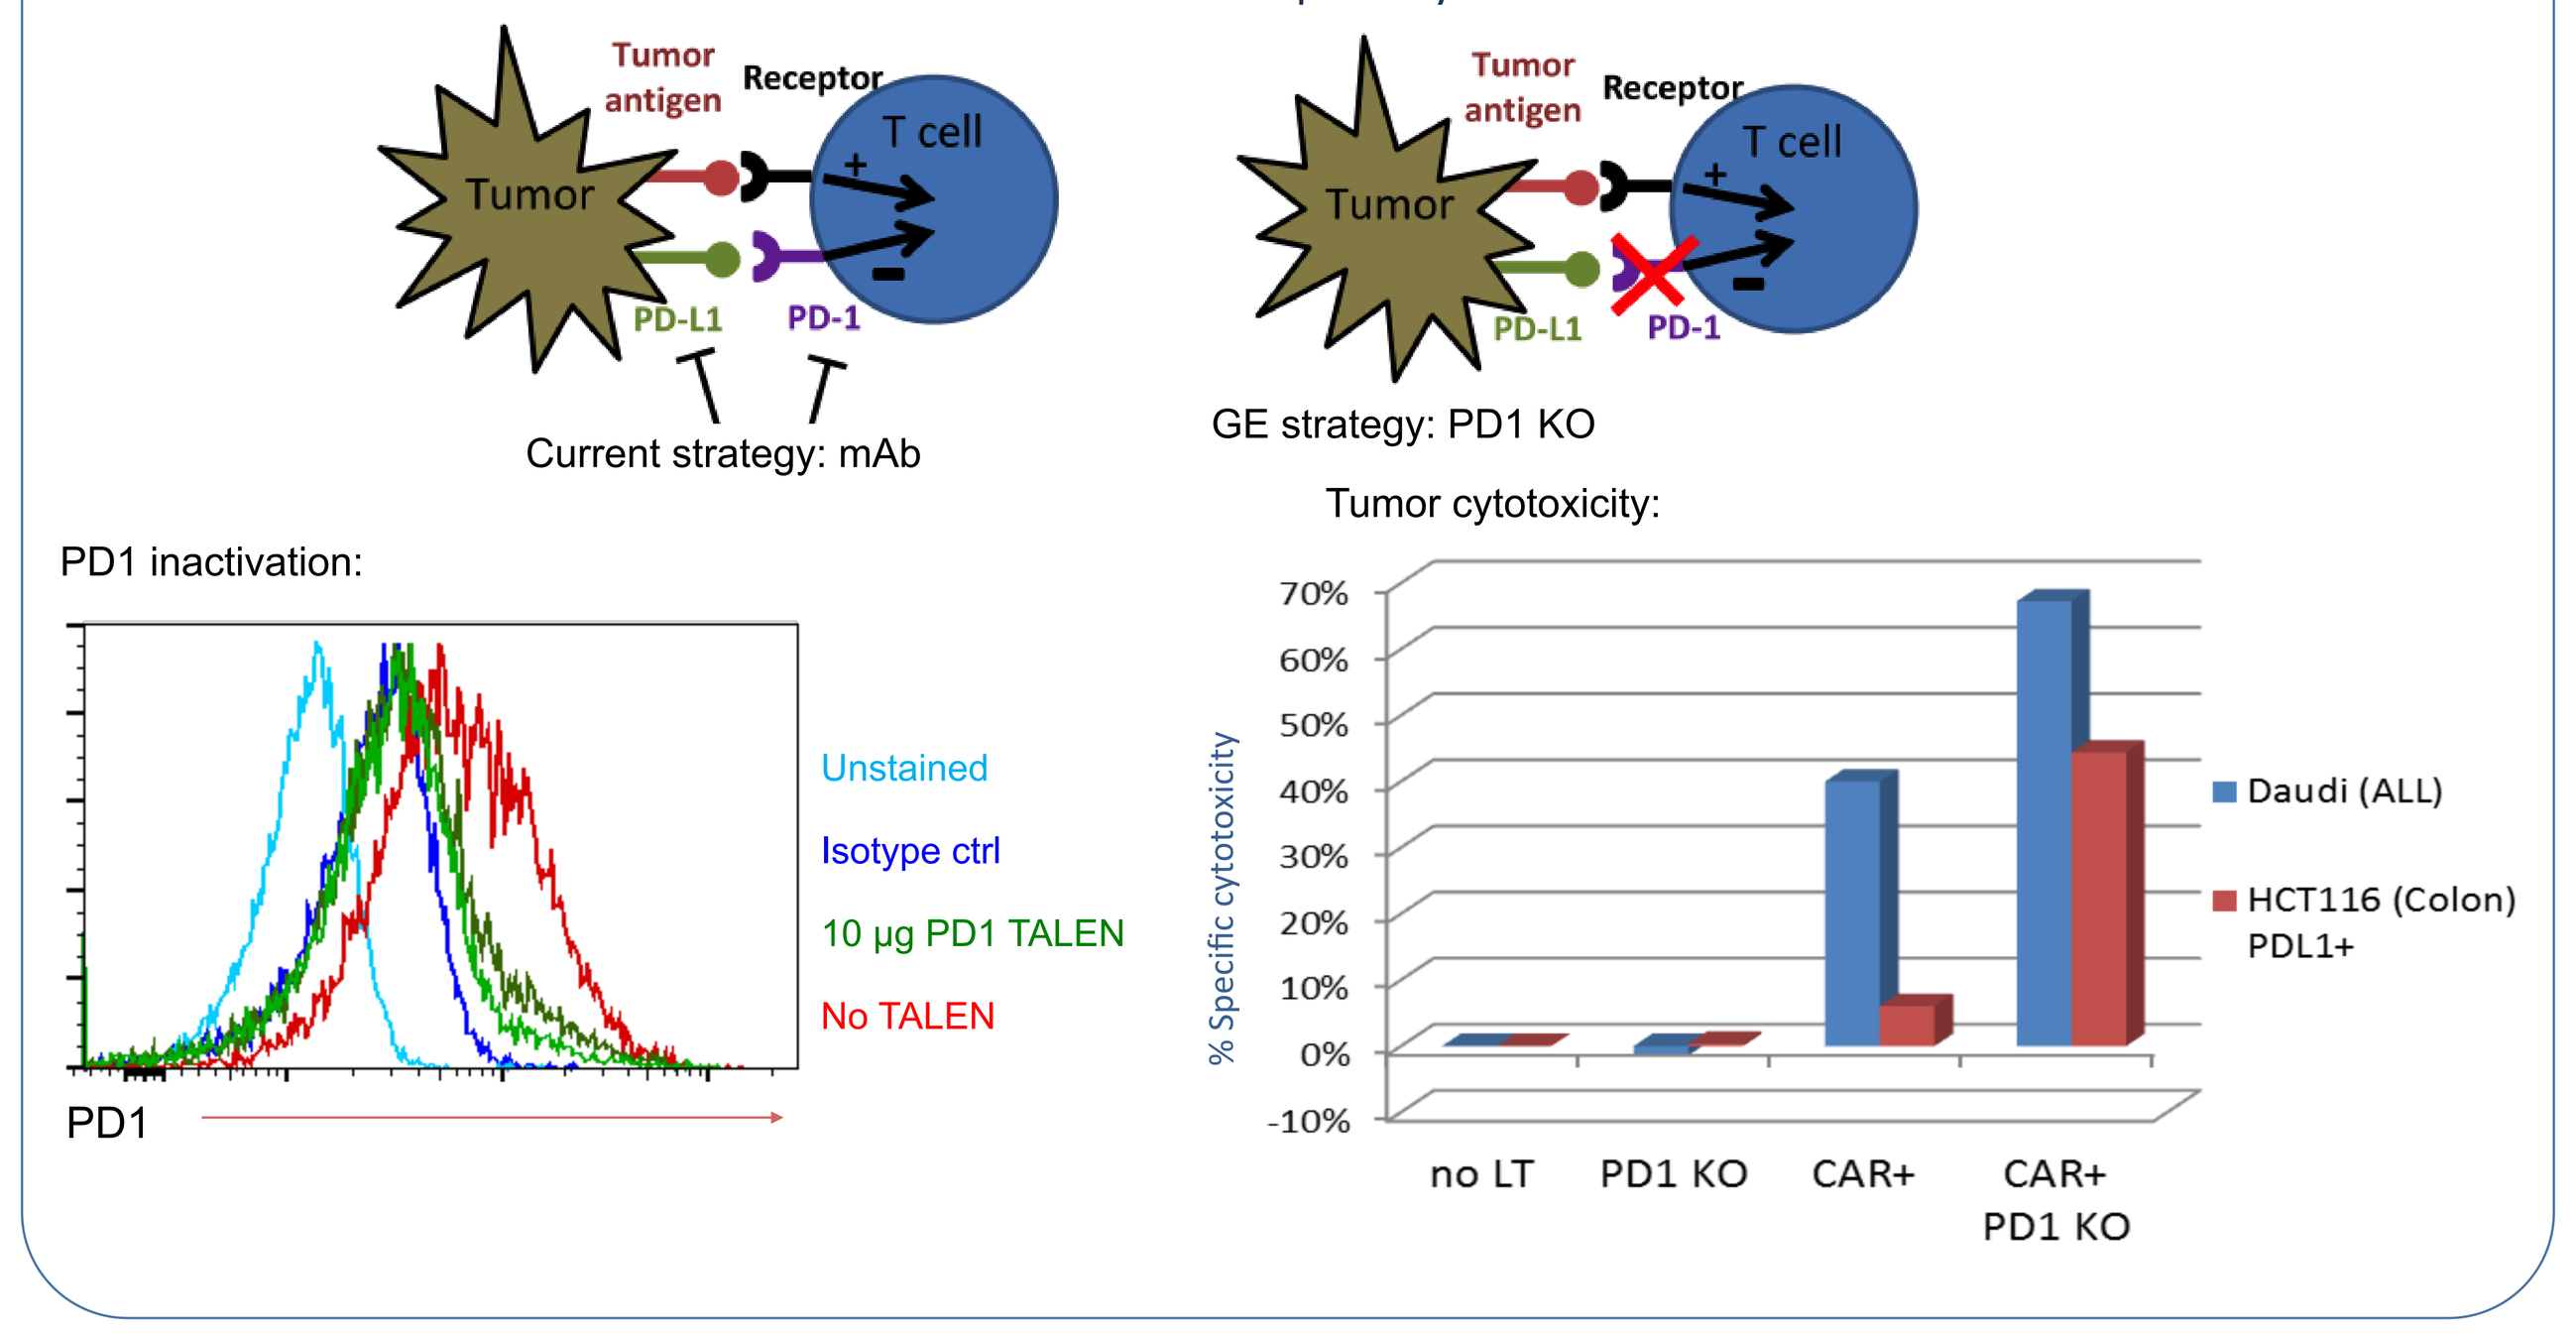
Tumor
Tumor antigen
Receptor
T cell
PD-L1
PD-1
Current strategy: mAb
GE strategy: PD1 KO
PD1 inactivation:
Unstained
Isotype ctrl
10 µg PD1 TALEN
No TALEN
PD1
Tumor cytotoxicity:
% Specific cytotoxicity
Daudi (ALL)
HCT116 (Colon) PDL1+
no LT
PD1 KO
CAR+
CAR+ PD1 KO

#8 Conclusions

- Autologous CAR T-cell therapy has been very successful to cure acute Lymphoblastic Leukemia (75% CR at 12month)

- Gene editing via TALEN have the power to enhance the potential of T-cells therapy

- We have developed highly efficient TALEN platform for T-cell engineering

- TALEN modified T-cells expressing an exogenous CAR have *In vitro & in vivo* antitumor reactivity comparable to "wt" CAR T-cells

- Gene editing via TALEN have the power to alleviate some of the challenges of adoptive T-cell immunotherapy

 - ❑ Endogenous T-cell receptor → prevent GvHD → off the shelf product
 - ❑ Cellular target for a lymphodepleting treatment → proliferation and activity of T- cells in presence of immunosuppressive drugs
 - ❑ Traits can be built into T-cells to tackle tumor immune evasion and support efficient immune access to the tumor while avoiding systemic effect
 - ❑ Combined inactivation provides therapeutic solutions for indications where multiple mechanisms of escape are involved

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis SA and its business. Cellectis SA is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. This communication contains Cellectis' proprietary information. Talen® and Cellectis® are trademarks owned by the Cellectis Group.

