Poster # 380

NON-VIRAL DNA DELIVERY COUPLED TO TALEN GENE EDITING EFFICIENTLY CORRECT THE SICKLE CELL MUTATION IN LONG-TERM HSCS

Arianna Moiani[1], Gil Letort[1], Sabrina Lizot[1], Patrick Hong[2], Sonal Temburni-Blake[2], Tristan Felix[3], Anne Chalumeau[3], Annarita Miccio[3], Agnès Gouble[1], Alexandre Juillerat[1], Aymeric Duclert[1], Philippe Duchateau[1], Julien Valton[1]

[1]Cellectis S.A., 8 Rue de la Croix Jarry, 75013 Paris, France; [2]Cellectis, Inc., 430 East 29th Street, New York, NY 10016, USA; [3]Université de Paris, Imagine Institute, Laboratory of Chromatin and Gene Regulation During Development, INSERM UMR 1163, Paris, France

Background

Sickle Cell Disease (SCD)

- SCD is an inherited blood disorder that stems from a single point mutation (A>T) in exon 1 of the *HBB* gene. *HBB* encodes the hemoglobin beta subunit (Hb) that associates with hemoglobin alpha subunit to generate the tetrameric protein complex called adult hemoglobin (HbA).[1]
- Mutated HbA polymerization results in sickle shaped red blood cells (RBCs) that cause reduced oxygen transfer to tissues throughout the body and alter normal blood flow.[1,2]

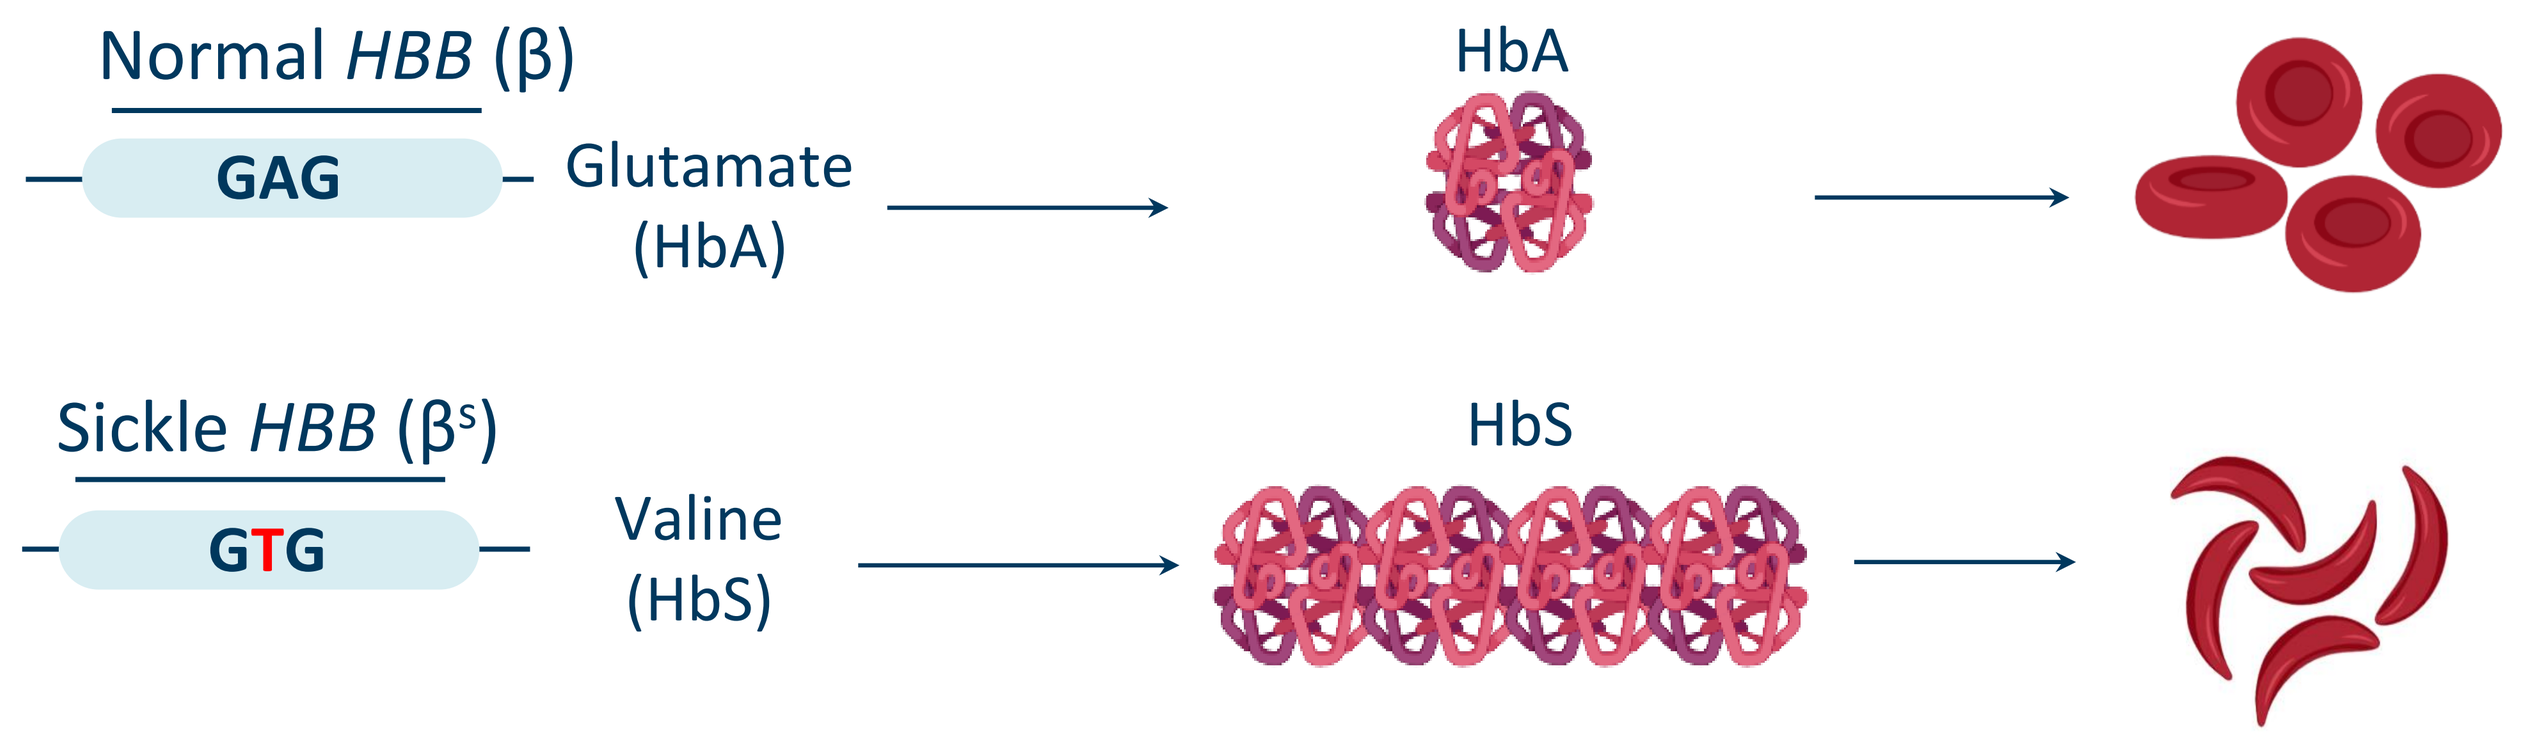
Normal HBB (β)
GAG
Glutamate (HbA)
HbA
Sickle HBB (βˢ)
GTG
Valine (HbS)
HbS

- People with SCD often suffer from anemia, painful vaso-occlusive crises, frequent infections, stroke and many other symptoms that can, ultimately, reduce quality of life and expected lifespan[2].
- SCD can be cured with hematopoietic stem cell transplant. However, this procedure is only available to patients with severe disease, it requires an HLA-matched donor and is associated to a substantial rate of morbidity and mortality[2]. Thus, alternative treatments are highly regarded
- This work proposes an autologous gene therapy approach based on *ex vivo HBB* gene correction in HSPCs to treat SCD.

References: 1. Renaudier P, Transfus Clin Biol. 2014; 2. Piel FB et al, Lancet 2013;Eapen et al. Lancet Hematol. 2019

