TALEN®- Mediated non-viral Transgene Insertion for the Advancement of Cellular and Gene Therapies.

Gil Letort[1], Aymeric Duclert[1], Diane Leclerre[1], Isabelle Chion-Sotinel[1], Emilie Dessez[1], Margaux Sevin[1], Roman Galetto[1], Marco Rotondi[1], Jordan Skinner[2], Shipra Das[2], Cosimo Ducani[3], Roger Salvatori[3], Laurent Poirot[1], Beatriz Aranda-Orgilles[2], Philippe Duchateau[1], Julien Valton[1].

[1]Cellectis SA, Paris, France ; [2]Cellectis Inc, New York, NY; [3]Moligo Technologies, Anderstorpsvägen 16, 171 54, Solna, Sweden

SCAN ME

EDITING LIFE

Cell and gene therapy approaches can rely on using gene-editing tools to modify disease-associated genes and introducing transgenes in the genome. These modifications can restore or create new cellular functions, thereby providing a potential therapeutic solution for a wide array of diseases. Despite clinical successes in the immuno-oncology and gene therapy fields, several challenges persist. The traditional application of lentiviral vectors for payload delivery results in random genomic integration, which may pose risks of genomic toxicity or cellular transformation. Advanced techniques use targeted transgene integration facilitated by DNA repair templates with specific homology arms, which can be delivered in AAV particles. These templates are introduced to the cell along with gene-specific double-strand breaks created by gene-editing tools. While more specific, this approach also faces challenges such as manufacturing constraints, potential AAV-associated toxicities or limited payload size. Increasing evidence suggests that non-viral methods, such as single-stranded DNA, to deliver transgenes have the potential to address current efficiency, safety and manufacturing challenges. Consequently, there is a need to integrate these methods into the next-generation of therapies to treat a broad range of diseases.

Here, we explore the combination of TALEN® mediated gene editing and the delivery of non-viral donor templates, particularly single-stranded DNA in its linear (LssDNA) and circular (CssDNA) formats, to mediate gene insertion in T-cells and hematopoietic stem and progenitor cells (HSPCs). We show that non-viral templates are amenable to be used to efficiently edit T-cells with CssDNA leading to higher integration levels. In HSPCs, we also observe an increase in transgene insertion frequency using CssDNA compared to LssDNA, alongside increased viability and insertion specificity. Additionally, we use CITE-seq coupled with long-term engraftment capacity assessment in NCG mice to compare CssDNA with traditional viral DNA repair templates (AAV). Our results demonstrate that CssDNA-mediated cell engineering favors stem-cell fitness and editing stability compared to traditional viral DNA repair templates (AAV). Our comprehensive dataset revealed critical transcriptomic and phenotypic differences regarding the efficacy of non-viral versus viral DNA donor template delivery methods.

Our findings highlight the potential of non-viral ssDNA delivery combined with TALEN® gene editing for gene insertion in long-term repopulating hematopoietic stem cells and primary T-cells. Implementing these advanced gene-editing techniques can pave the way for next-generation of therapies, aiming to provide alternative efficient, and safe therapeutic options for patients with cancer, autoimmune diseases, monogenic disorders, and other conditions.