Allogenic CAR T-cells Targeting CD123 for Adoptive Immunotherapy of Acute Myeloid Leukemia (AML)

Roman Galetto[1], Céline Lebuhotel[1], Patricia Françon[1], Agnès Gouble[1] and Julianne Smith[1]

[1] Cellectis SA, Paris, France

#1 Abstract

Chimeric antigen receptor (CAR)-redirected T-cells have given rise to long-term durable remissions and remarkable objective response rates in patients with refractory leukemia, raising hopes that a wider application of CAR technology may lead to a new paradigm in cancer treatment. A limitation of the current autologous approach is that CAR T-cells must be manufactured on a "per patient basis". We have developed a standardized platform for manufacturing T-cells from third-party healthy donors to generate allogeneic "off-the-shelf" engineered CAR+ T-cell–based frozen products. This platform utilizes Transcription Activator-Like Effector Nuclease (TALEN) gene editing technology to inactivate the TCRα constant (TRAC) gene, eliminating the potential for T-cells bearing alloreactive TCR's to mediate Graft versus Host Disease (GvHD). We have previously demonstrated that editing of the TRAC gene can be achieved at high frequencies, obtaining up to 80% of TCRα negative cells. This allows us to efficiently produce TCR-deficient T-cells that have been shown to no longer mediate alloreactivity in a xeno-GvHD mouse model. In addition to CAR expression, our T-cells are engineered to co-express the RQR8 gene as a safety feature, with the aim of rendering them sensitive to the monoclonal antibody rituximab.

In this work we present the adaptation of this allogeneic platform to the production of T cells targeting CD123, the transmembrane alpha chain of the interleukin-3 receptor, which is expressed in tumor cells from the majority of patients with Acute Myeloid Leukemia (AML). To identify an effective CAR targeting CD123, we have screened multiple antigen recognition domains in the context of several different CAR architectures to identify candidates displaying activity against cell lines expressing variable levels of the CD123 antigen. Furthermore, experiments in an AML mouse model using anti-CD123 CAR T cells demonstrate important anti-tumor activity in vivo. The ability to carry out large scale manufacturing of allogeneic, non alloreactive CD123 specific T cells from a single healthy donor will thus offer the possibility of an off-the-shelf treatment that would be immediately available for administration to a large number of AML patients.

#2 Allogenic approach targeting CD123 to treat AML

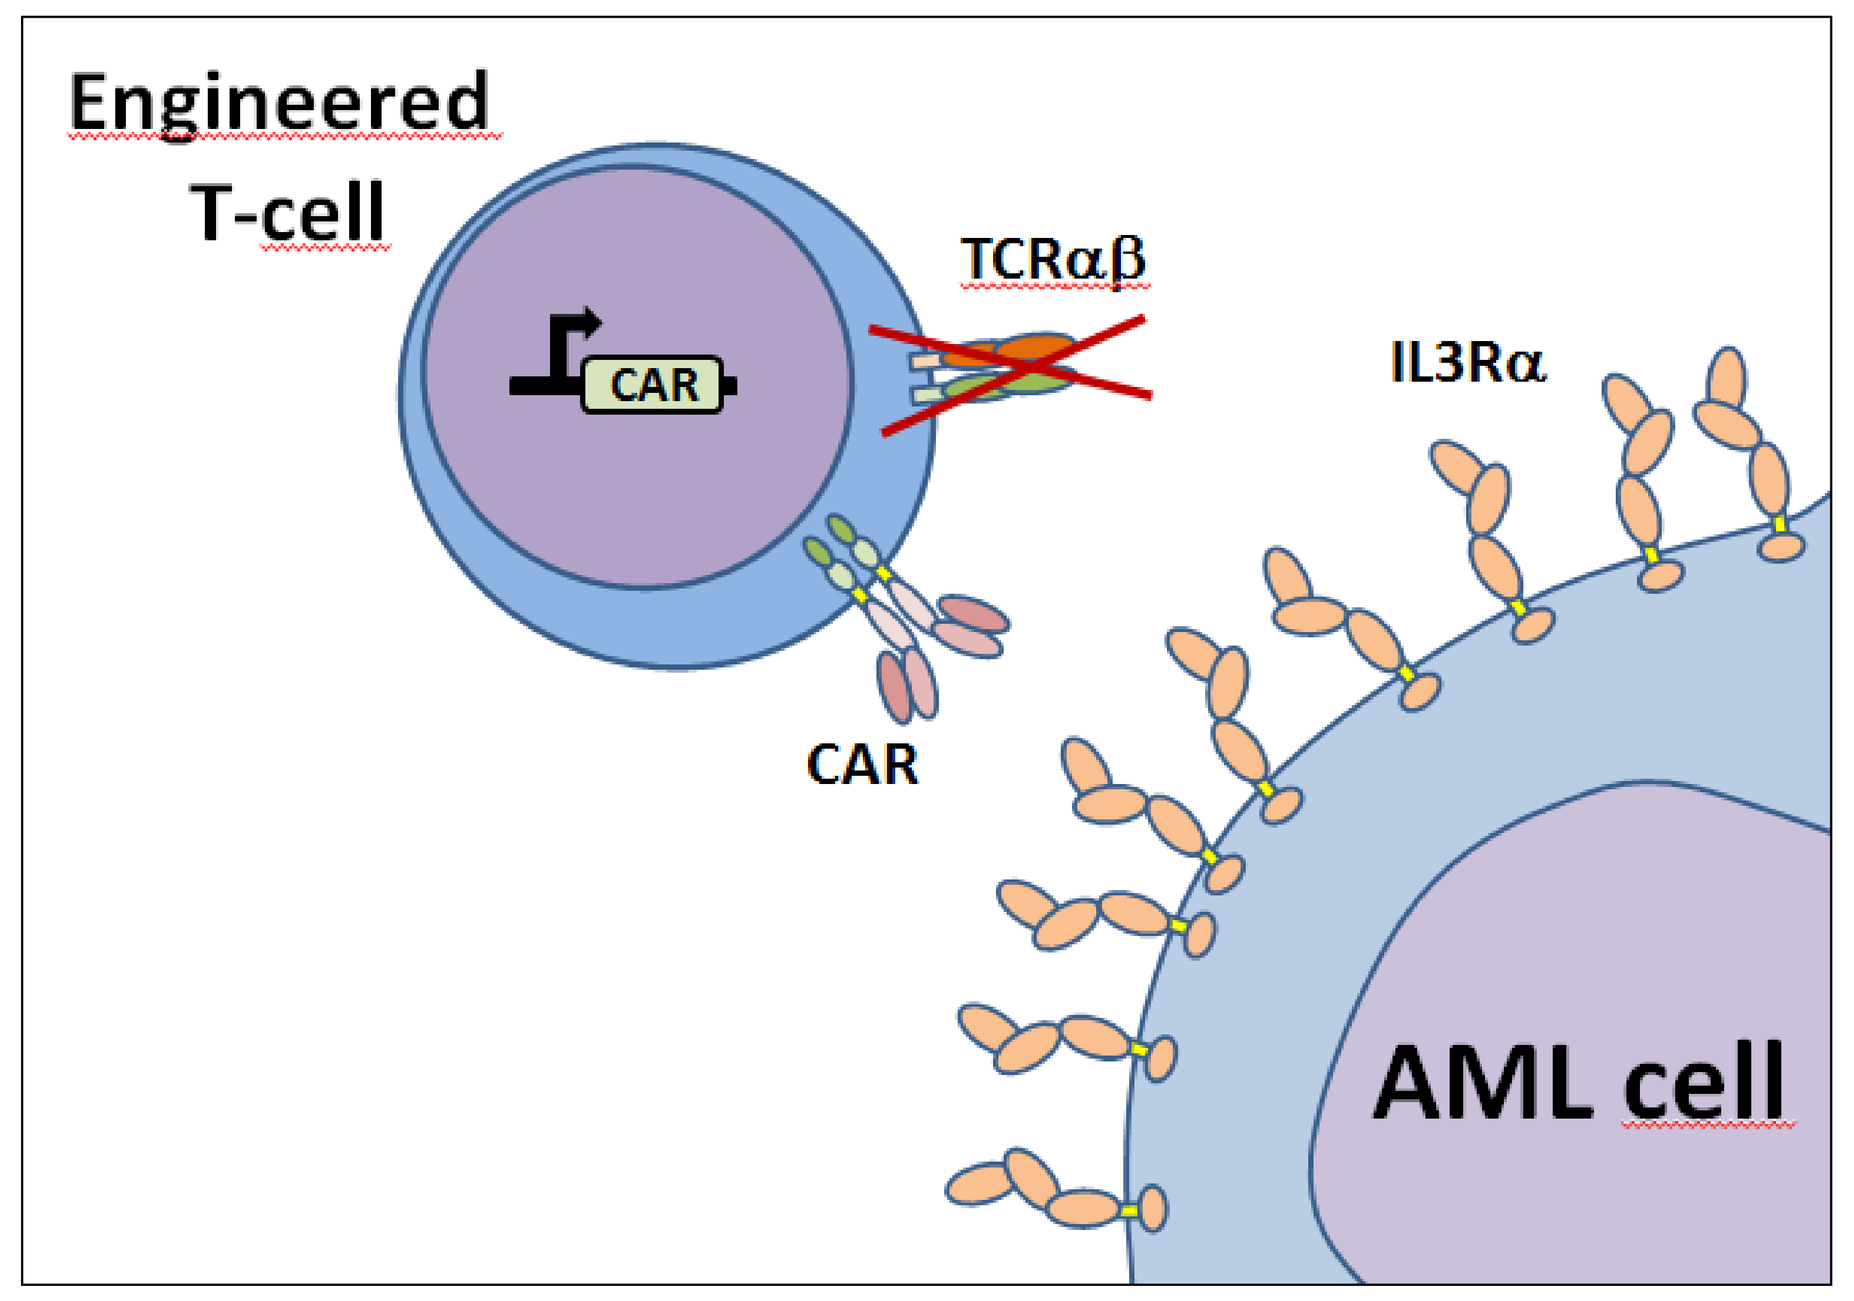

Engineered
T-cell
CAR
TCRαβ
IL3Rα
CAR
AML cell

One of the key barriers to the adoptive transfer of 3rd party CAR T-cells can be overcome via the application of TALEN gene editing technology, deletion of the TCR gene and thus the use of any donors' T-cells without the risk of GvHD.