TALEN®-mediated *HBB* gene correction strategy

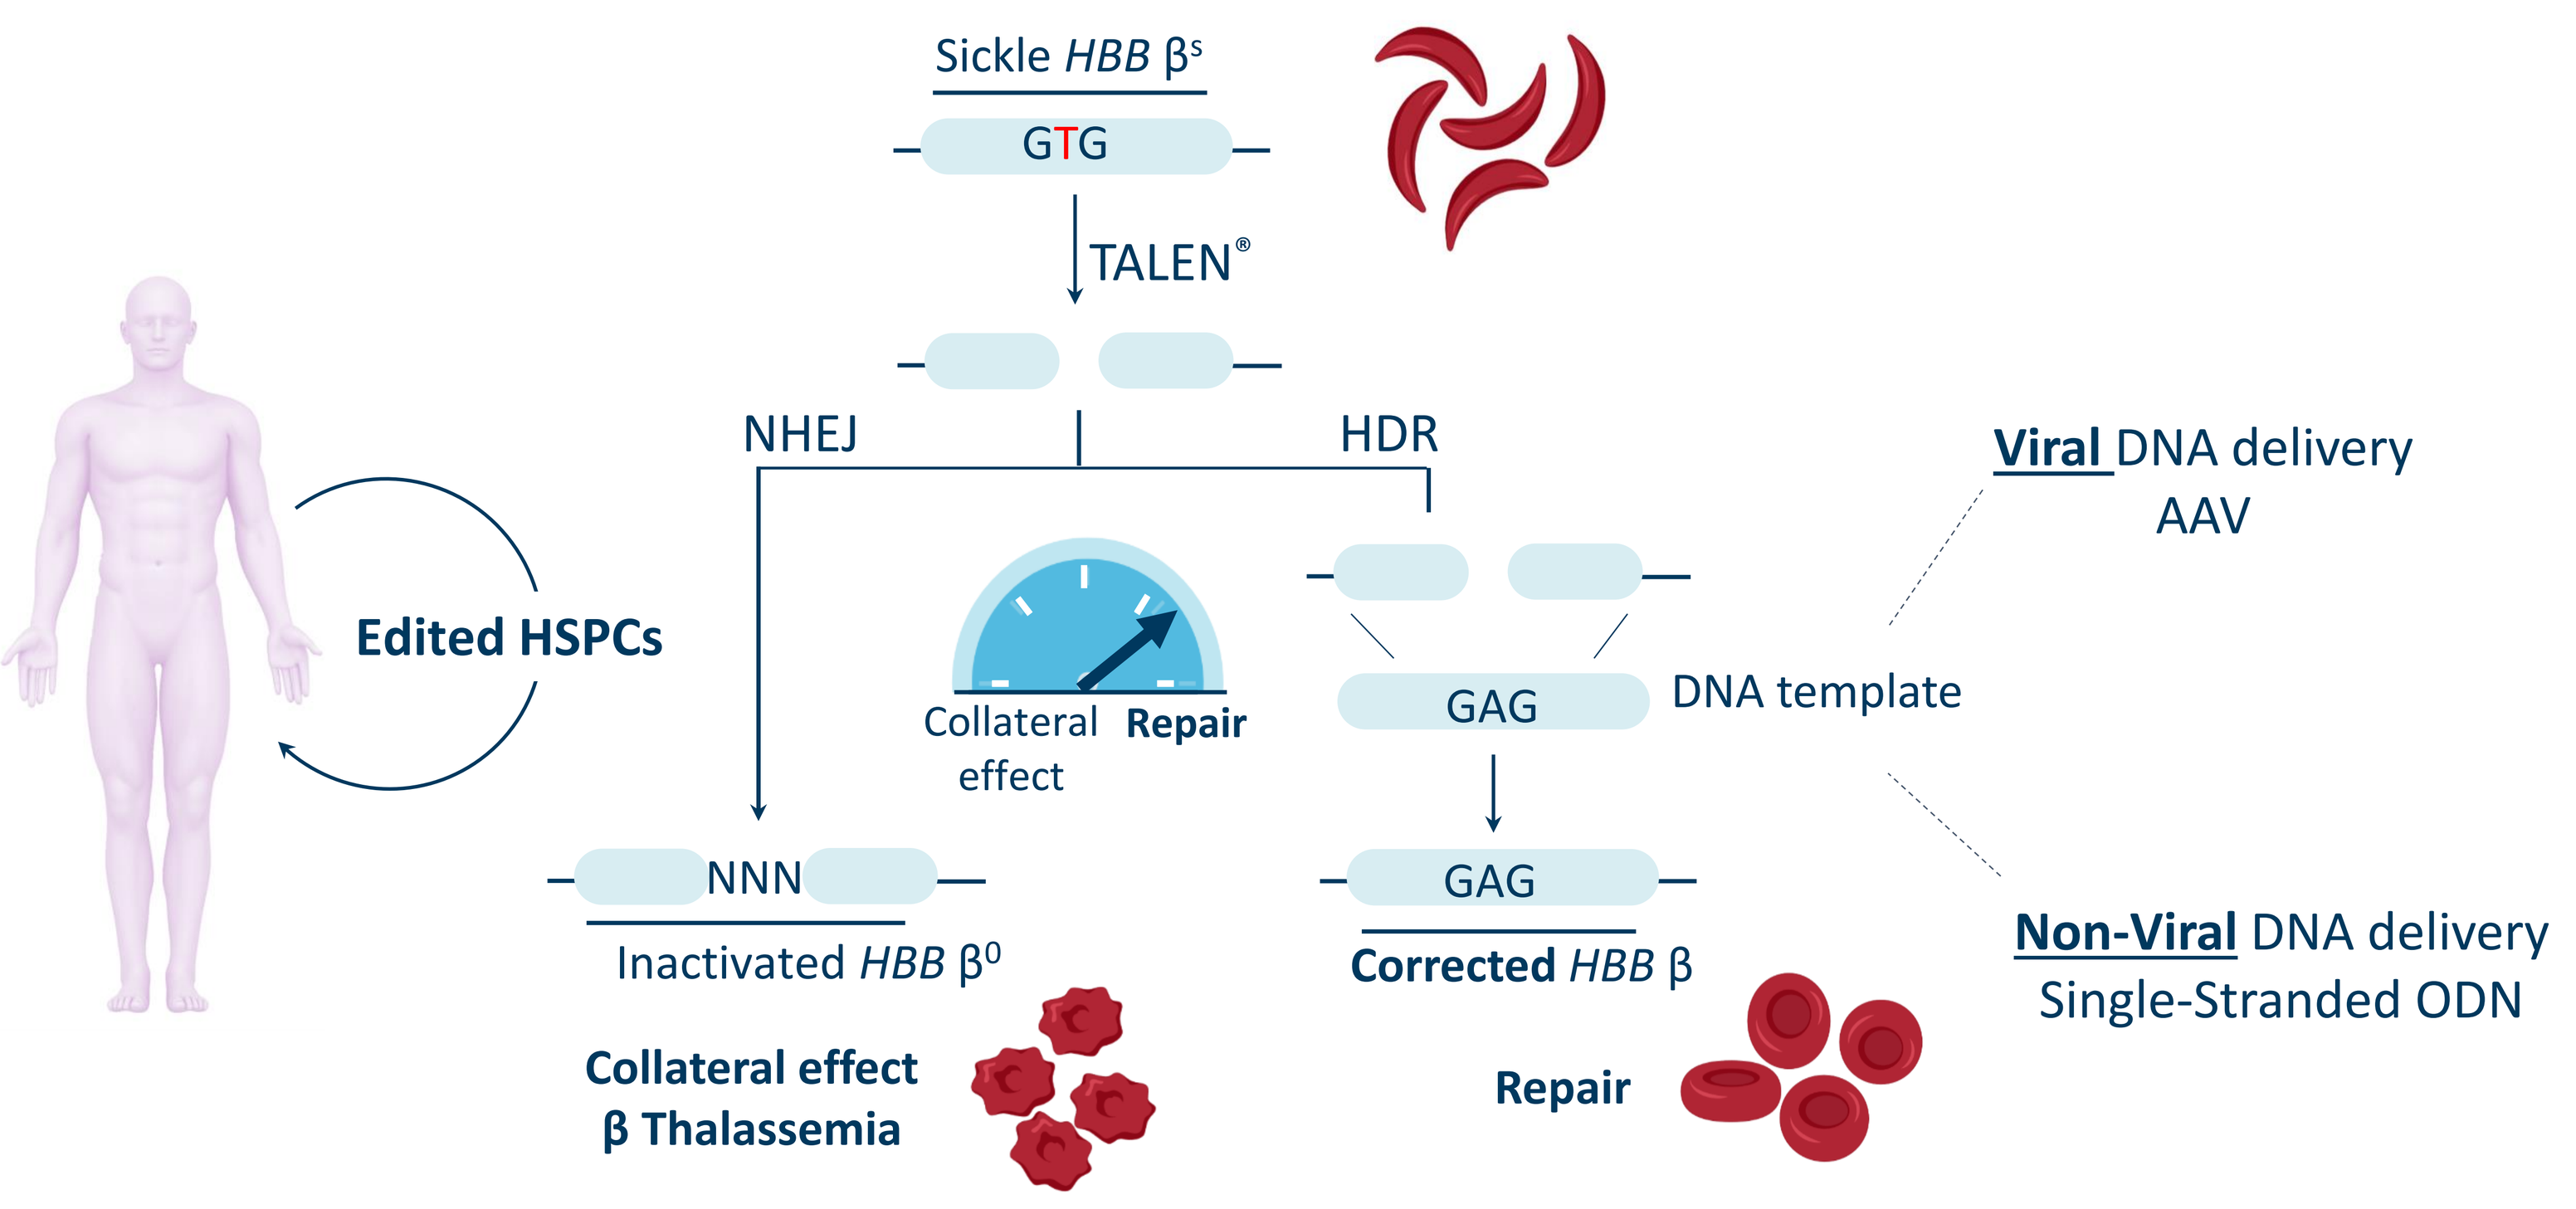
Sickle HBB βˢ
GTG
TALEN®
NHEJ
HDR
Edited HSPCs
Collateral effect
Repair
NNN
Inactivated HBB β⁰
Collateral effect
β Thalassemia
GAG
DNA template
GAG
Corrected HBB β
Repair
Viral DNA delivery
AAV
Non-Viral DNA delivery
Single-Stranded ODN

- We developed an autologous gene therapy strategy to treat SCD with a TALEN® targeting the mutated *HBB* gene followed by the delivery of a DNA template to repair the *HBB* mutation via homologous directed repair (HDR).
- Here, we report that our protocol 1), efficiently corrects *HBB* in long-term repopulating hematopoietic stem cells (LT-HSCs) and repairs the sickle cell phenotype in differentiated RBCs 2), provides a direct comparison of viral and non-viral strategies for DNA delivery and 3), mitigates collateral effect derived from bi-allelic inactivation of *HBB*.