1 Circular and linear ssDNA delivery

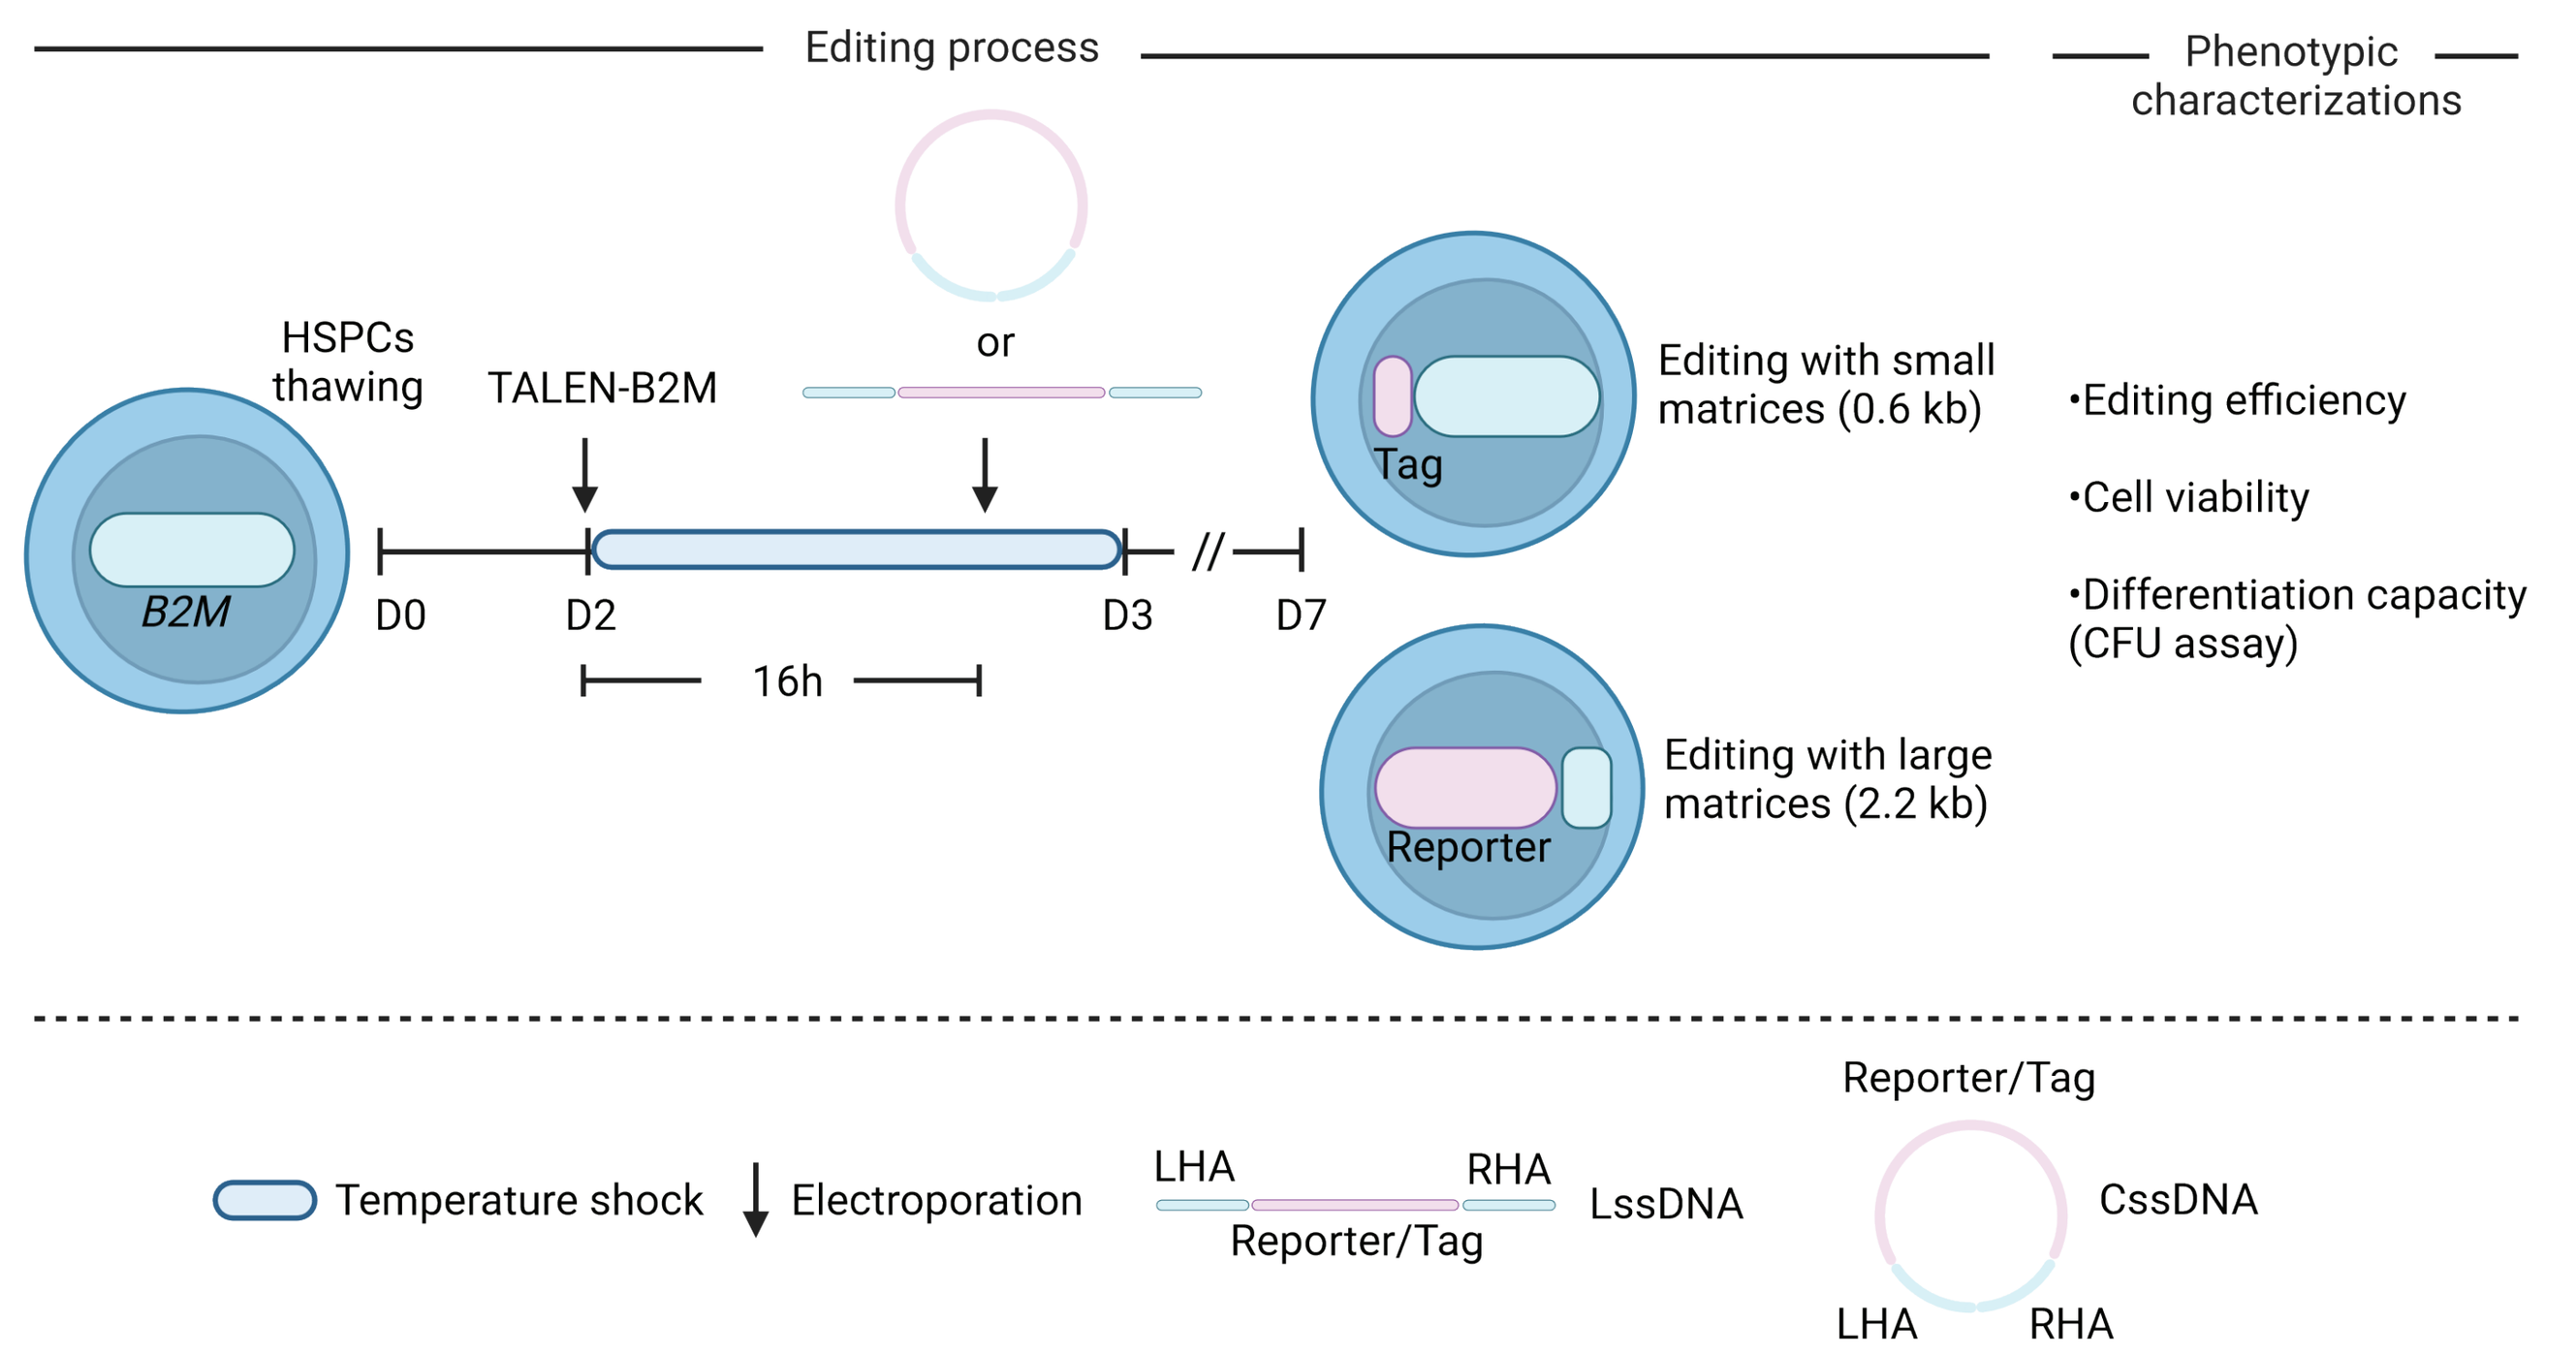
Editing process
Phenotypic characterizations
HSPCs thawing
TALEN-B2M
or
B2M
D0
D2
D3
D7
16h
Tag
Editing with small matrices (0.6 kb)
Reporter
Editing with large matrices (2.2 kb)
•Editing efficiency
•Cell viability
•Differentiation capacity (CFU assay)
Temperature shock
Electroporation
LHA
RHA
Reporter/Tag
LssDNA
CssDNA