#3 TCR alpha inactivation prevents Graft vs. Host disease

TALEN were designed to recognize and cleave the coding sequence of human TCRα gene

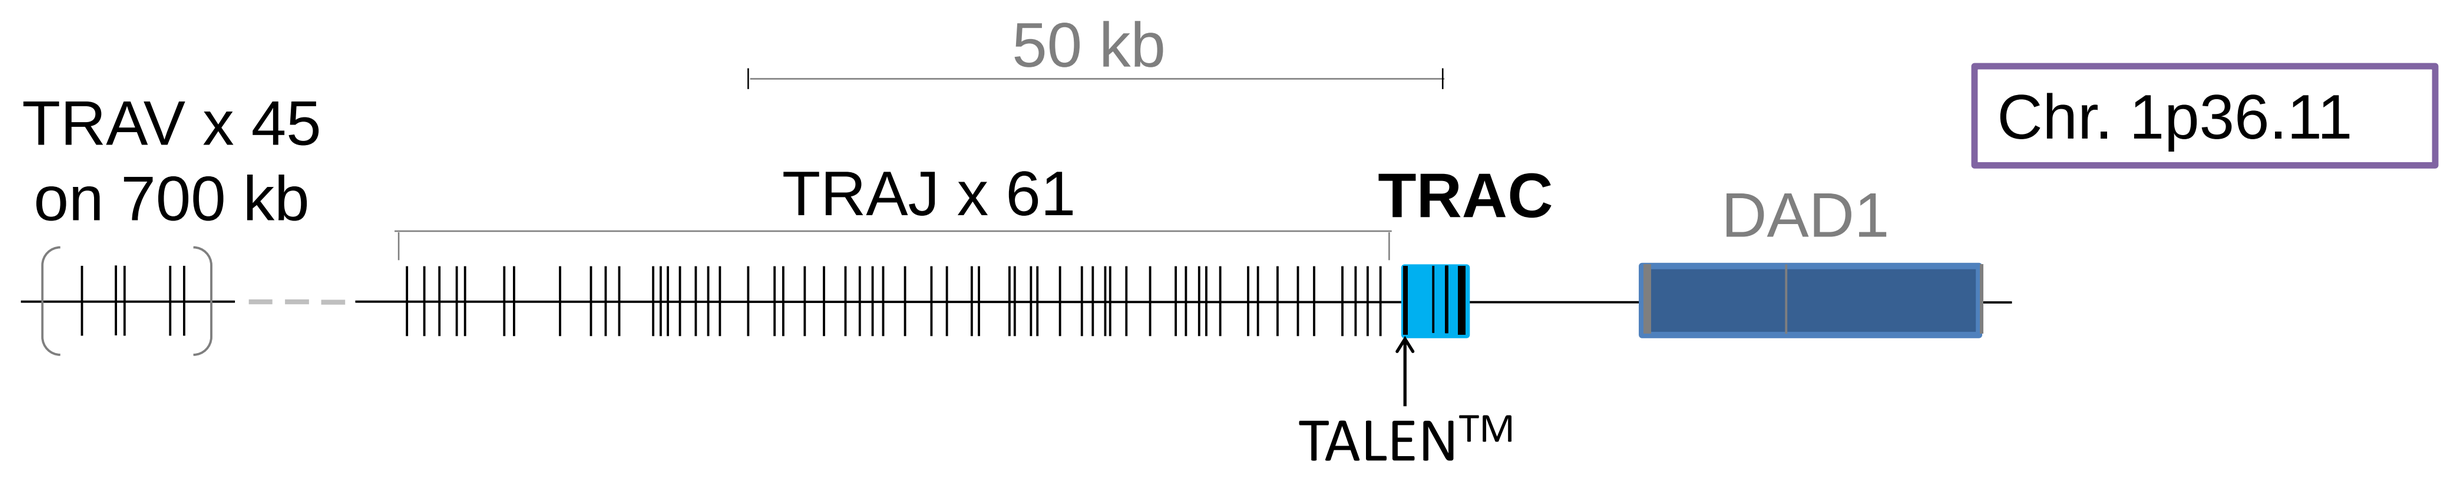

50 kb
Chr. 1p36.11
TRAV x 45
on 700 kb
TRAJ x 61
TRAC
DAD1
TALEN™

High levels of gene inactivation can be obtained in primary T cells by electrotransfer of TALEN mRNAs using a PulseAgile® electroporation technology.

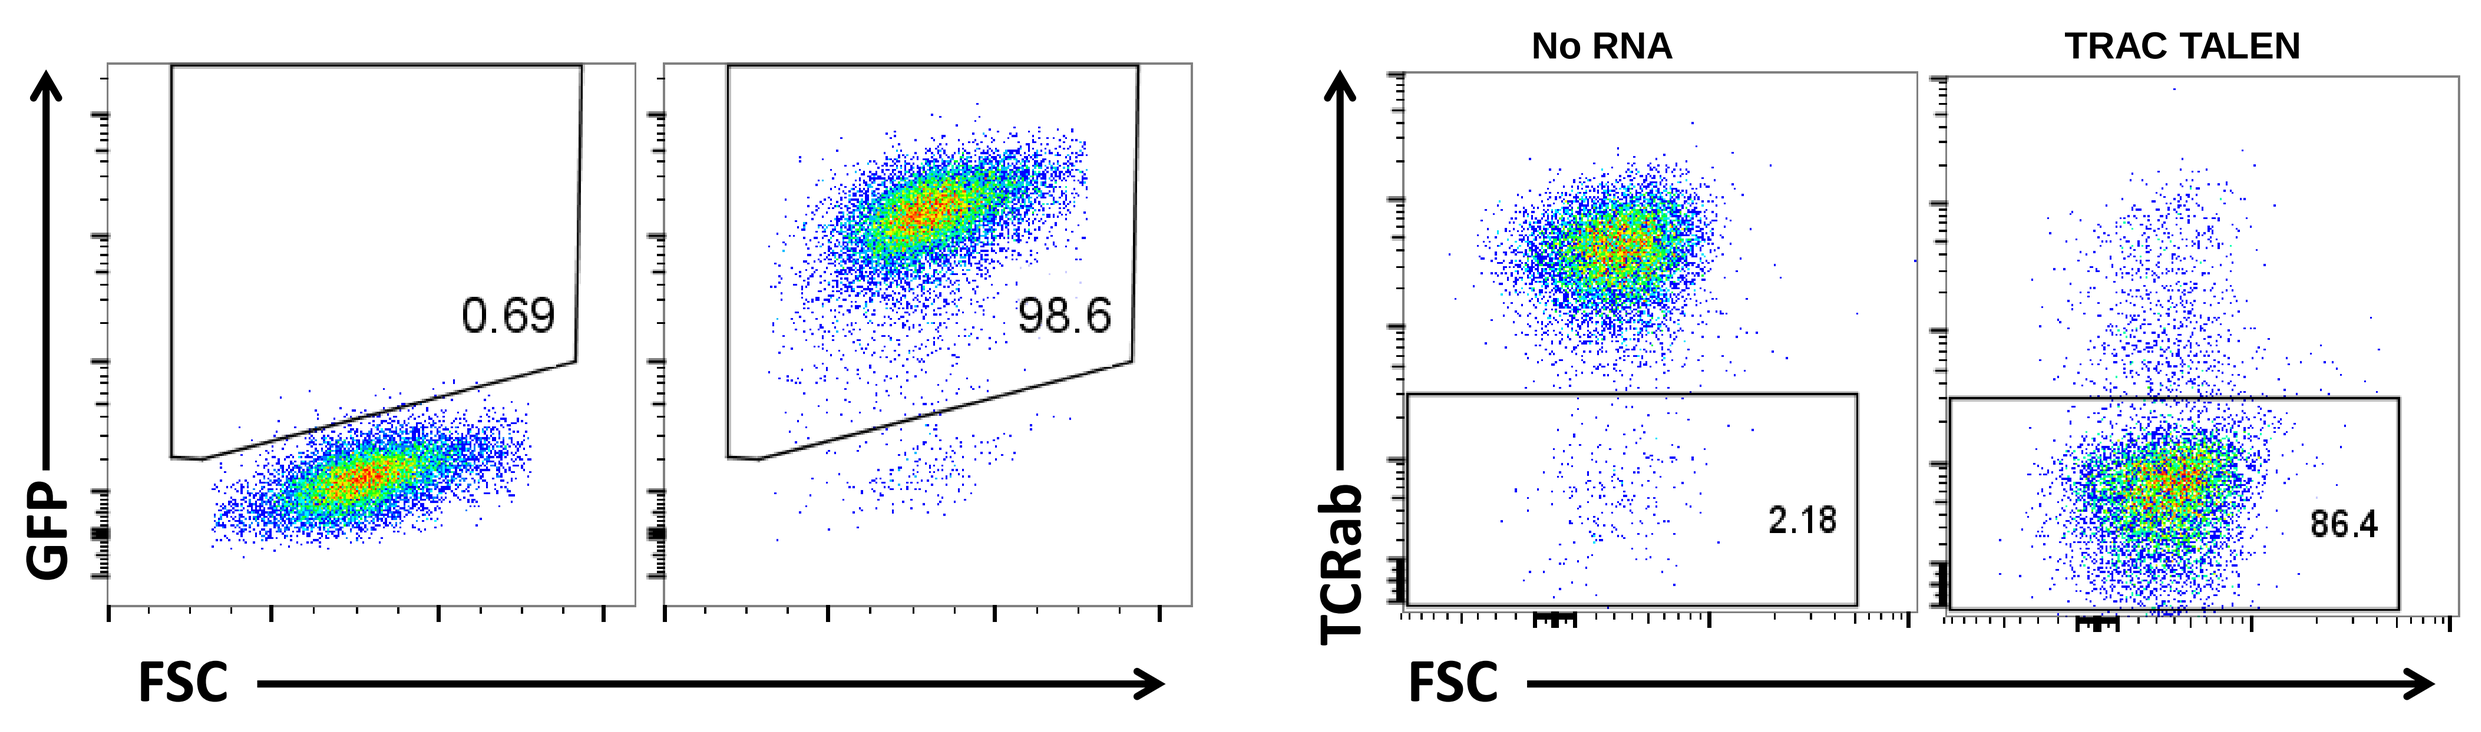

GFP
FSC
0.69
98.6
No RNA
TRAC TALEN
TCRab
2.18
86.4

Xenogeneic model of Graft vs. Host Disease: Irradiated NOG mice were injected with PBS (group 1), or 30x10^6 human T cells from the same donor. The cells were obtained from activated PBMCs after 3 day expansion (group 2), 15-day expansion (group 3), mock electroporation and 15-day expansion (group 4), or TRAC TALEN-electroporation, 15-day expansion and depletion of TCR+ cells (group 5).