TALEN®-HBB displays editing activity at *HBB* locus and high specificity with only one off-target site detected

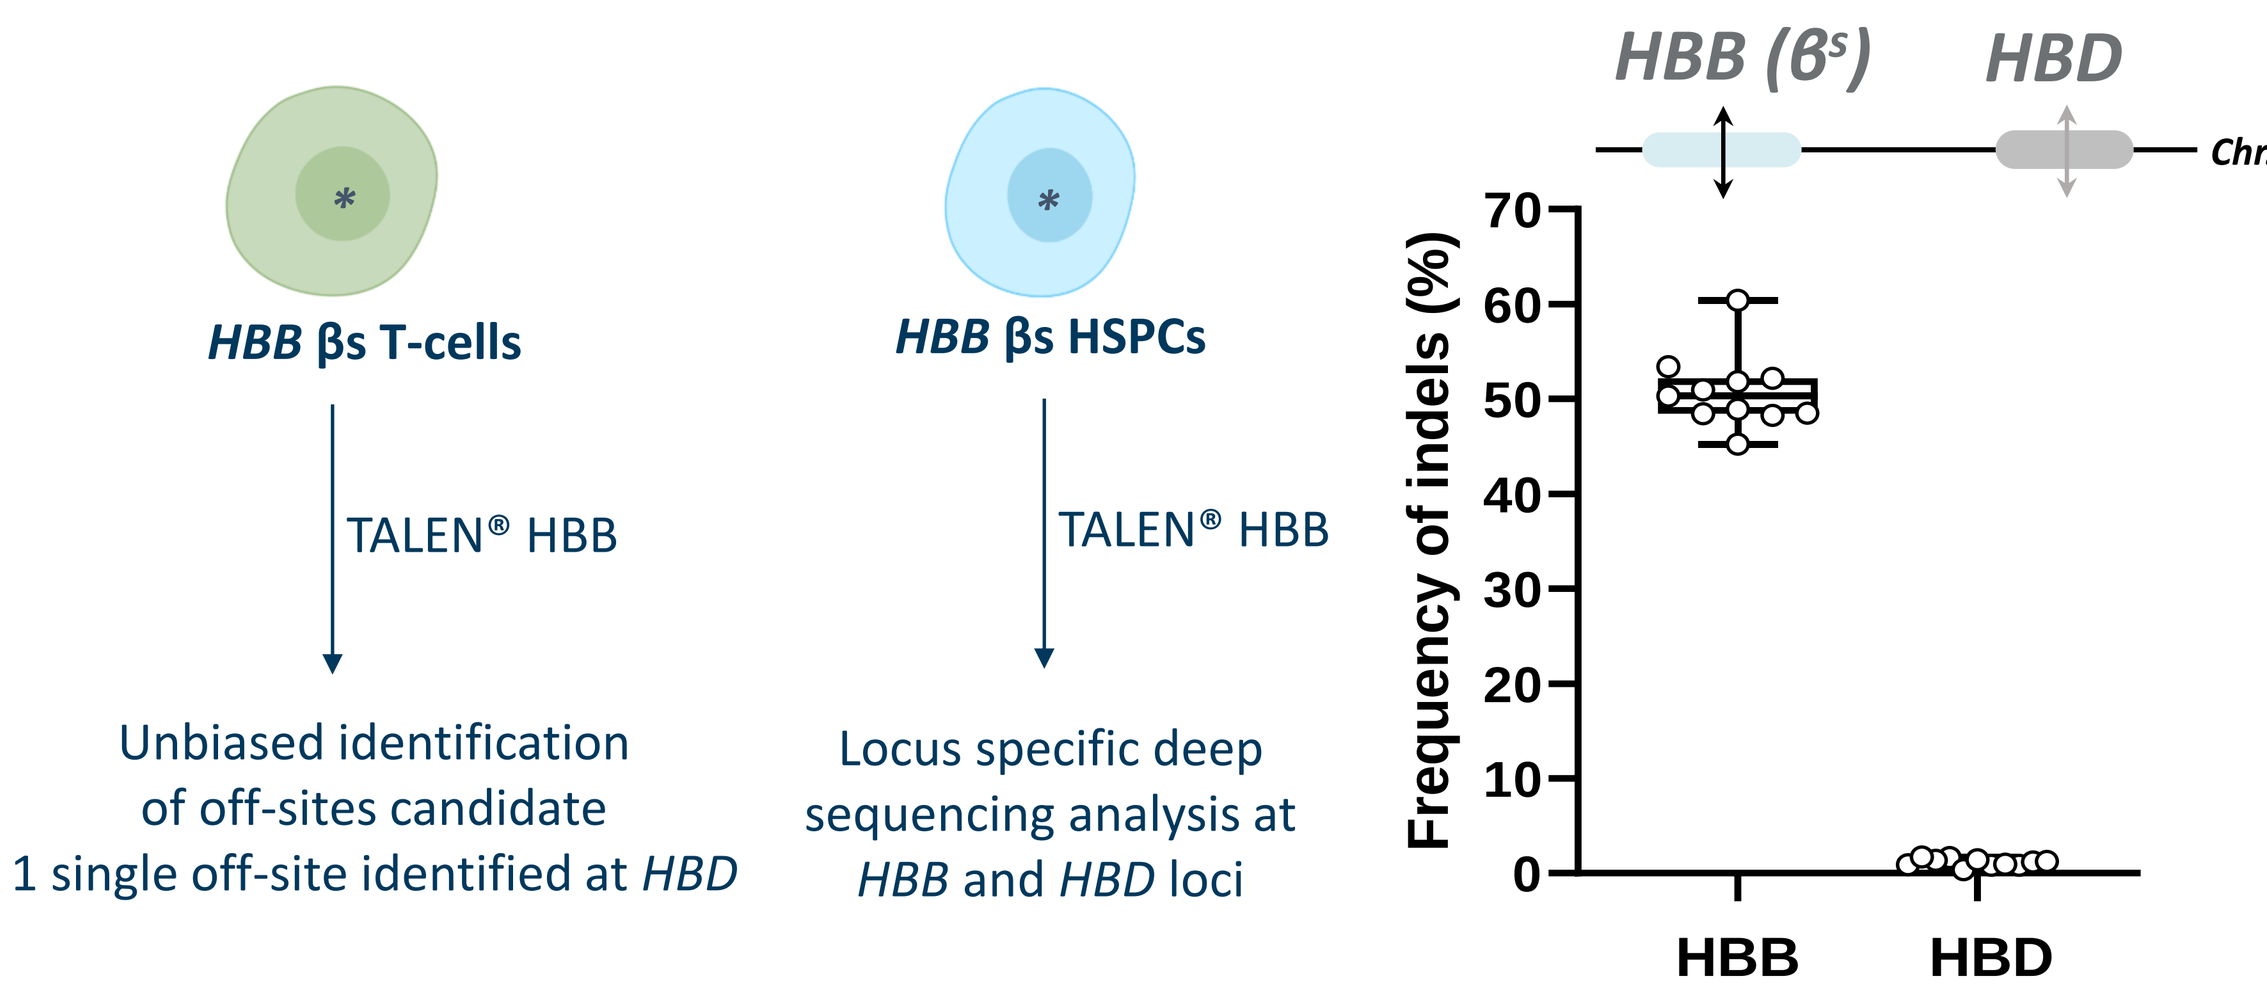
HBB βs T-cells
TALEN® HBB
Unbiased identification of off-sites candidate
1 single off-site identified at HBD
HBB βs HSPCs
TALEN® HBB
Locus specific deep sequencing analysis at HBB and HBD loci
HBB (βˢ)
HBD
Chr.11
Frequency of indels (%)
HBB
HBD

- High activity of TALEN®-HBB detected at HBB onsite in HBB βs HSPCs
- High specificity of TALEN®-HBB assessed by unbiased Oligo Capture Assay and locus specific deep sequencing. Only one off-site was confirmed at the *HBD* gene. TALEN® cleavage off-site activity was found to be very low compared to the on-site cleavage activity (1.2% +/- 0.4 at the off-site versus 50.7% Indels +/- 3.9 at the on-site).

Highly efficient TALEN® based gene editing at *HBB* locus in mobilized HSPCs from healthy donors

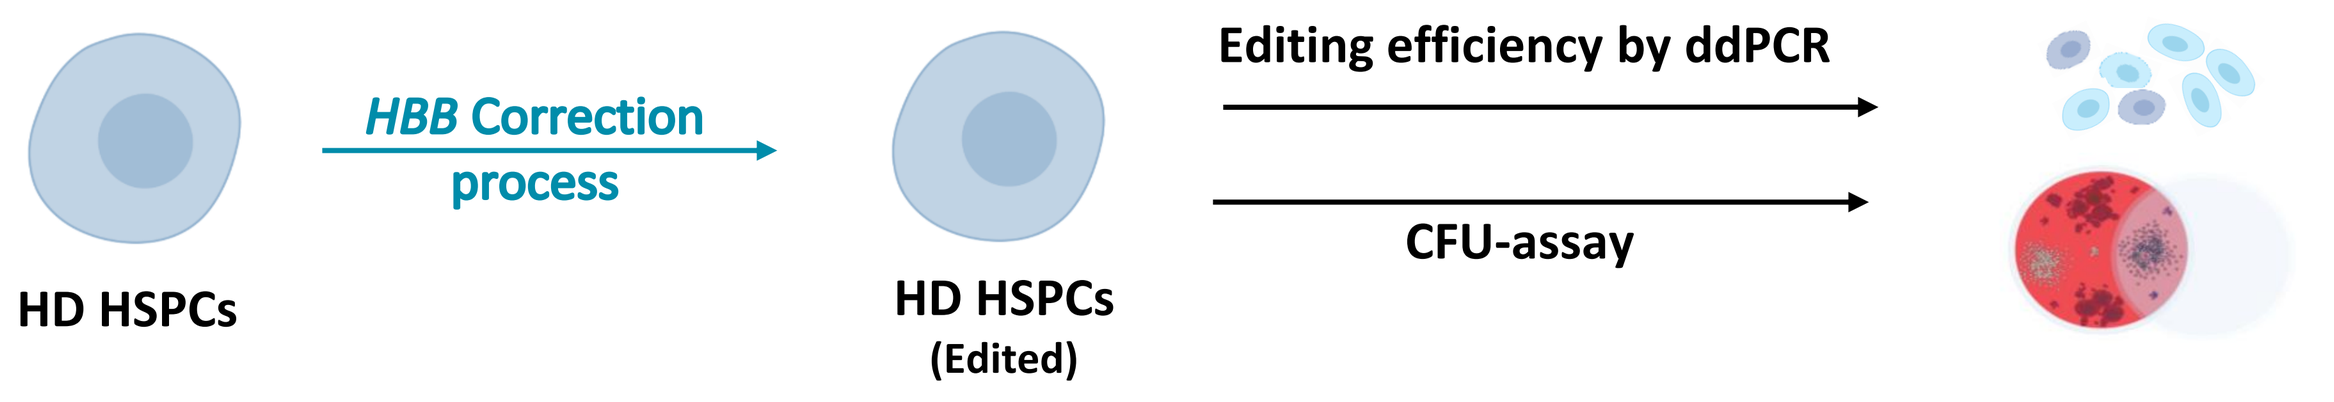
A.
HD HSPCs
HBB Correction process
HD HSPCs (Edited)
Editing efficiency by ddPCR
CFU-assay

- Mobilized HSPCs from healthy donors were expanded, edited with TALEN® coupled to either AAV6 or ssODN delivering the correct *HBB* sequence. Edited cells were analyzed 48h after editing to assess editing efficiency, viability, purity and clonogenic potential by CFU-assay (A).