Representative schema of TALEN-mediated editing protocol using an mRNA encoding TALEN targeting the B2M locus and lssDNA or cssDNA as DNA donor templates to insert a tag (0.6 kb) or a reported gene (2.2 kb) via non-disruptive and disruptive insertions, respectively. mRNAs encoding a viability enhancer and a HDR enhancer (Via-Enh01 and HDR-Enh01, respectively) were also incorporated in the process. The timing is indicated in days (D0-D7). Edited cells obtained 7 days post thawing were characterized by flow cytometry to assess the level of knock-in (KI) of DNA donor templates and knock-out (KO) of B2M as well as their viability. The differentiation capacity of HSPCs into erythroid and myeloid progenitors was also assessed by colony forming unit (CFU) assay.

2 ssDNA circularization increases TALEN-mediated KI in HSPCs

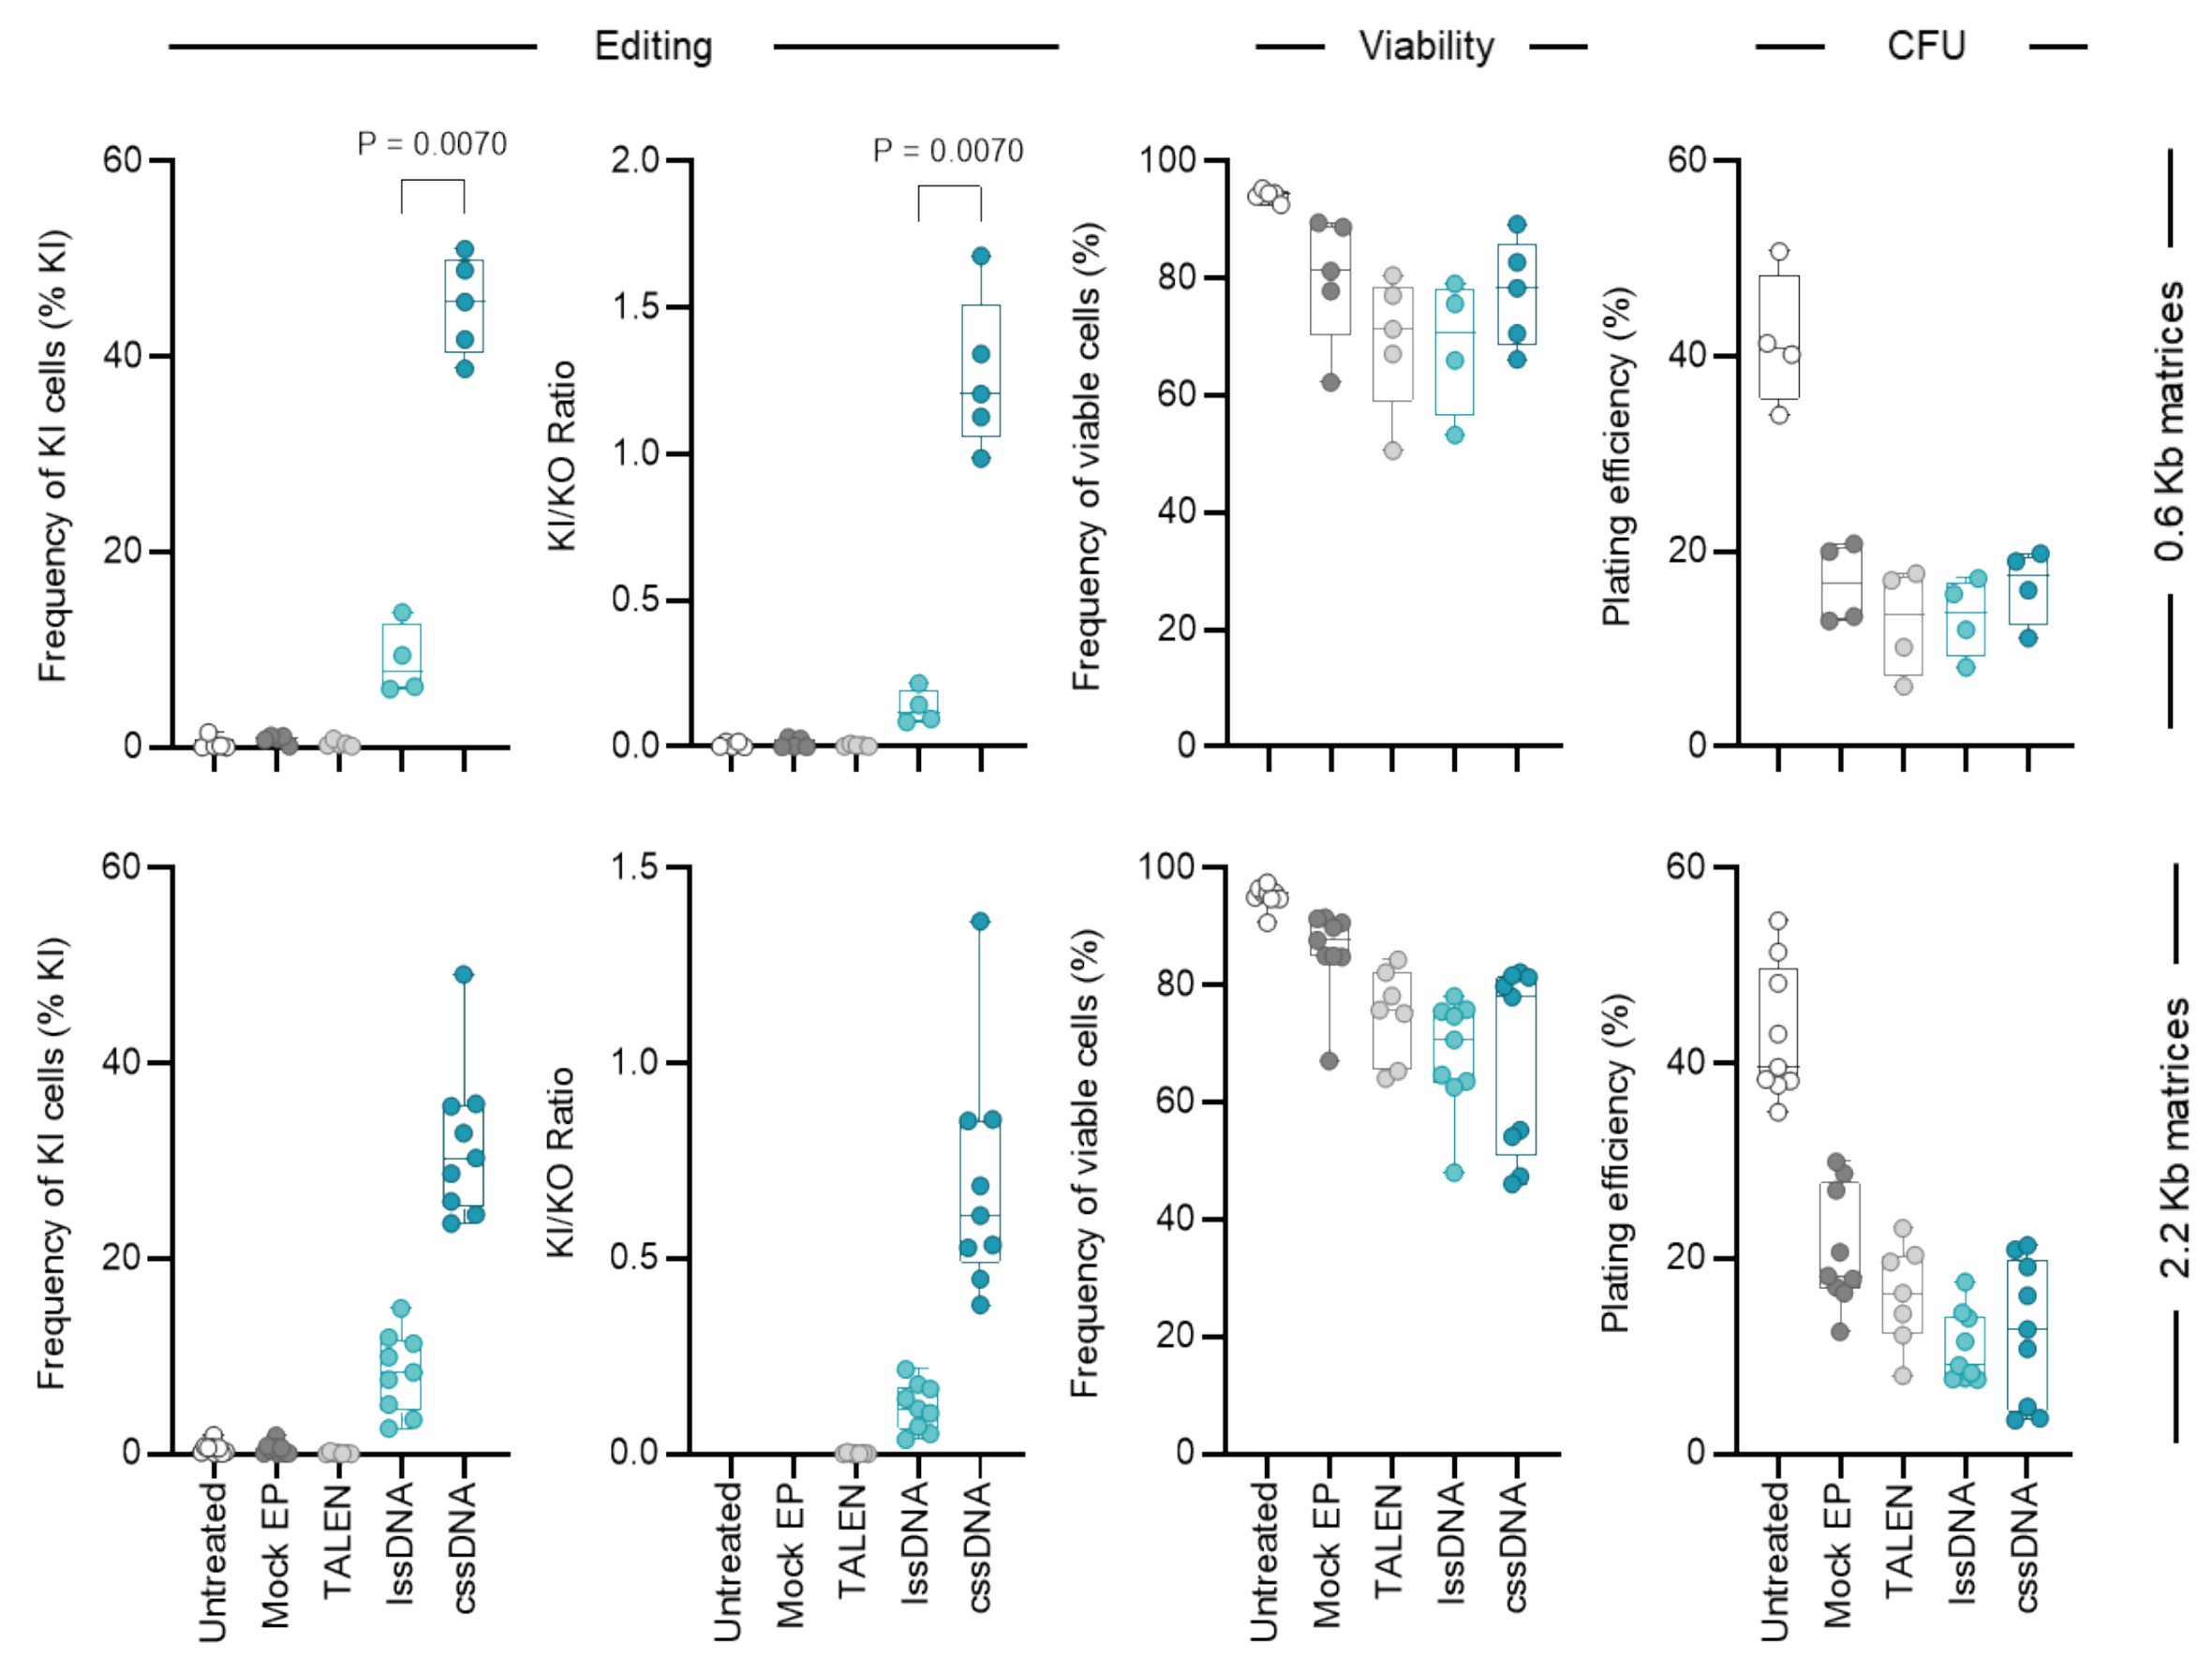
Editing
Viability
CFU
P = 0.0070
P = 0.0070
Frequency of KI cells (% KI)
KI/KO Ratio
Frequency of viable cells (%)
Plating efficiency (%)
0.6 Kb matrices
2.2 Kb matrices
Untreated
Mock EP
TALEN
lssDNA
cssDNA