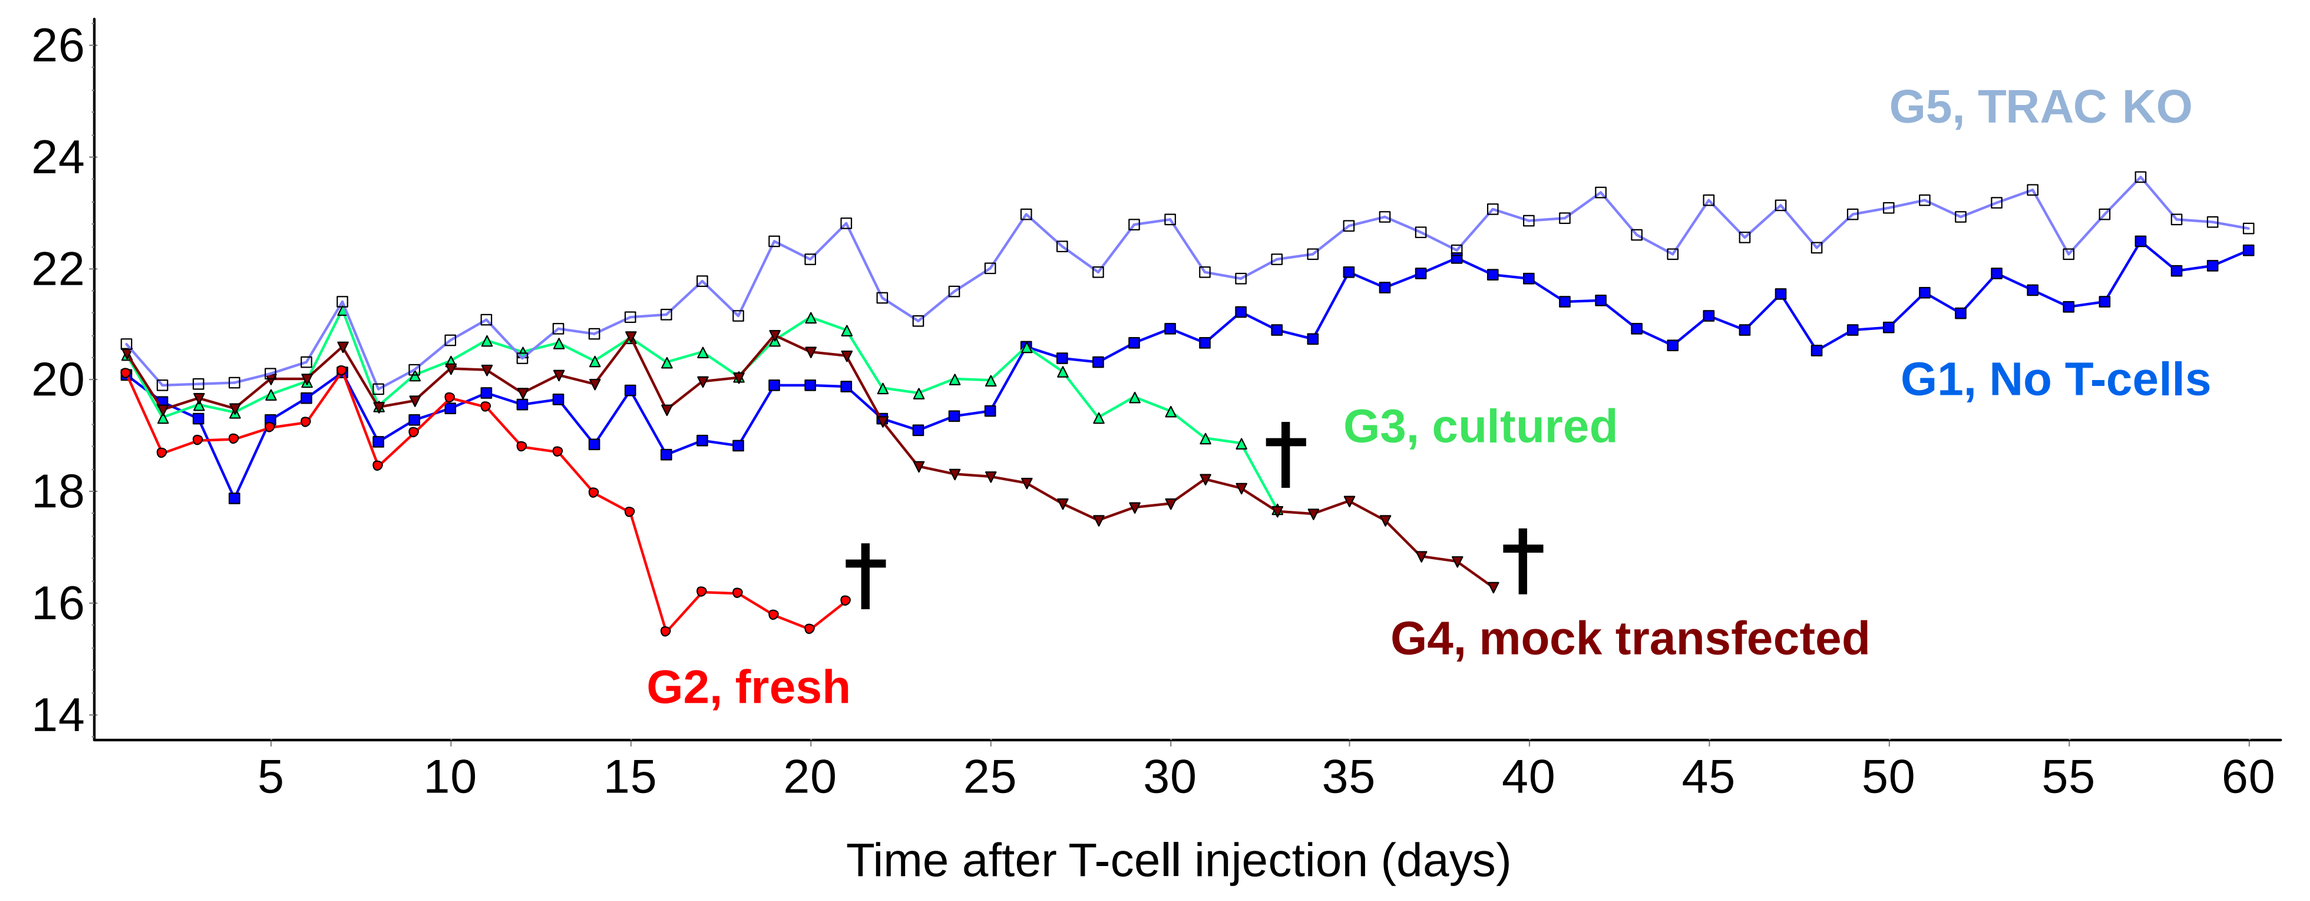

G5, TRAC KO
G1, No T-cells
G3, cultured
G4, mock transfected
G2, fresh
Time after T-cell injection (days)

No mice receiving TRAC TALEN-treated cells showed signs of GvHD, whereas all mice receiving non engineered cells developped GvHD and had to be sacrificed. Human cells survived at least 2 weeks in all groups.

#4 Generation of anti-CD123 Chimeric Antigen Receptors

An scFv targeting the extracellular domain of the human CD123 antigen was used to construct three chimeric antigen receptors, each containing hinge sequences of different lengths.

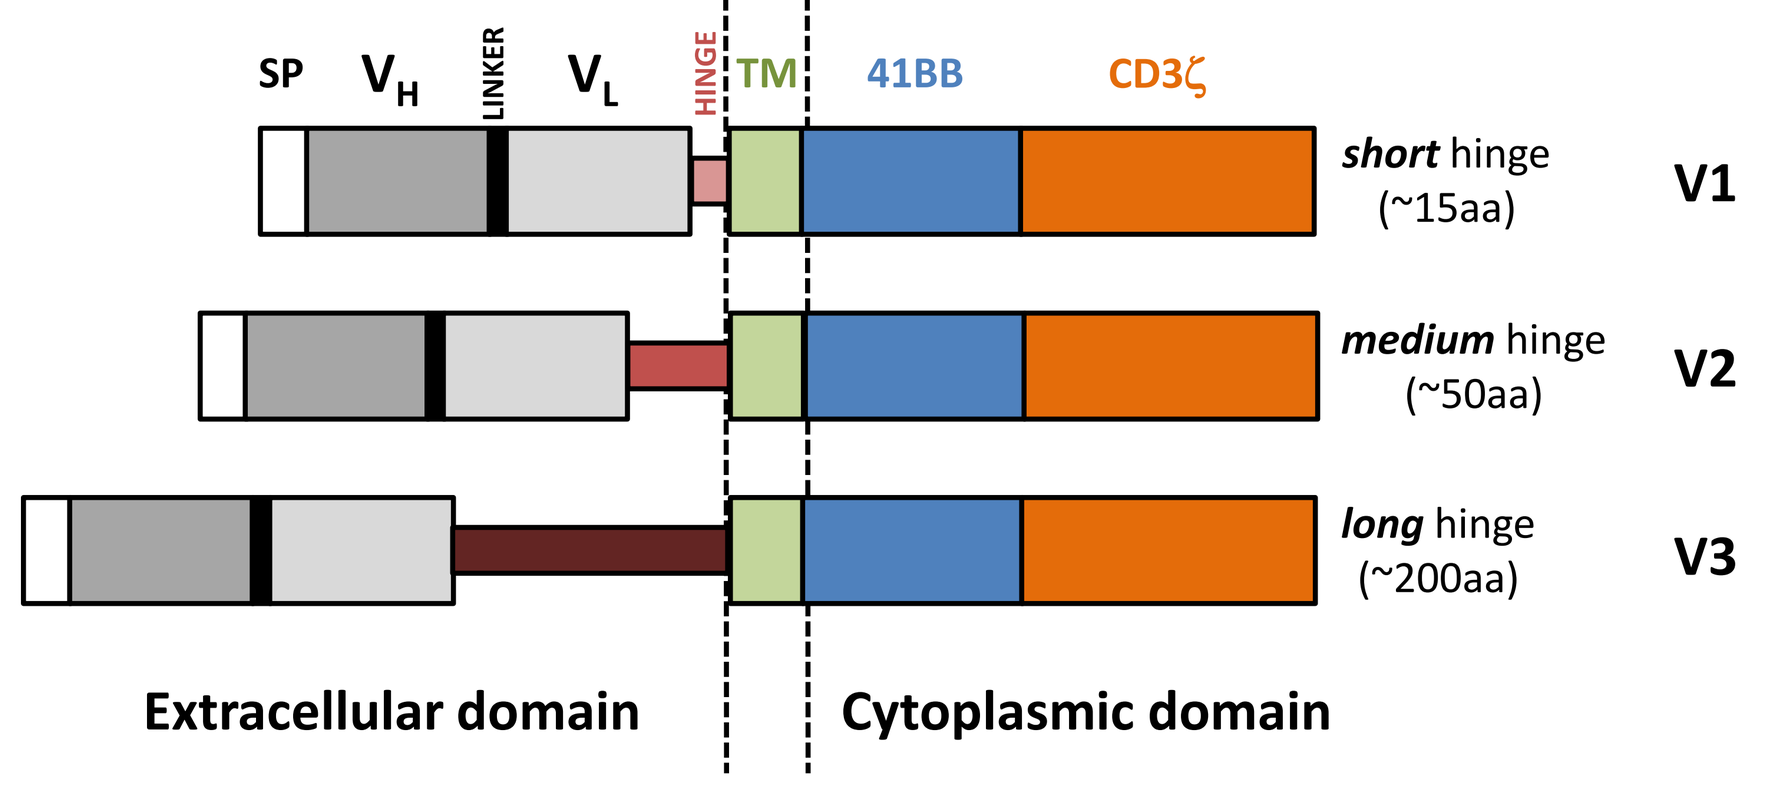

SP
VH
LINKER
VL
HINGE
TM
41BB
CD3ζ
short hinge (~15aa)
V1
medium hinge (~50aa)
V2
long hinge (~200aa)
V3
Extracellular domain
Cytoplasmic domain

#5 CD123 CAR T-cell in vitro activity

T-cells stably expressing the three CAR constructs and a benchmark CAR (32716) were generated by lentiviral transduction and their activity was assessed 10 days after transduction.