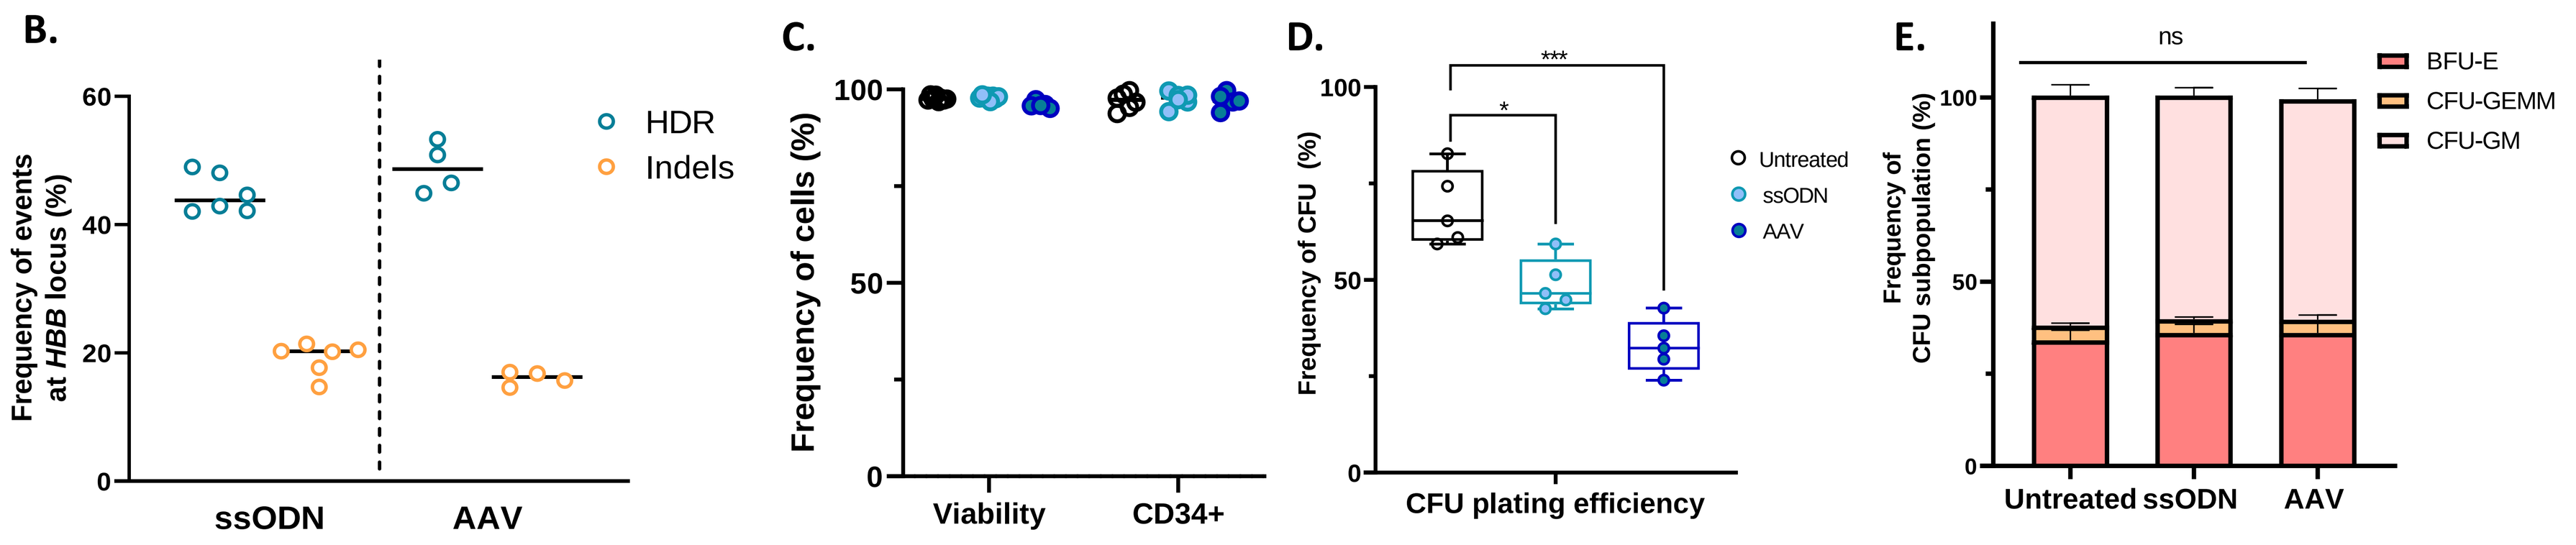
B.
Frequency of events at HBB locus (%)
HDR
Indels
ssODN
AAV
C.
Frequency of cells (%)
Viability
CD34+
D.
Frequency of CFU (%)
Untreated
ssODN
AAV
CFU plating efficiency
E.
Frequency of CFU subpopulation (%)
BFU-E
CFU-GEMM
CFU-GM
Untreated
ssODN
AAV

- Our process led to high HDR frequencies (>50%), while maintaining low levels of indels (<20%) either using AAV6 or ssODN (B). It was reproducible in six different donors and did not affect HSPCs viability (>95% mean viable cells), or purity (> 95% mean CD34+ cells assessed by FACS) (C). We observed a decreased HSPCs clonogenic potential after editing which was further impacted by AAV transduction (D), without lineage skewing between edited and untreated HSPCs (E).

Non-viral gene editing mitigates p53 response activation, maintains LT-HSC pool and fitness and enables high level gene correction *in vivo*

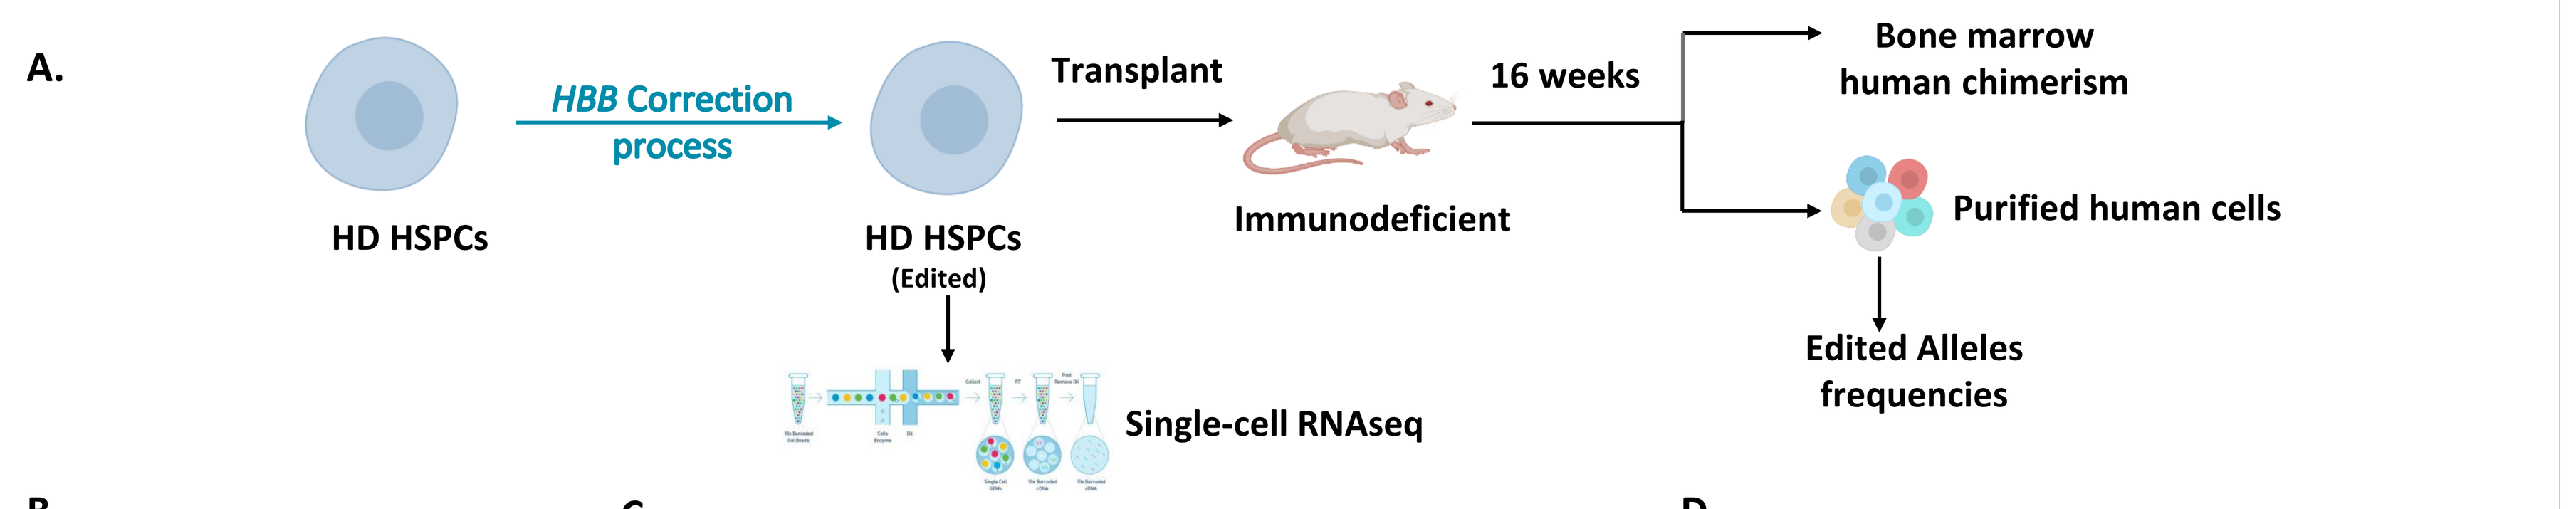
A.
HD HSPCs
HBB Correction process
HD HSPCs (Edited)
Transplant
Immunodeficient
16 weeks
Bone marrow human chimerism
Purified human cells
Edited Alleles frequencies
Single-cell RNAseq