Results showing the frequency of cells harboring KI events, the ratio KI/KO, the viability and plating efficiency of HSPCs either untreated, electroporated (Mock EP), edited with TALEN only (TALEN), or edited with TALEN and lssDNA or cssDNA donor templates (lssDNA or cssDNA, respectively) following the protocol from panel 1. Results with 0.6 kb donor templates are shown in the top panel and results with 2.2 kb DNA donor templates are shown in the bottom, On each box plot, the central mark indicates the median, the bottom and top edges of the box indicate the interquartile range (IQR), and the whiskers represent the maximum and minimum data point. Each dot represents data obtained from different HSPCs donors.

3 CssDNA-mediated KI leads to higher HSPCs engraftment than AAV

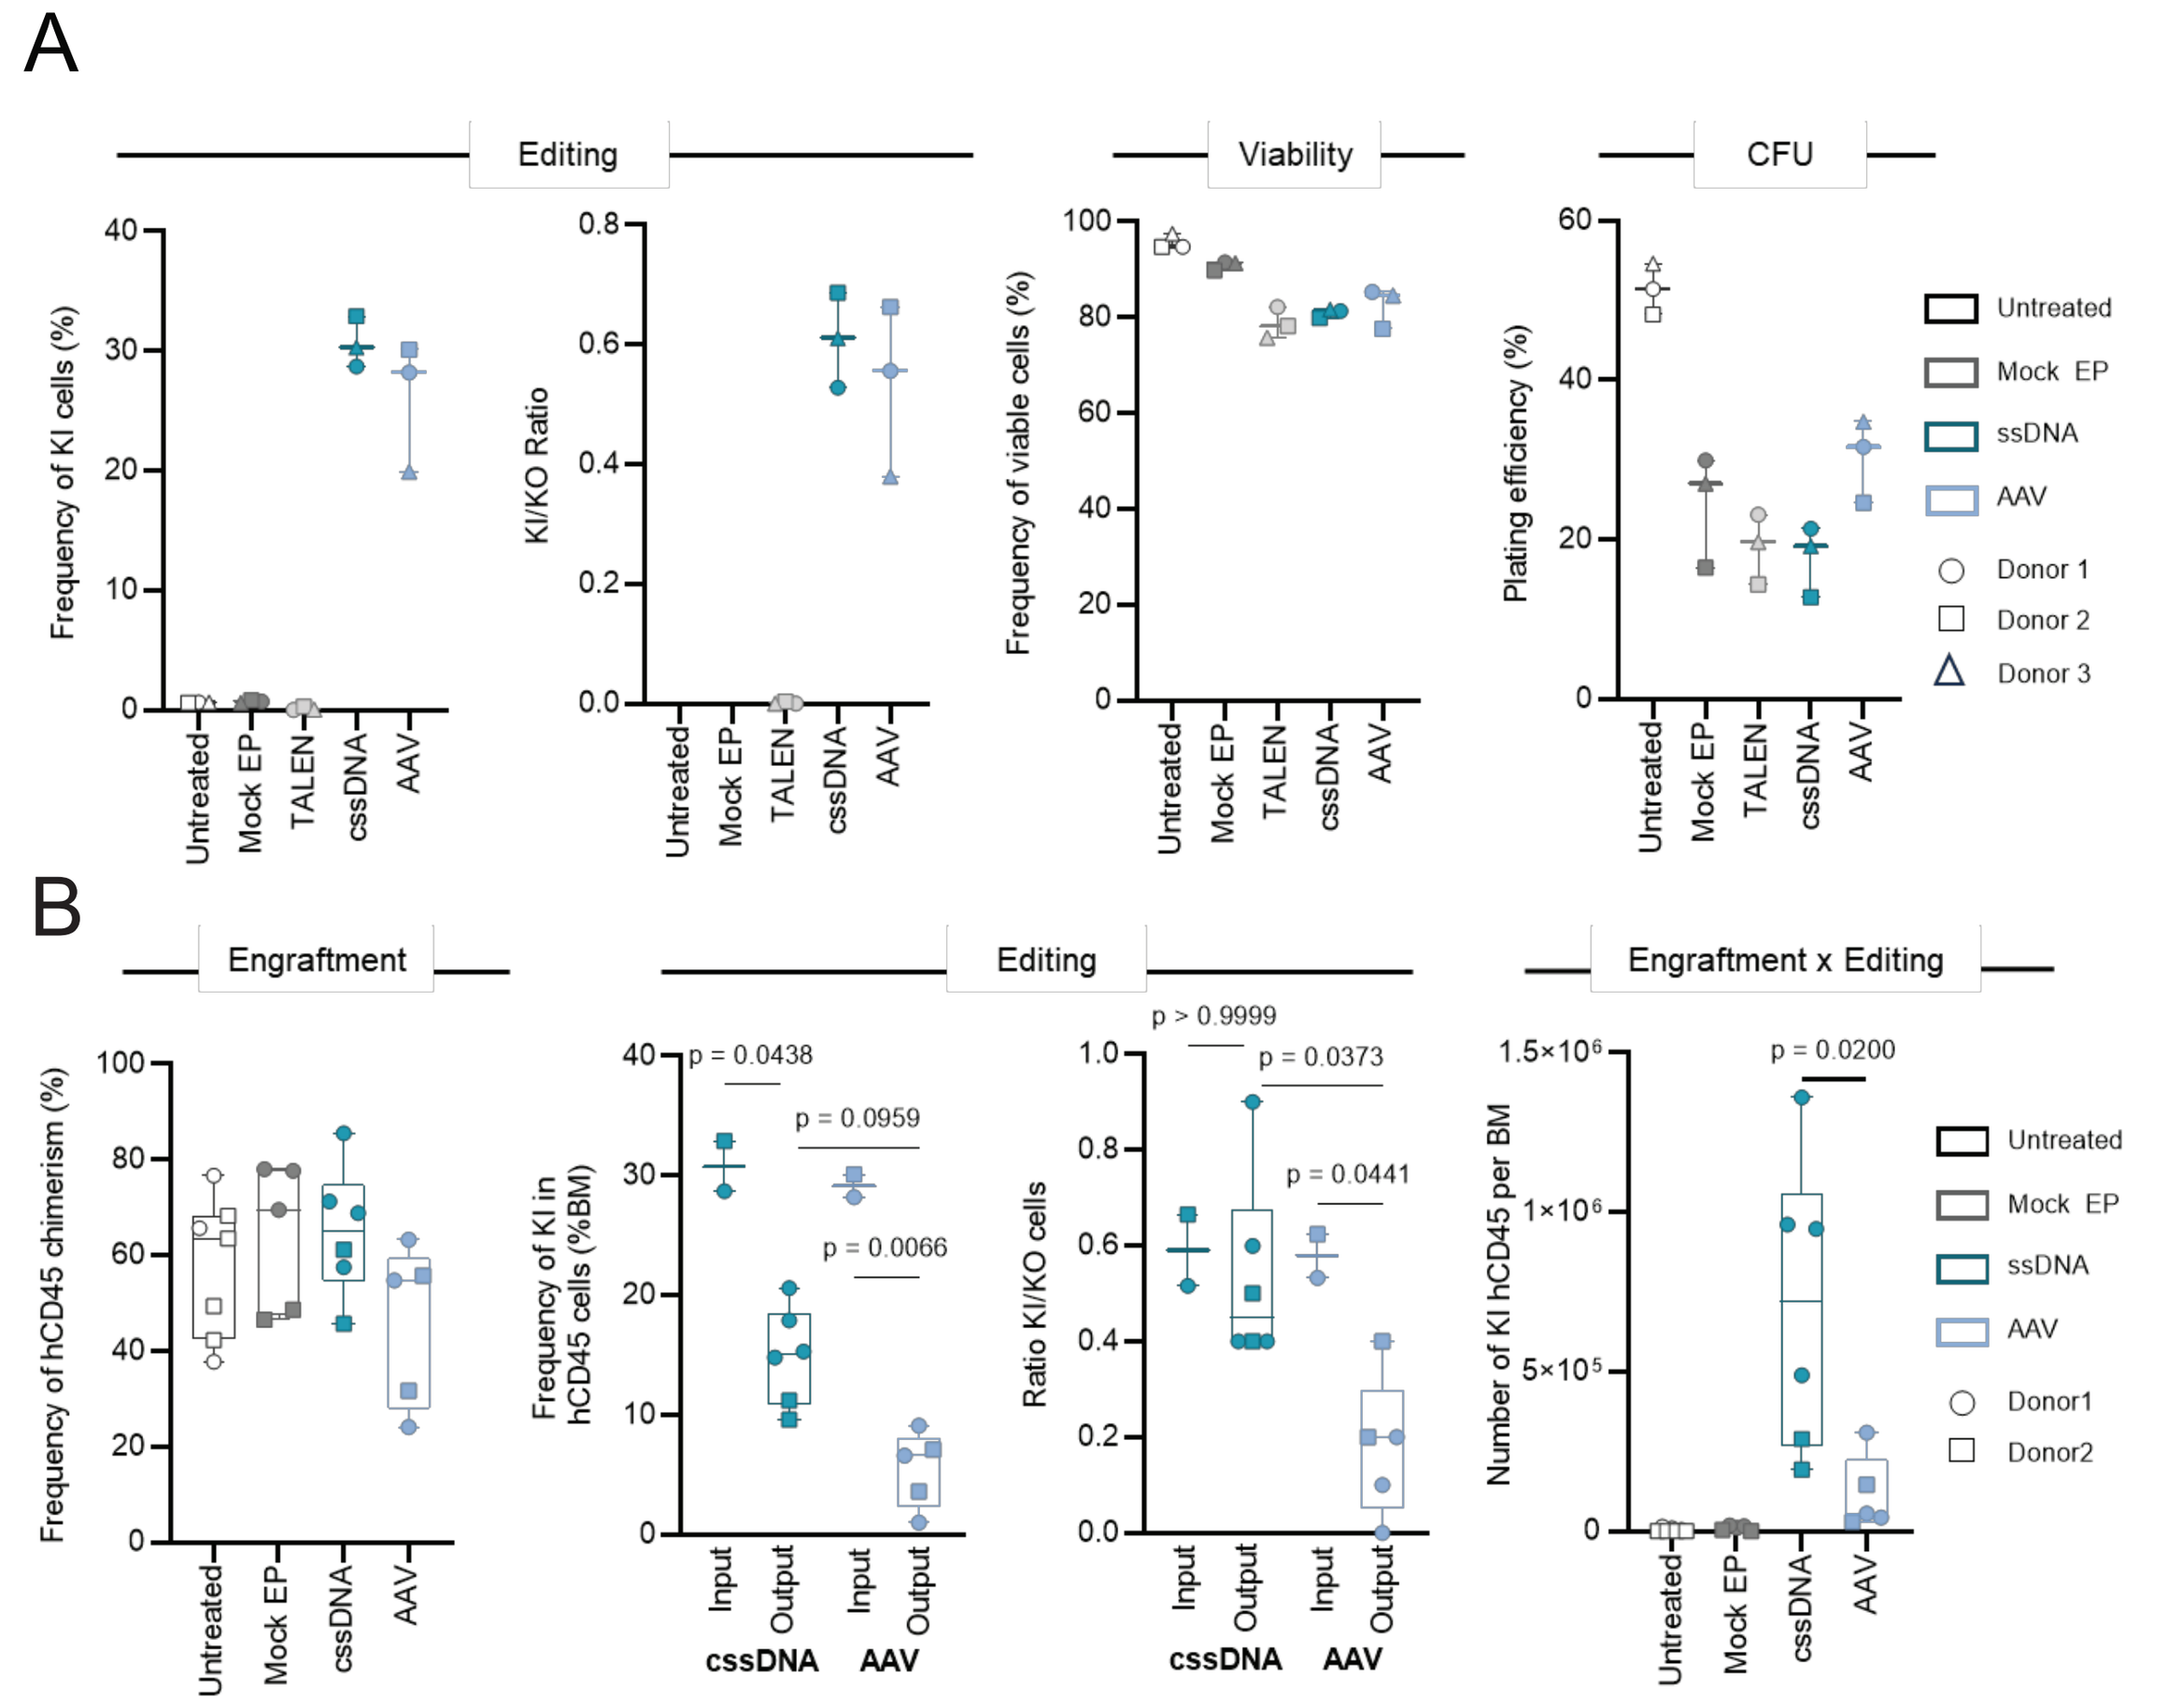
A
Editing
Viability
CFU
Frequency of KI cells (%)
KI/KO Ratio
Frequency of viable cells (%)
Plating efficiency (%)
Untreated
Mock EP
TALEN
cssDNA
AAV
Untreated
Mock EP
ssDNA
AAV
Donor 1
Donor 2
Donor 3
B
Engraftment
Editing
Engraftment x Editing
Frequency of hCD45 chimerism (%)
Frequency of KI in hCD45 cells (%BM)
Ratio KI/KO cells
Number of KI hCD45 per BM
p = 0.0438
p = 0.0959
p = 0.0066
p > 0.9999
p = 0.0373
p = 0.0441
p = 0.0200
Input
Output
cssDNA
AAV
Donor1
Donor2