DEGRANULATION ASSAY

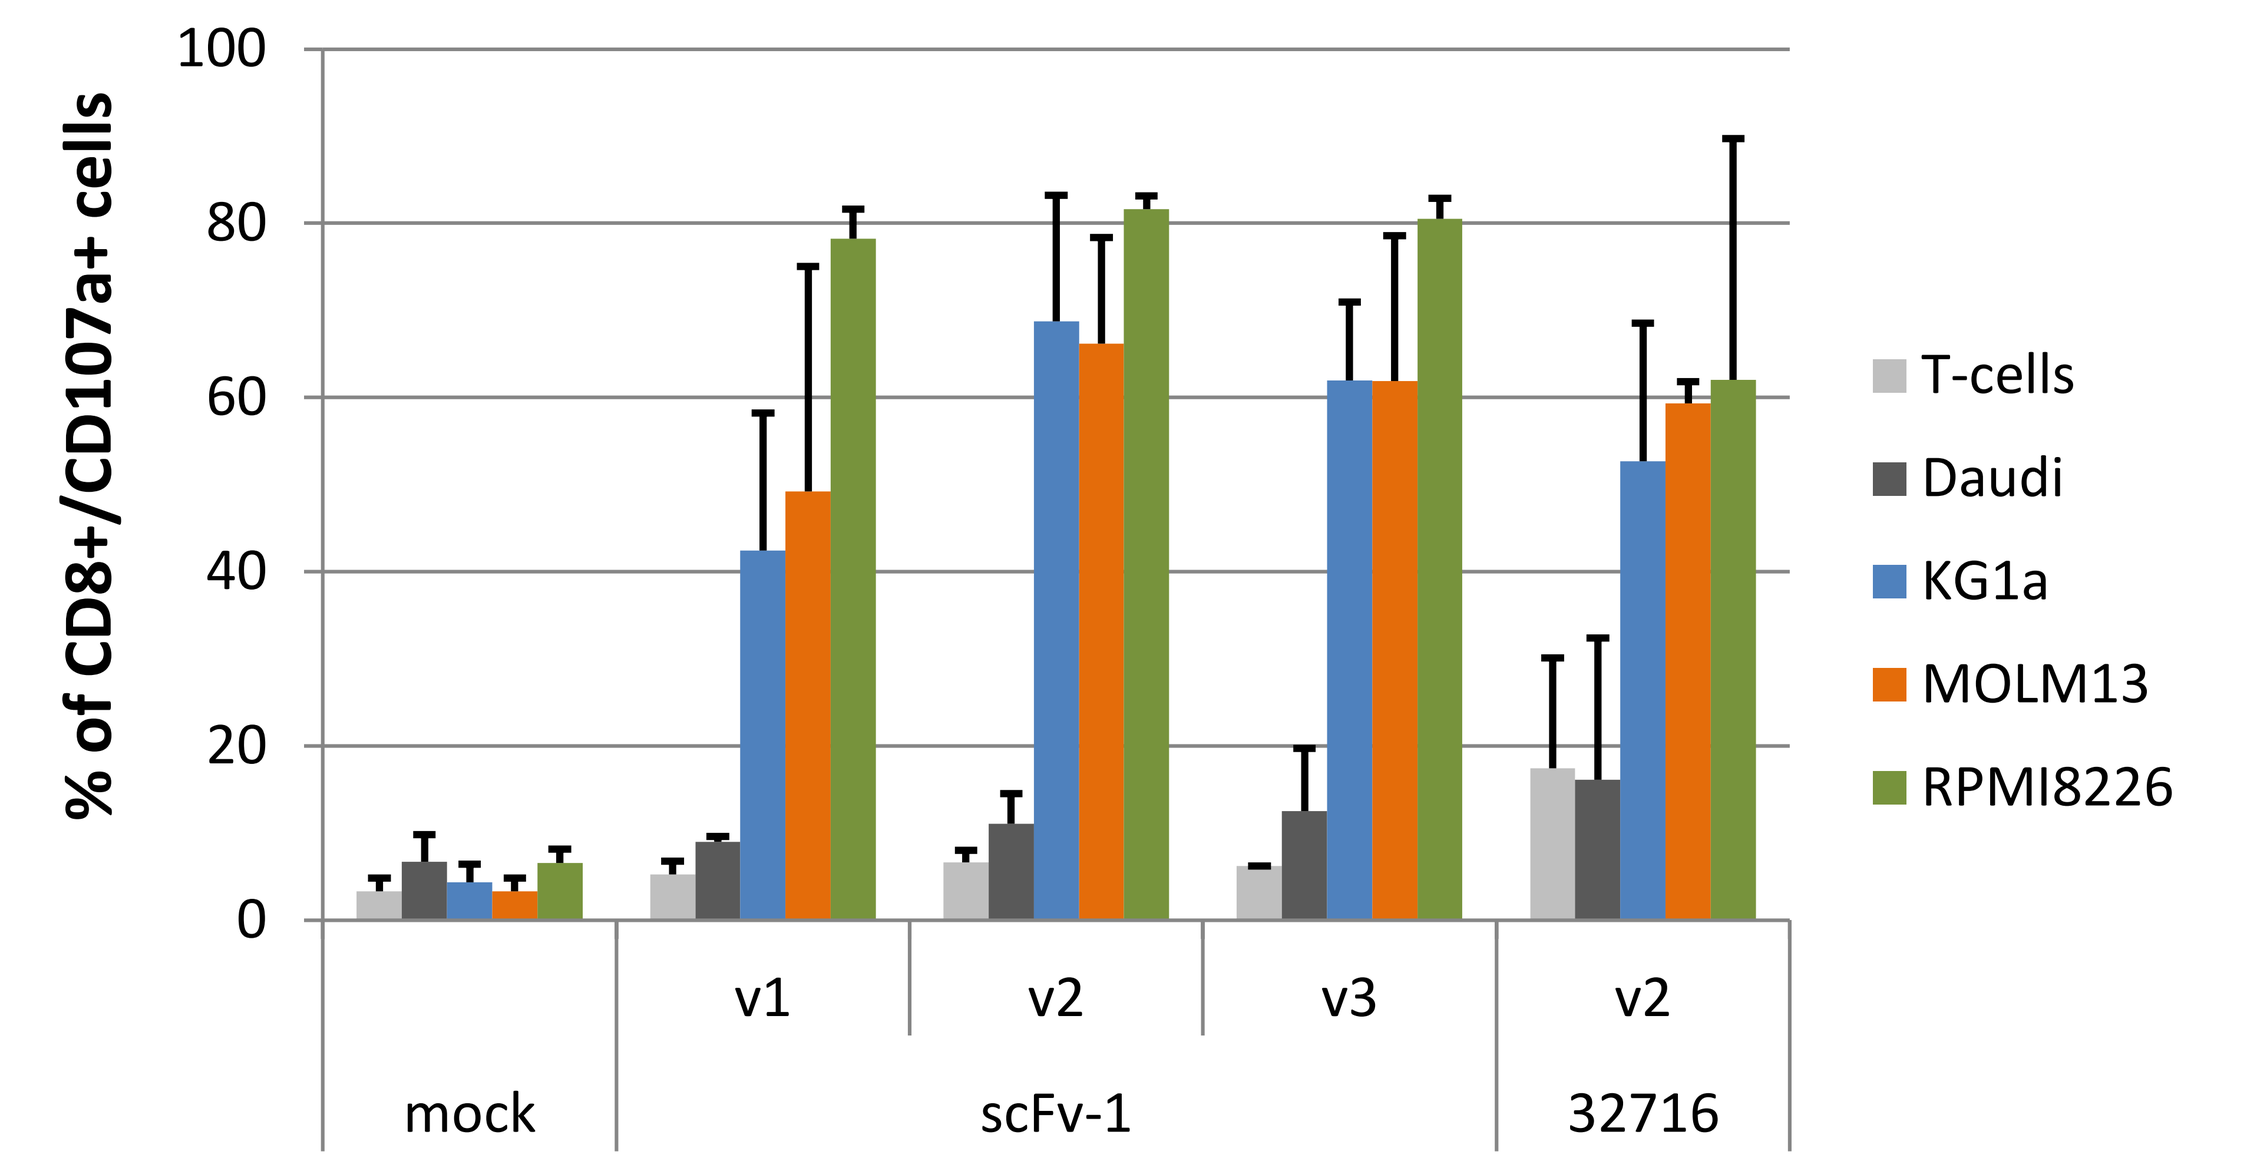

% of CD8+/CD107a+ cells
T-cells
Daudi
KG1a
MOLM13
RPMI8226
mock
v1
v2
v3
v2
scFv-1
32716

CAR+ T-cells were co-cultured with CD123+ target cells (1:1). Degranulation activity was measured by staining with CD8 and CD107a antibodies and analyzed by flow cytometry. Daudi cells were used as a negative control.