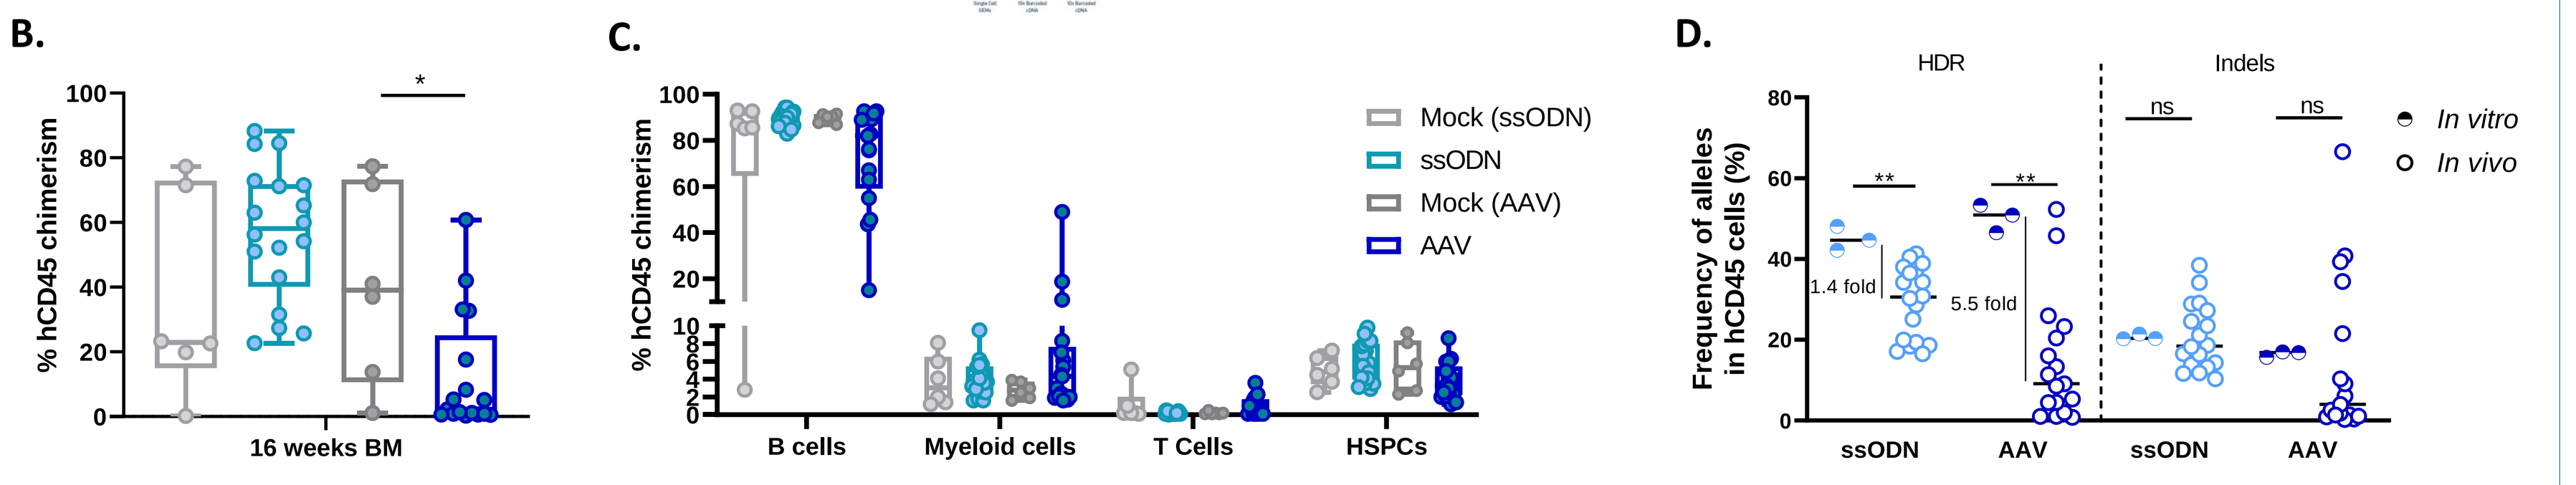
B.
% hCD45 chimerism
16 weeks BM
C.
% hCD45 chimerism
Mock (ssODN)
ssODN
Mock (AAV)
AAV
B cells
Myeloid cells
T Cells
HSPCs
D.
HDR
Indels
Frequency of alleles in hCD45 cells (%)
1.4 fold
5.5 fold
In vitro
In vivo
ssODN
AAV

- To assess the ability of TALEN® editing to achieve high HDR efficiencies in LT-HSCs, edited plerixafor-mobilised HSPCs from three healthy donors were injected in immunodeficient mice and analyzed 16 weeks post-engraftment to assess bone marrow (BM) human chimerism, multilineage engraftment and editing efficiencies (A).
- Non-viral DNA delivery performed better than viral-mediated editing *in vivo*, reaching high level BM engraftment, comparable to non-edited HSPCs (B), multilineage reconstitution (C) and long-term maintenance of HDR in engrafted LT-HSCs at levels comparable to the input (average 30% HDR at 16 weeks in BM) (D).
- Single-cell RNAseq (scRNAseq) was performed in HSPCs before *in vivo* injection to better characterize the discrepancies observed between viral and non-viral editing of LT-HSCs (A).

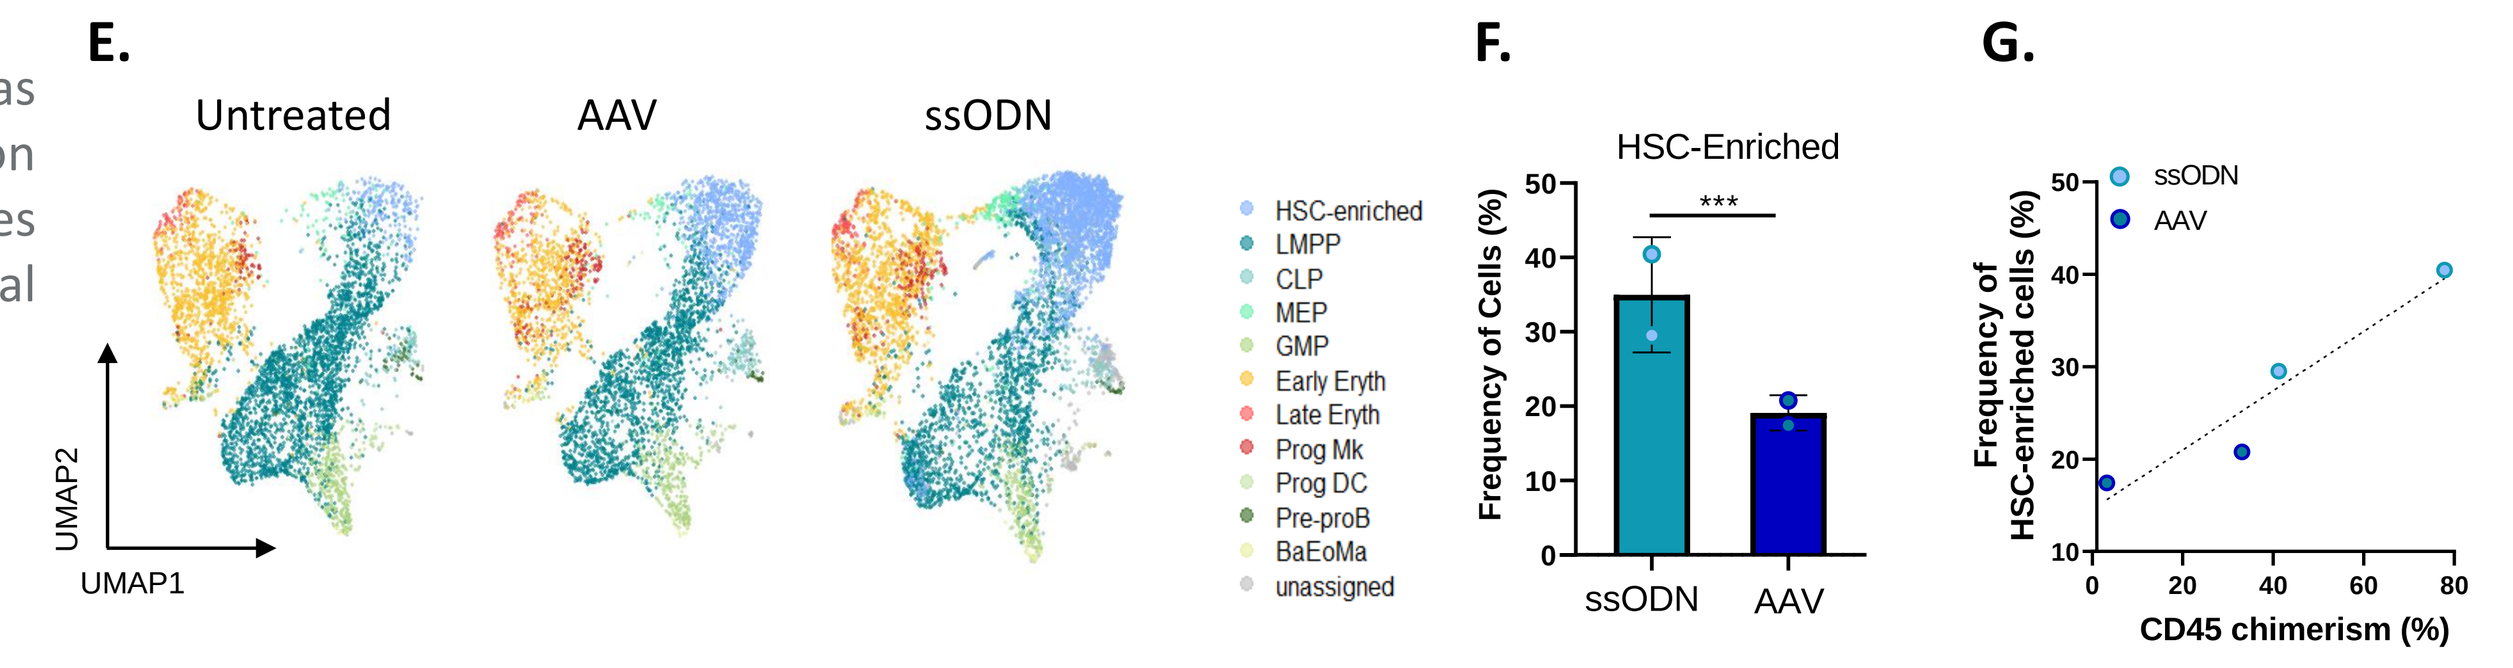
E.
Untreated
AAV
ssODN
UMAP2
UMAP1
HSC-enriched
LMPP
CLP
MEP
GMP
Early Eryth
Late Eryth
Prog Mk
Prog DC
Pre-proB
BaEoMa
unassigned
F.
HSC-Enriched
Frequency of Cells (%)
ssODN
AAV
G.
ssODN
AAV
Frequency of HSC-enriched cells (%)
CD45 chimerism (%)

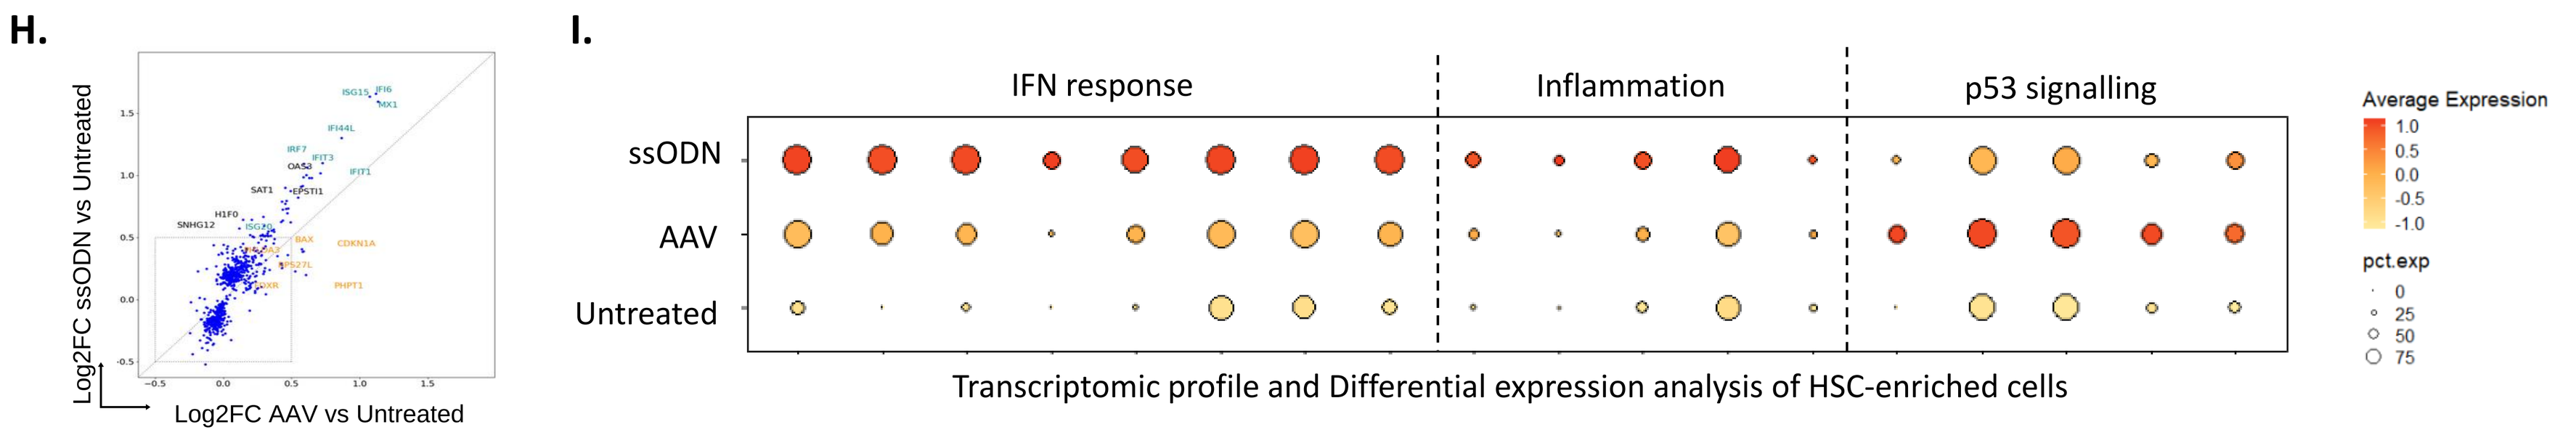
H.
Log2FC ssODN vs Untreated
Log2FC AAV vs Untreated
I.
IFN response
Inflammation
p53 signalling
ssODN
AAV
Untreated
Average Expression
pct.exp
Transcriptomic profile and Differential expression analysis of HSC-enriched cells

- A higher proportion of primitive HSC-enriched cells was found in ssODN- vs AAV6-edited sample (E-F) correlating with engraftment data (G). By looking at transcription profile of HSC-enriched cells, we observed that edited HSCs upregulated several pathways including interferon, inflammation and p53 signaling, compared to untreated sample (H-I). Interestingly, a mitigated p53 signaling activation was observed in ssODN edited samples suggesting that non-viral gene editing better preserves HSC fitness than viral gene editing (I).

TALEN®-mediated gene editing reaches high level of gene correction in SCD patient's derived HSPCs with minimal collateral effect

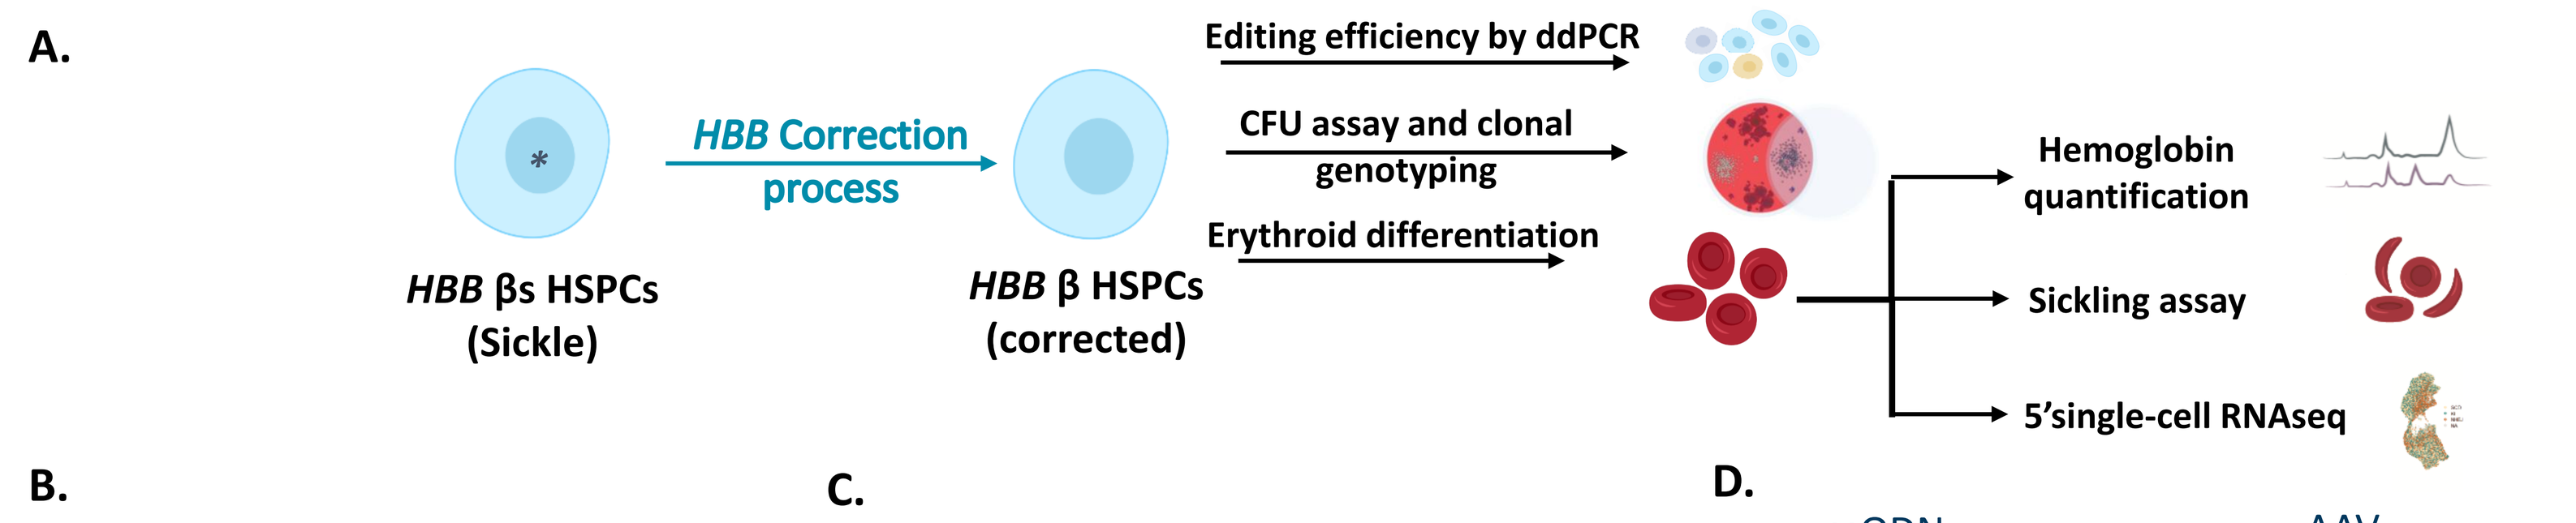
A.
HBB βs HSPCs (Sickle)
HBB Correction process
HBB β HSPCs (corrected)
Editing efficiency by ddPCR
CFU assay and clonal genotyping
Erythroid differentiation
Hemoglobin quantification
Sickling assay
5'single-cell RNAseq