IFNgamma RELEASE ASSAY

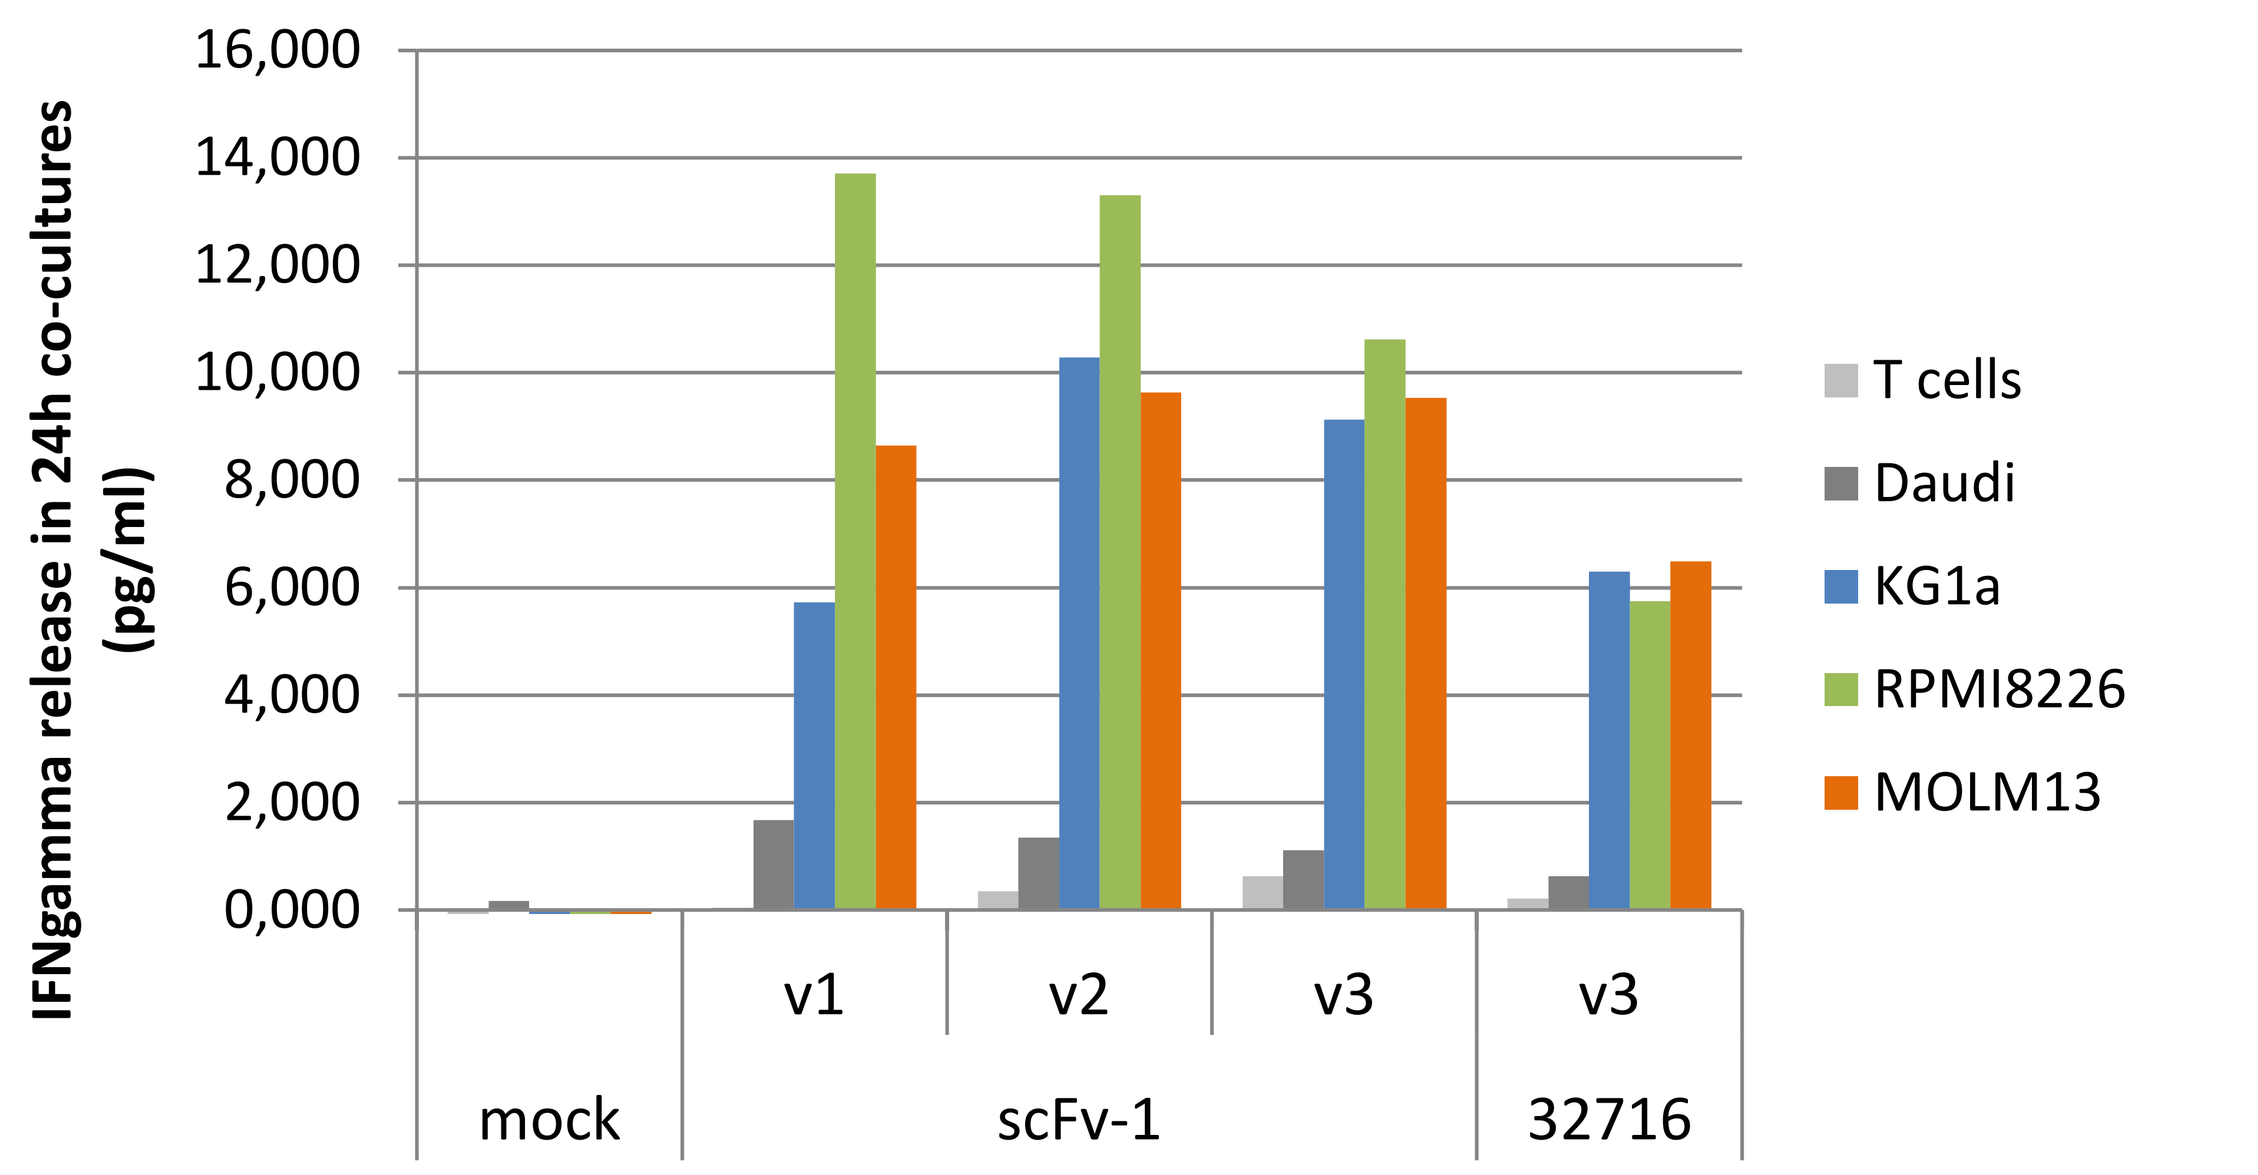

IFNgamma release in 24h co-cultures (pg/ml)
T cells
Daudi
KG1a
RPMI8226
MOLM13
mock
v1
v2
v3
v3
scFv-1
32716

CAR+ T-cells were co-cultured with CD123(+) target cells (1:1). The amount of IFNg released in the culture supernatants was determined by ELISA.

CYTOTOXICITY ASSAY

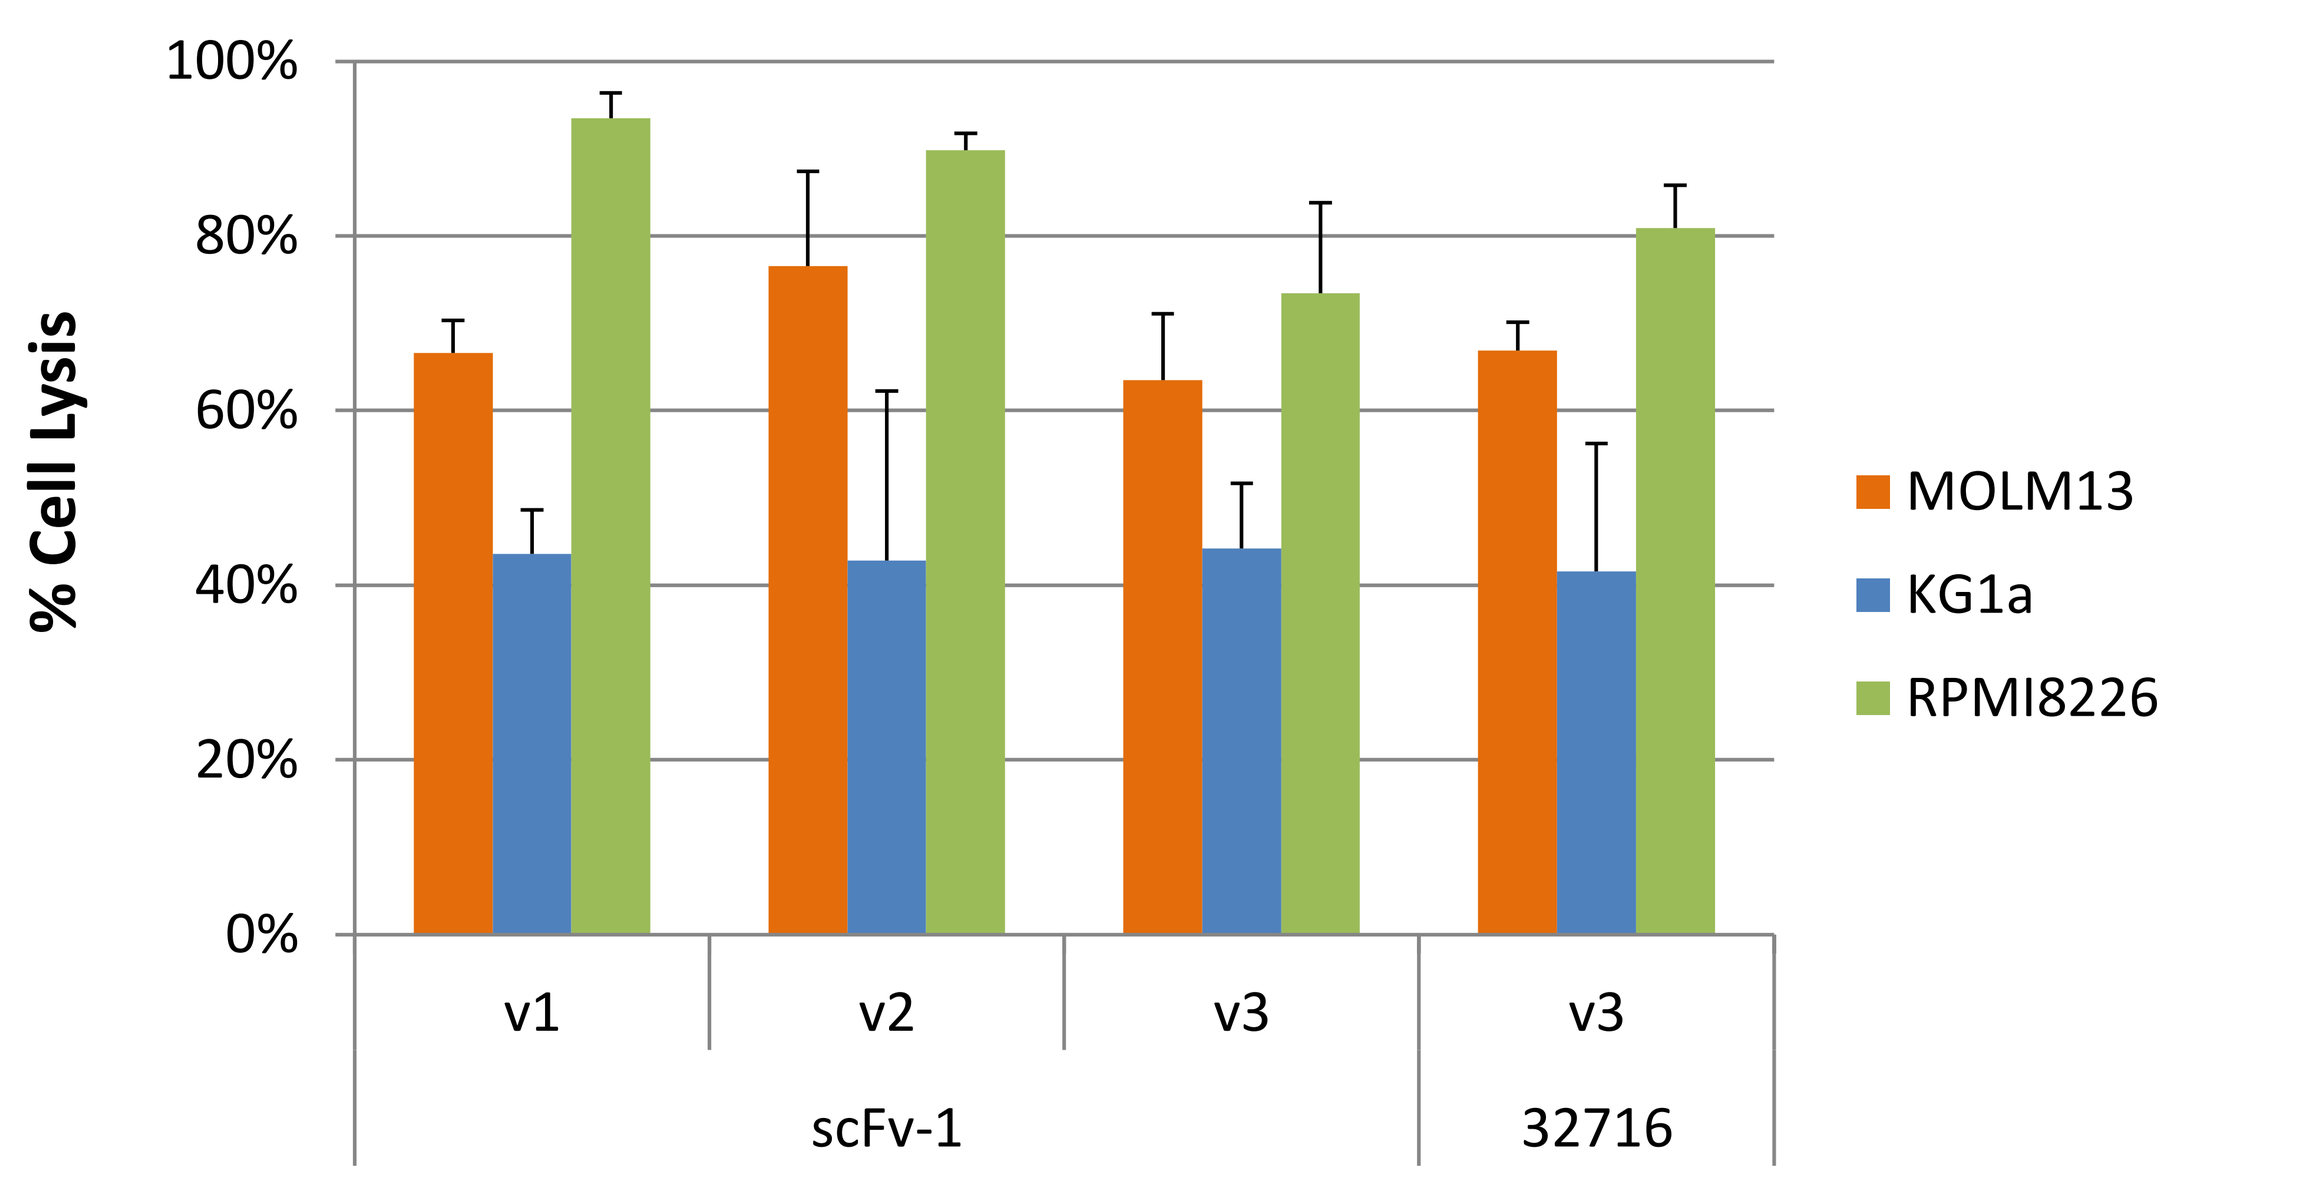

% Cell Lysis
MOLM13
KG1a
RPMI8226
v1
v2
v3
v3
scFv-1
32716

CAR+ T-cells were co-cultured with CD123(+) target cells and CD123(-) cells (10:1:1 ratio). The viability of each cell line -CD123(+) and CD123(-)- was determined after 4h of co-culture to determine the % of specific cell lysis.

#6 CD123 CAR T cell in vivo anti-tumor activity

All three CAR constructs were evaluated for vivo anti-tumor activity: 5×10^6 CAR-positive T-cells were intravenously injected in NSG mice 7 days after tumor cell injection (2.5×10^5 Molm13-ffluc cells).