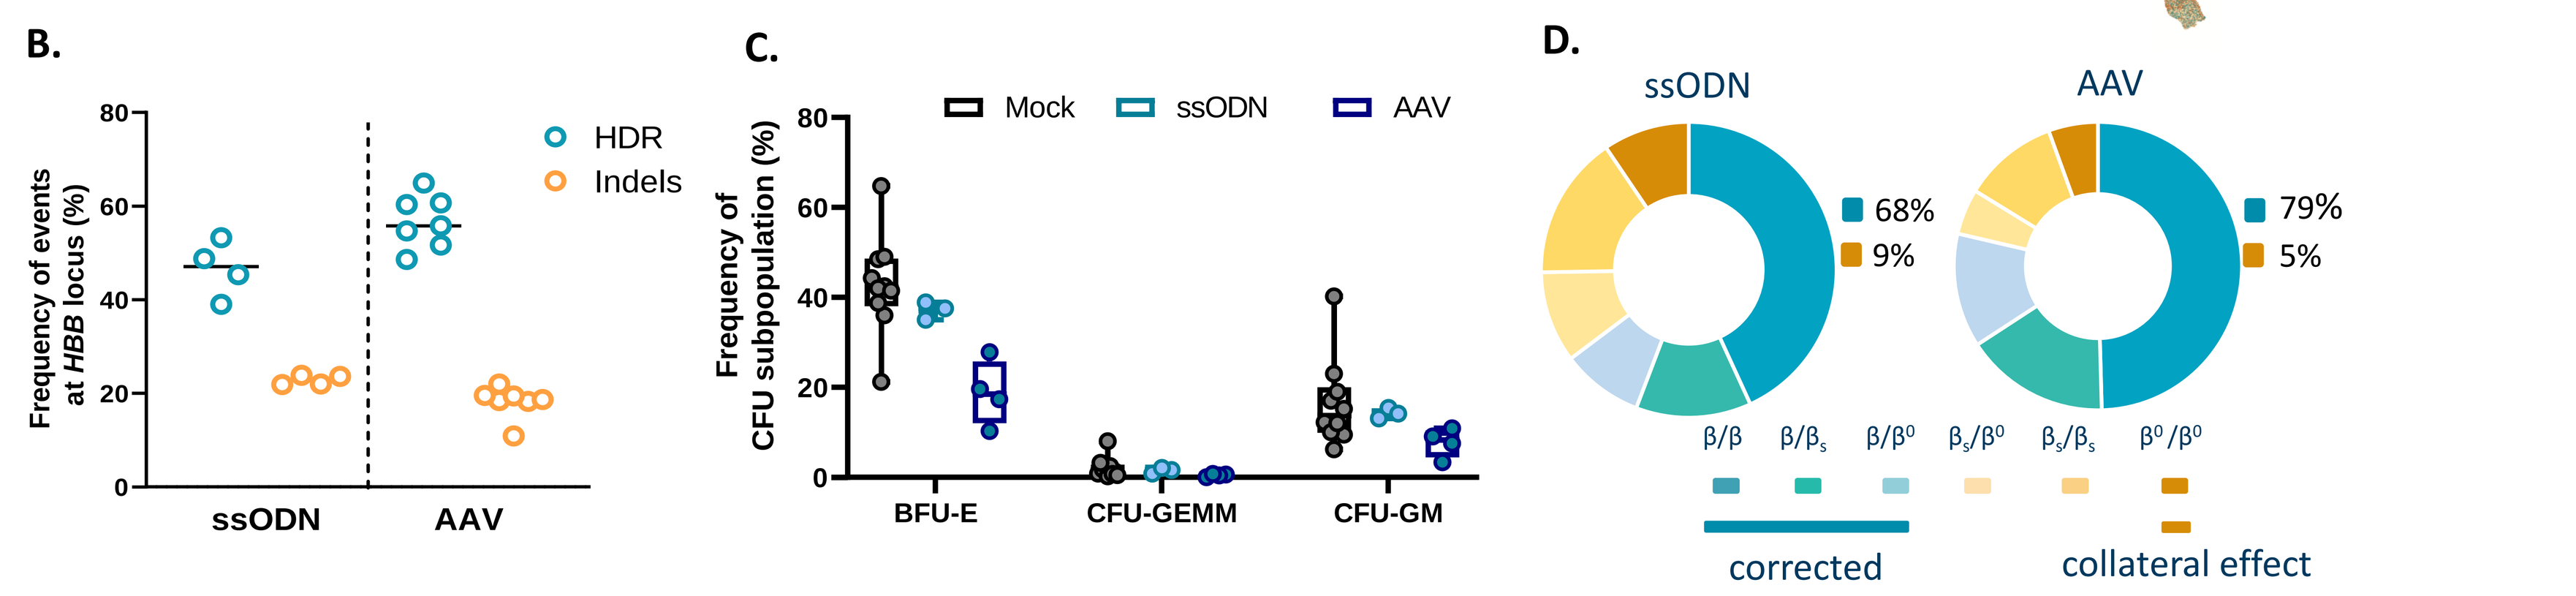
B.
Frequency of events at HBB locus (%)
HDR
Indels
ssODN
AAV
C.
Frequency of CFU subpopulation (%)
Mock
ssODN
AAV
BFU-E
CFU-GEMM
CFU-GM
D.
ssODN
AAV
68%
9%
79%
5%
β/β
β/βs
β/β0
βs/β0
βs/βs
β0/β0
corrected
collateral effect

- To assess whether our gene editing protocol would efficiently correct sickle-cell HSPCs, we purified HSPCs from multiple non-mobilized homozygous sickle patients (nmHbSS) and assessed gene editing efficiencies in CD34+ bulk and BFU-E single clones and clonogenic potential. We also differentiated edited HSPCs into fully mature red blood cells (RBCs) to evaluated hemoglobin expression, sickling properties and the potential beta-thalassemic phenotype through scRNAseq (A)
- Our editing strategy achieved high gene correction frequencies (>50%), while maintaining low levels indels (<20%)(B). It was reproducible in multiple different donors and did not impact HSPCs clonogenic potential (C). Notably we reached an average of 68% and 79% of corrected BFU-E clones while generating less than 10% bi-allelic indels clones.

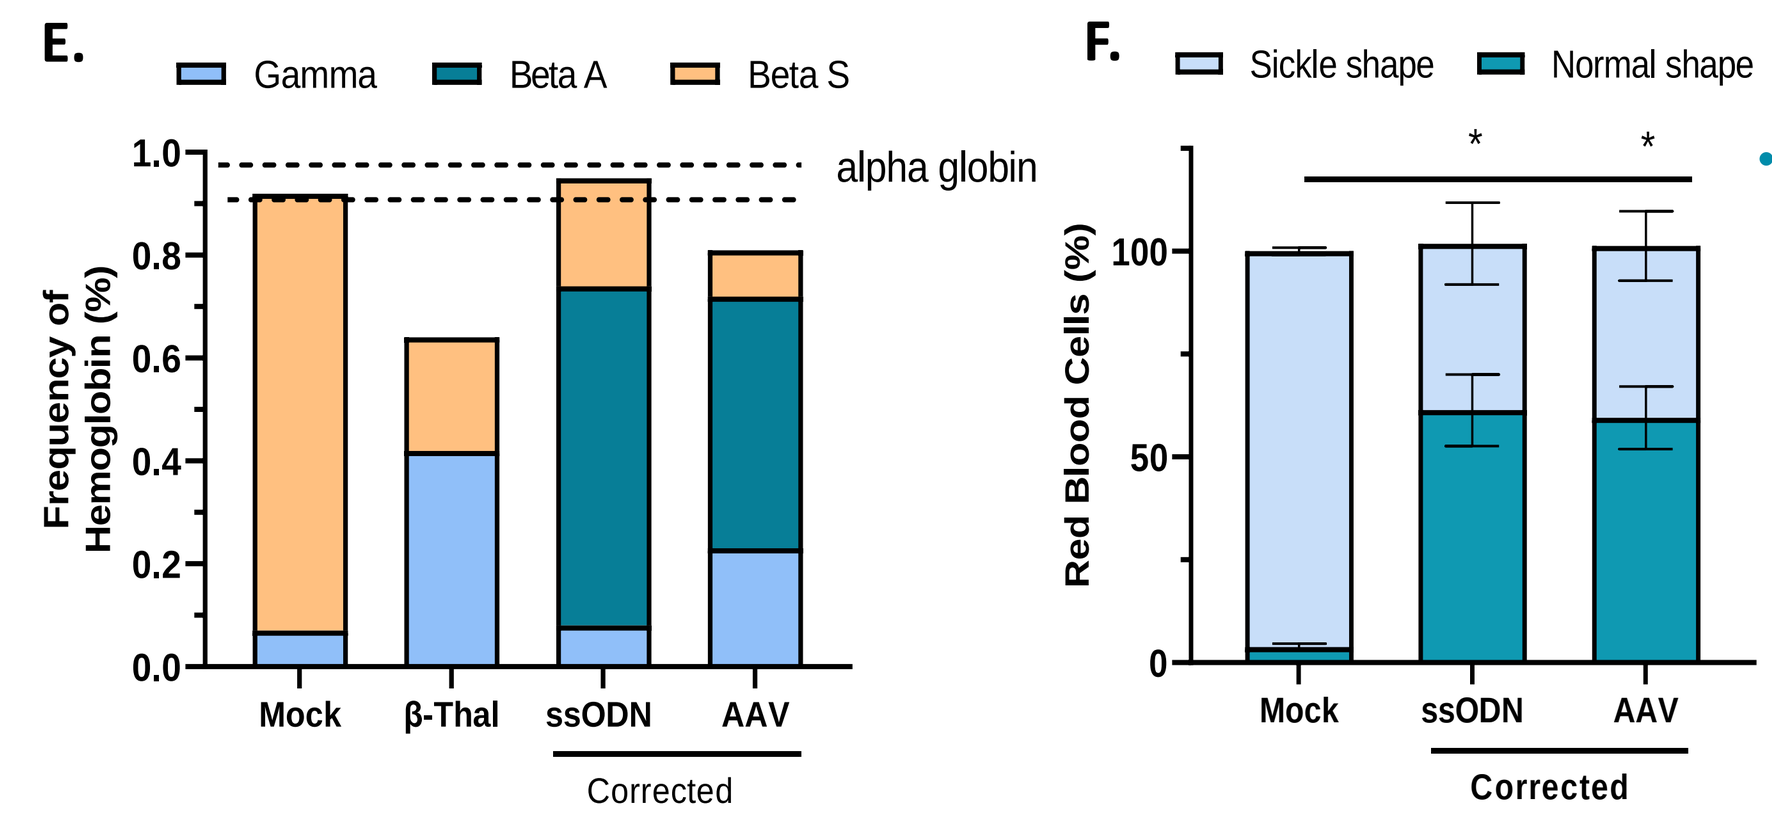
E.
Gamma
Beta A
Beta S
alpha globin
Frequency of Hemoglobin (%)
Mock
β-Thal
ssODN
AAV
Corrected
F.
Sickle shape
Normal shape
Red Blood Cells (%)
Mock
ssODN
AAV
Corrected

- Our protocol allowed high expression of adult hemoglobin (HbA) (>50%), while maintaining a correct level of total hemoglobin as assessed by alpha to non-alpha globin ratio (alpha globin level set to 1) (E). Moreover, it mitigated collateral effect derived from *HBB* inactivation observed in the β-thalassemic-like control (β-Thal). Finally, TALEN® gene editing enables rescuing the sickling phenotype (up to 70% of normal cells obtained) in hypoxic conditions (F).
- scRNaseq analysis enabling to simultaneously characterize the genotype and transcription profiles of edited RBCs, highlighted a cell cluster in the β-thal control, which was absent in the mock (G). Interestingly, this cluster was mainly composed of RBCs harboring bi-allelic indels and was reduced in the edited samples (<10% cells)(G-H).
- A β-thalassemic signature characterized by upregulation of heat shock protein genes (HSP), ribosomal proteins genes and genes encoding for fetal Hb was observed in the β-thal control, while edited RBCs had a transcription profile similar to the mock control(I). These data confirm the reduced risk of TALEN®-gene correction to generate β-thal phenotype which is restricted only to the cluster of cells harboring bi-allelic indels as shown by HSPA1A expression(G).

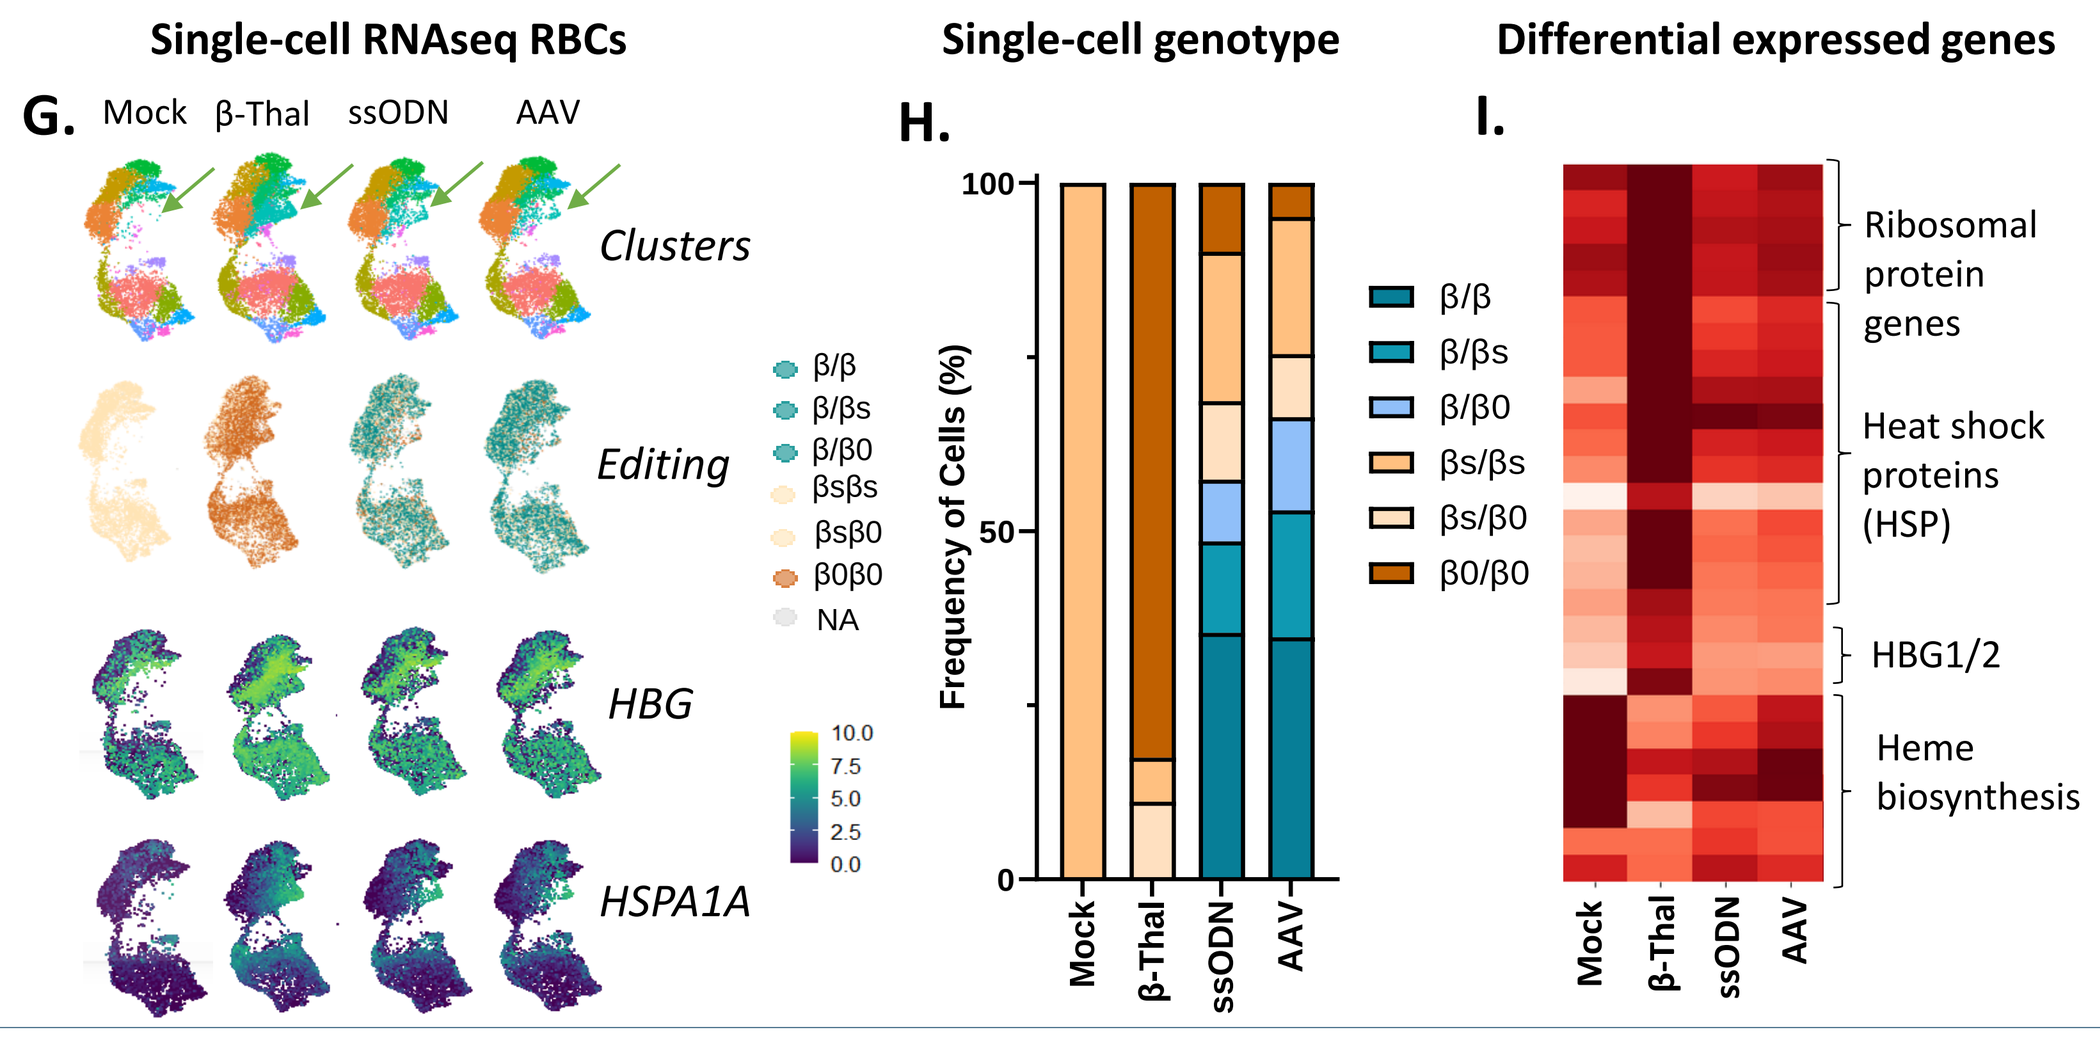
Single-cell RNAseq RBCs
G.
Mock
β-Thal
ssODN
AAV
Clusters
Editing
HBG
HSPA1A
Single-cell genotype
H.
Frequency of Cells (%)
Differential expressed genes
I.
Ribosomal protein genes
Heat shock proteins (HSP)
HBG1/2
Heme biosynthesis

Conclusions

-TALEN®-mediated engineering efficiently corrects the mutated *HBB* gene in clinically relevant HSPCs (68-79% corrected progenitors) and this translates into phenotype correction in fully mature RBCs (Up to 50% HbA expression)

-Our optimized TALEN® gene editing process mitigates potential safety challenges by reducing the frequency of *HBB* gene inactivation (<10% β-thal cells)

-Non-viral DNA template mediated repair mitigates p53 DNA damage response activation and preserve LT-HSCs fitness

-Non-viral DNA delivery template coupled to TALEN® editing enables higher engraftment and maintenance of gene correction in LT-HSCs (Up to 40% corrected LT-HSCs)

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis and its business. Cellectis is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. This communication contains Cellectis' proprietary information. TALEN® and Cellectis® are trademarks owned by the Cellectis Group.

Image Credit: Images of cells were adapted from "Cell Biology", by BioRender.com (2021); Retrieved from https://app.biorender.com/biorender-templates