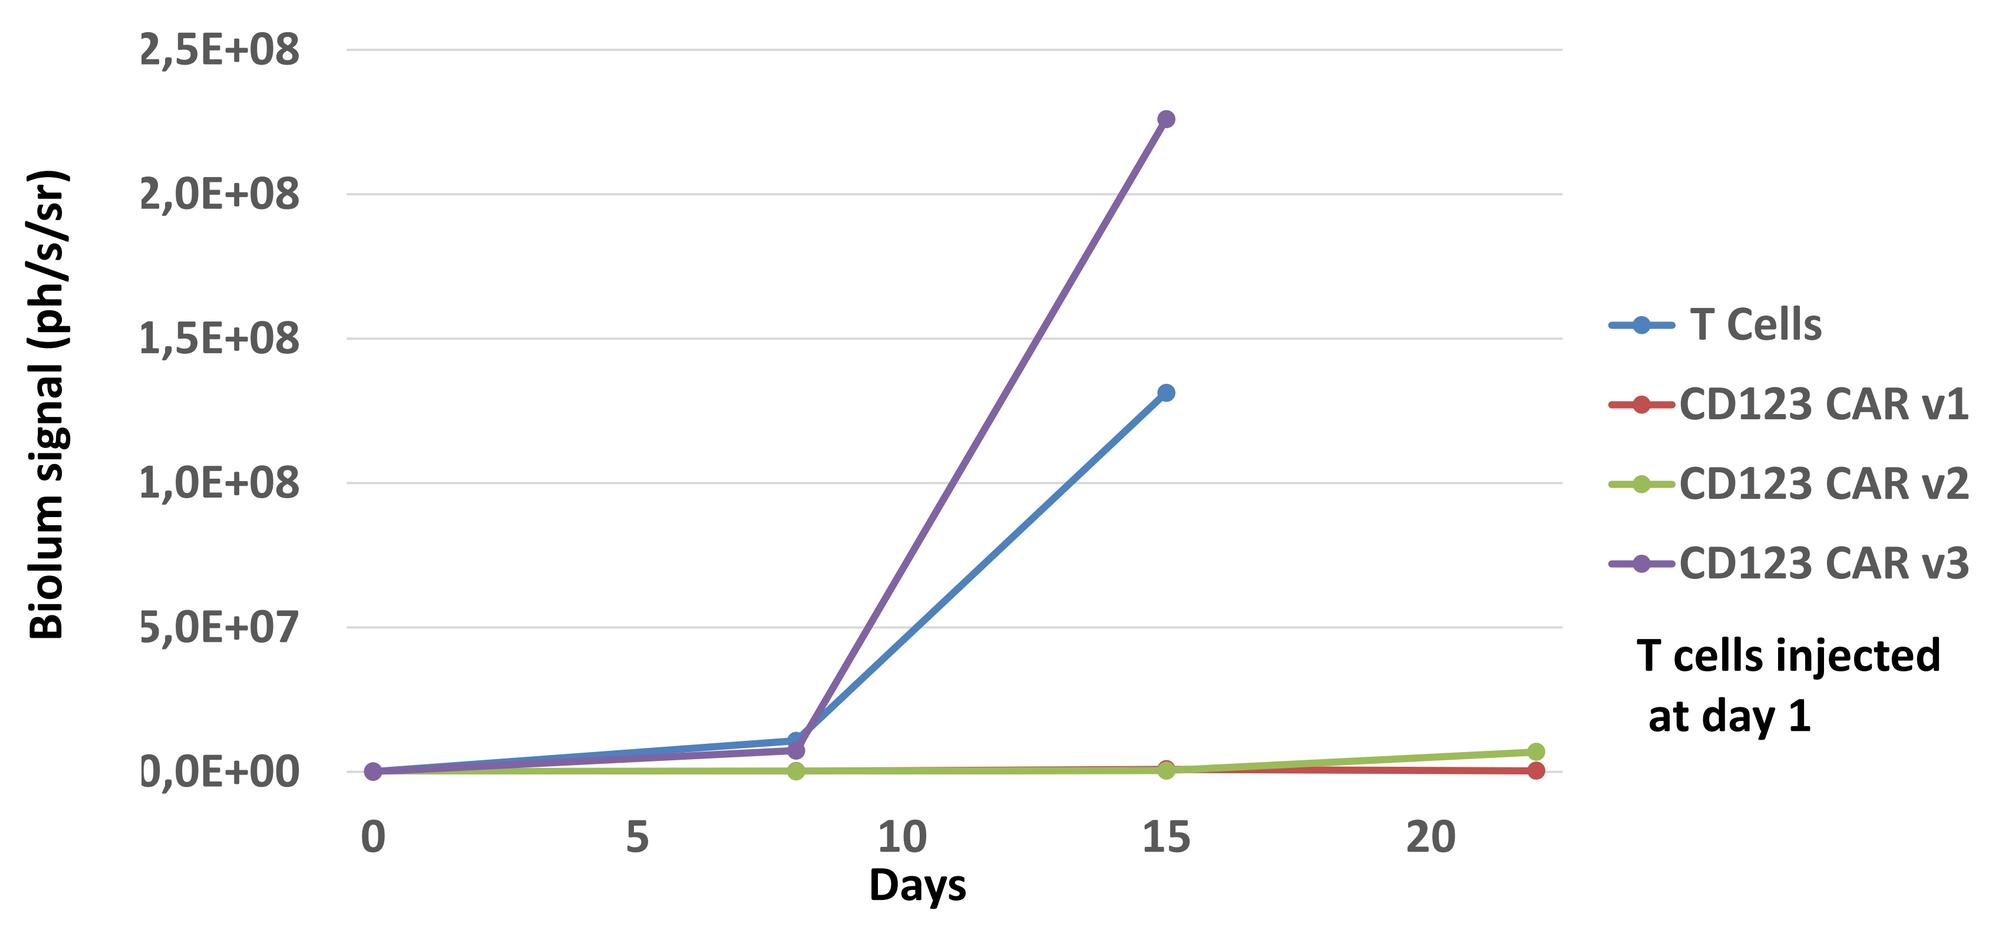

Biolum signal (ph/s/sr)
Days
T Cells
CD123 CAR v1
CD123 CAR v2
CD123 CAR v3
T cells injected at day 1

The CD123 CAR v1 and v2 display a strong anti-tumor effect while the CD123 CAR v3, despite significant in vitro activity, did not display any anti-tumor activity in vivo.

The anti-tumor activity of the v2 CAR was further characterized in a long term anti-tumor study: several doses of CAR-positive T-cells (1×10^6, 5×10^6 and 10×10^6 cells) were intravenously injected in NSG mice 7 days after tumor cell injection (2.5×10^5 Molm13-ffluc cells) and mice were followed for up to 60 days. The T cells were injected at Day 1 in the figures below.

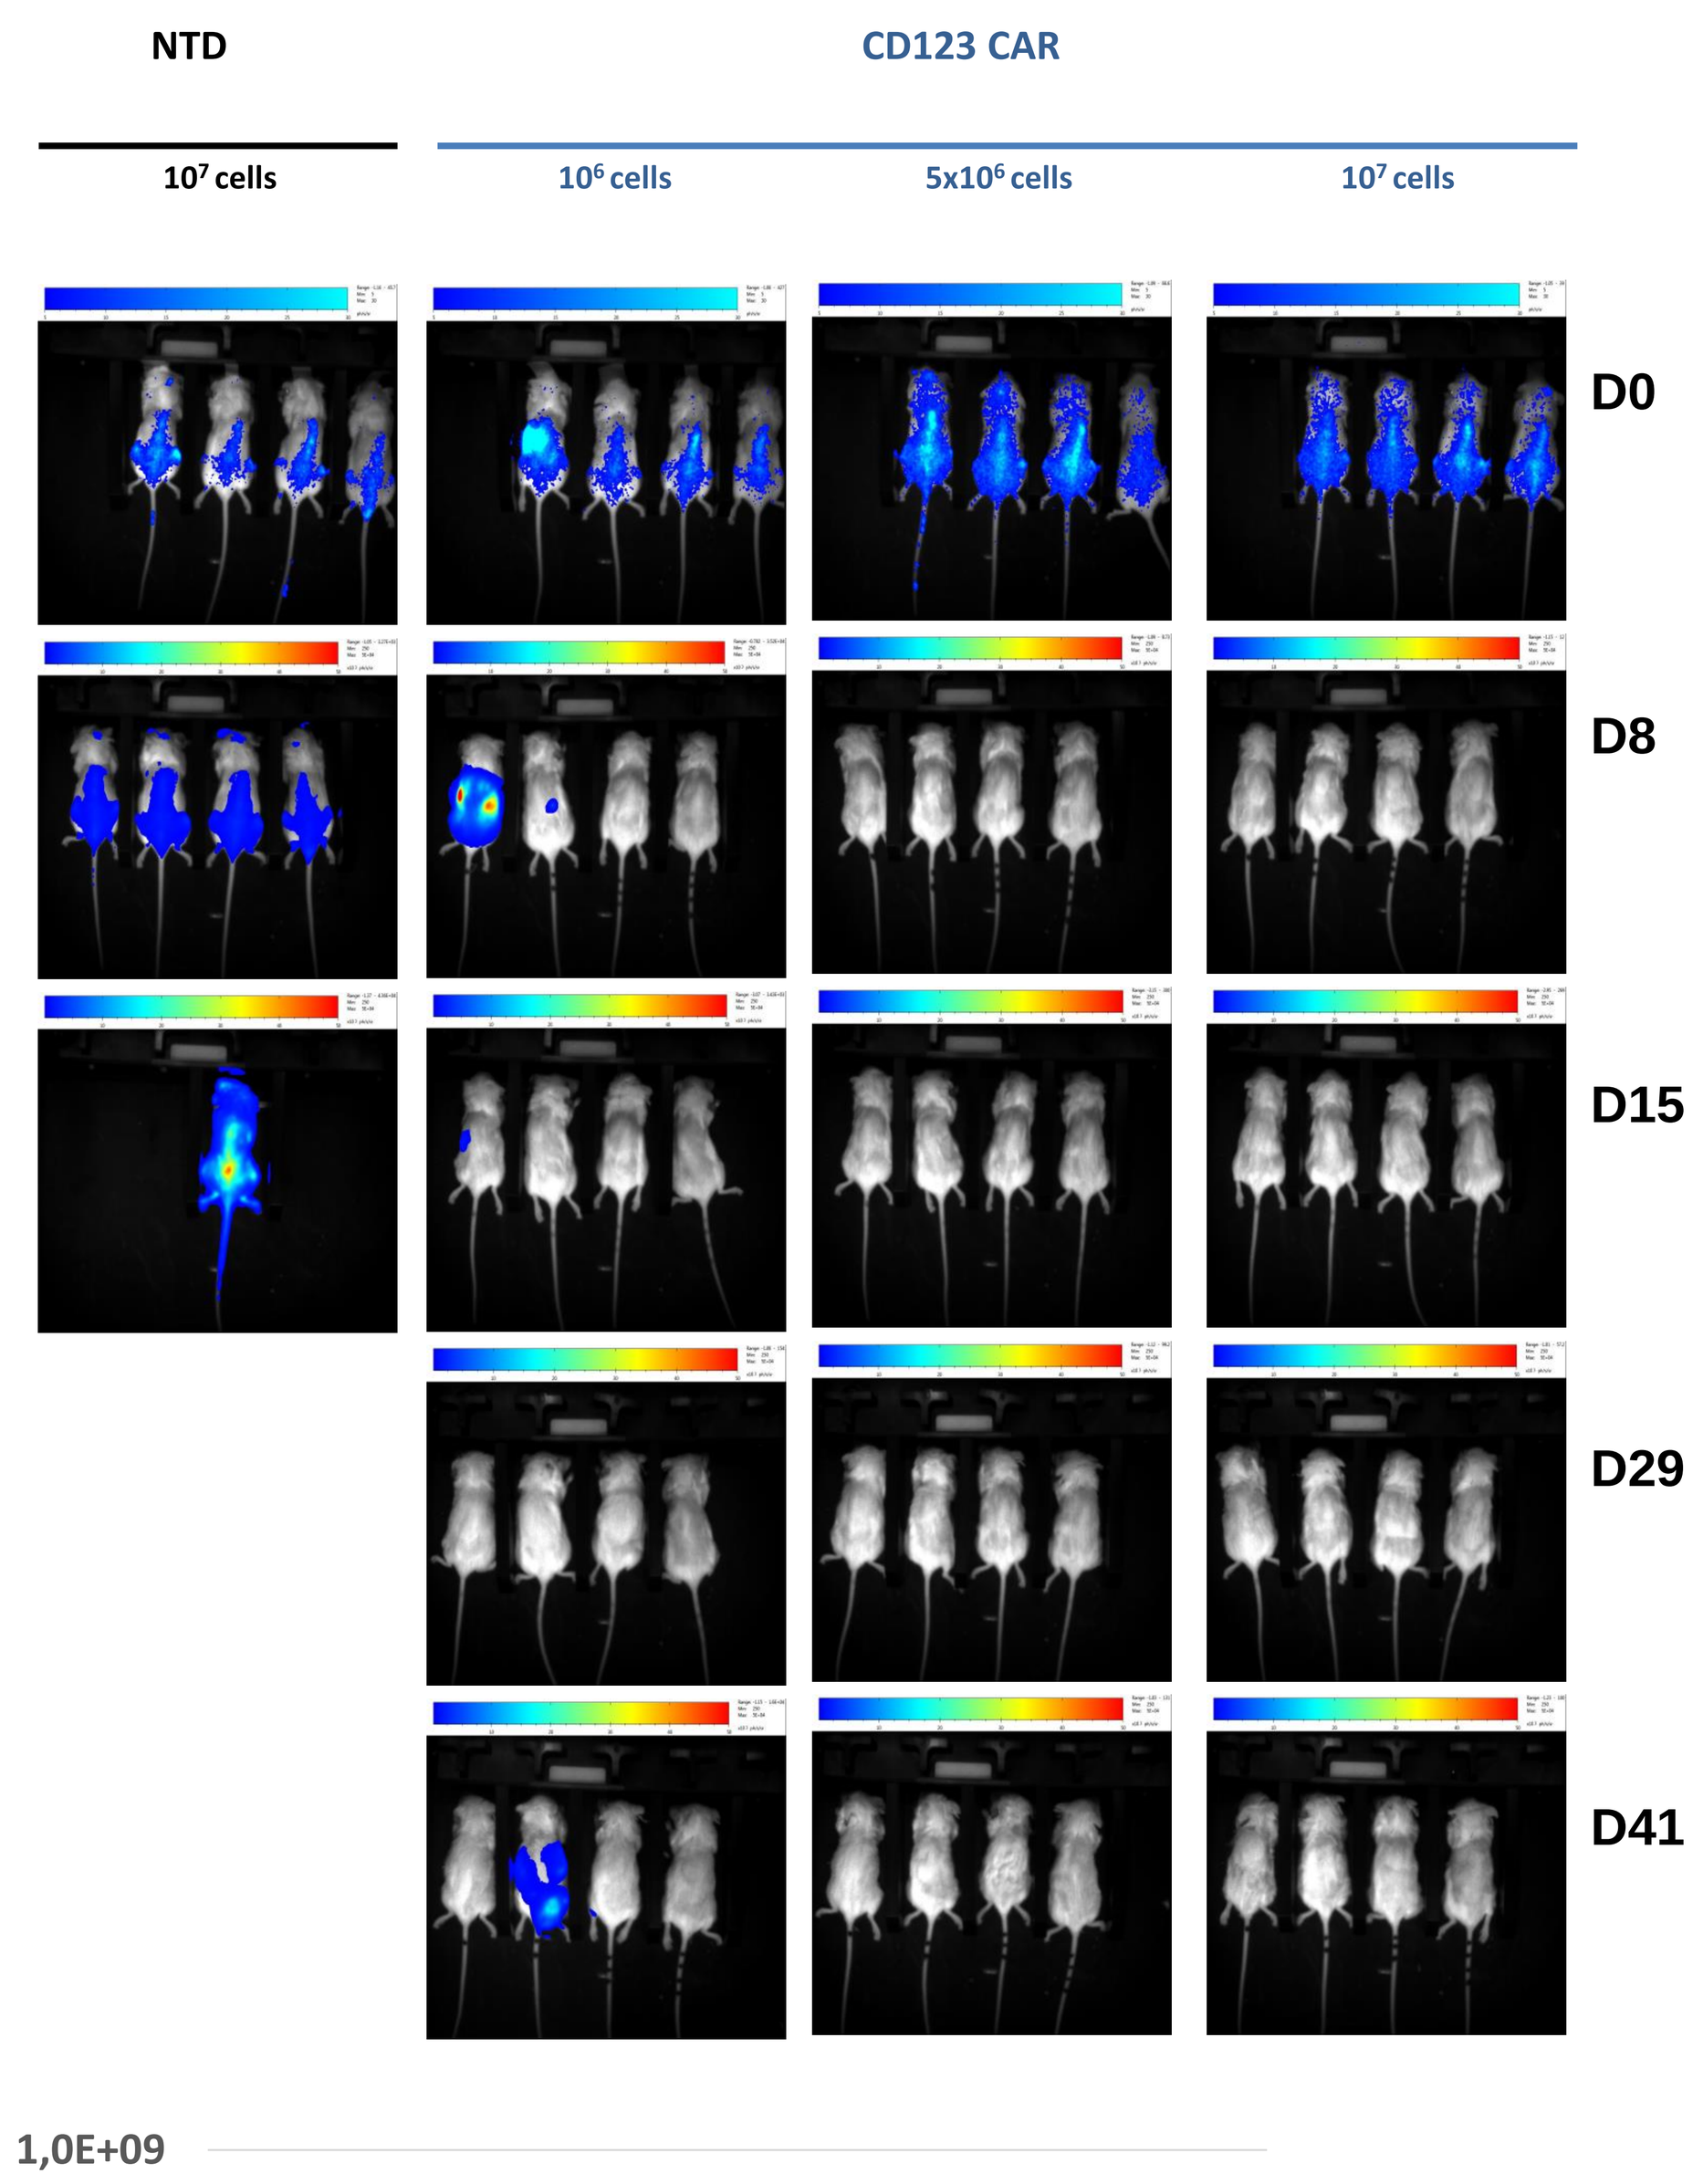

NTD
CD123 CAR
10^7 cells
10^6 cells
5x10^6 cells
10^7 cells
D0
D8
D15
D29
D41

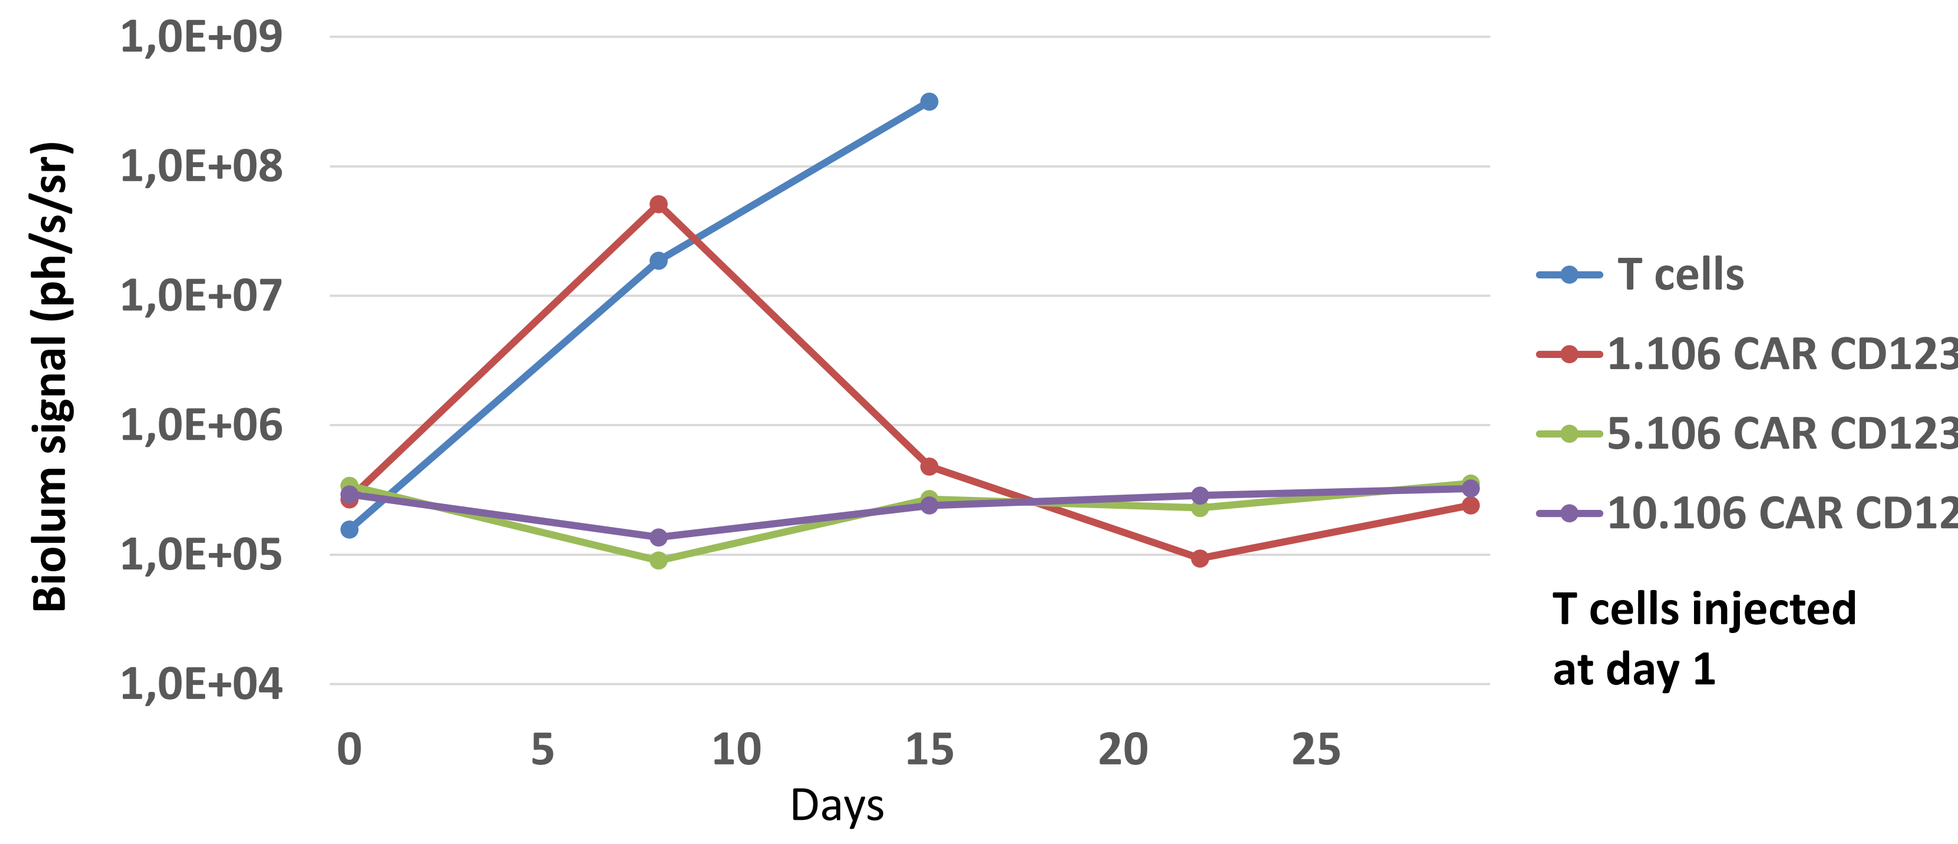

Biolum signal (ph/s/sr)
Days
T cells
1.106 CAR CD123
5.106 CAR CD123
10.106 CAR CD123
T cells injected at day 1

Except for one mouse (treated at 1x10^6), all the mice treated with CAR T-cells were alive at Day 60. In addition, flow cytometry analysis at Day 60 confirmed the complete elimination of the tumor cells in the blood, the spleen and the bone marrow.

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis and its business. Cellectis is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. Cellectis proprietary information