Experimental schematics

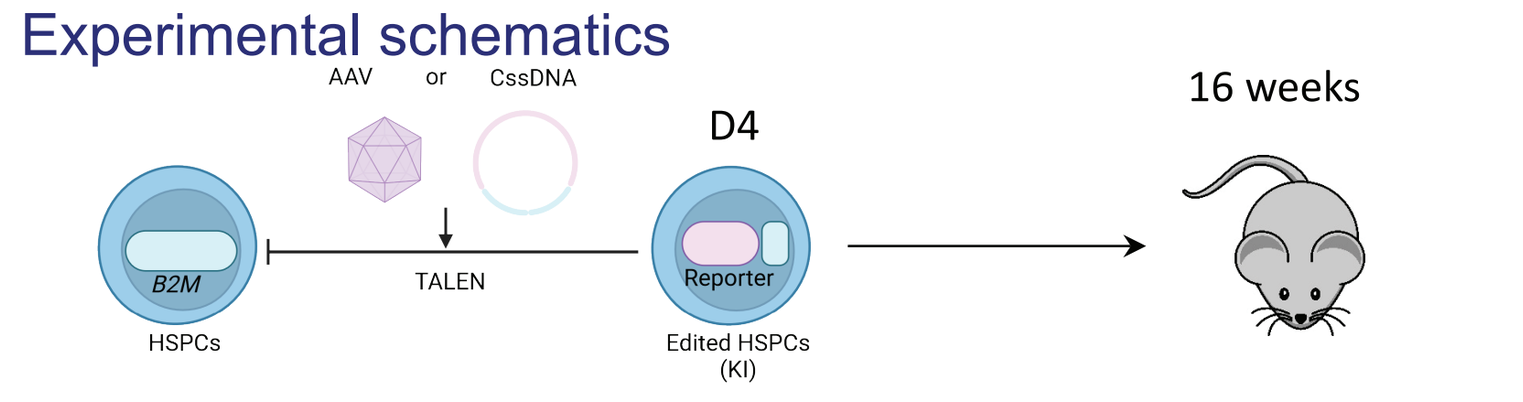
AAV
or
CssDNA
D4
16 weeks
B2M
HSPCs
TALEN
Reporter
Edited HSPCs (KI)

Edited HSPCs with a TALEN targeting the B2M locus and cssDNA or AAV (MOI=350 vg/cell) were retrieved 7 days post thawing and characterized by flow cytometry to assess transgene-KI and B2M KO levels. Differentiation capacity into erythroid and myeloid progenitors as well as their transcriptomics profile were also assessed with CFU assays and CITE-seq (panel 4), respectively. For *in vivo* experiments, edited HSPCs 4 days post thawing, were injected in NCG mice to assess their ability to engraft, differentiate and preserve editing efficiency. **A**. *In vitro* results showing the frequency of cells harboring KI events, KI/KO ratio, viability after editing and plating efficiency in the groups indicated. **B**. *In vivo* results showing hCD45+ cells engraftment and of KI and KO frequencies determined either before mice injection (input), or in hCD45+ cells engrafted in the bone marrow (BM) 16 weeks after cells injection (output). Two-way ANOVA followed by Bonferroni multi-comparison test. P-values are indicated. The product of the frequency of hCD45+ cells engraftment and frequency of KI is also shown to illustrate the overall efficiency of each protocol of HSPC editing. Mann–Whitney two-tailed non-parametric unpaired test with a confidence interval of 95%. P-value is indicated. On each box plot, the central mark indicates the median, the bottom and top edges of the box indicate the interquartile range (IQR), and the whiskers represent the maximum and minimum data point. Each dot represents data obtained from one HSPCs donor.

4 CssDNA-mediated KI yields higher number of edited of HSCs than AAV

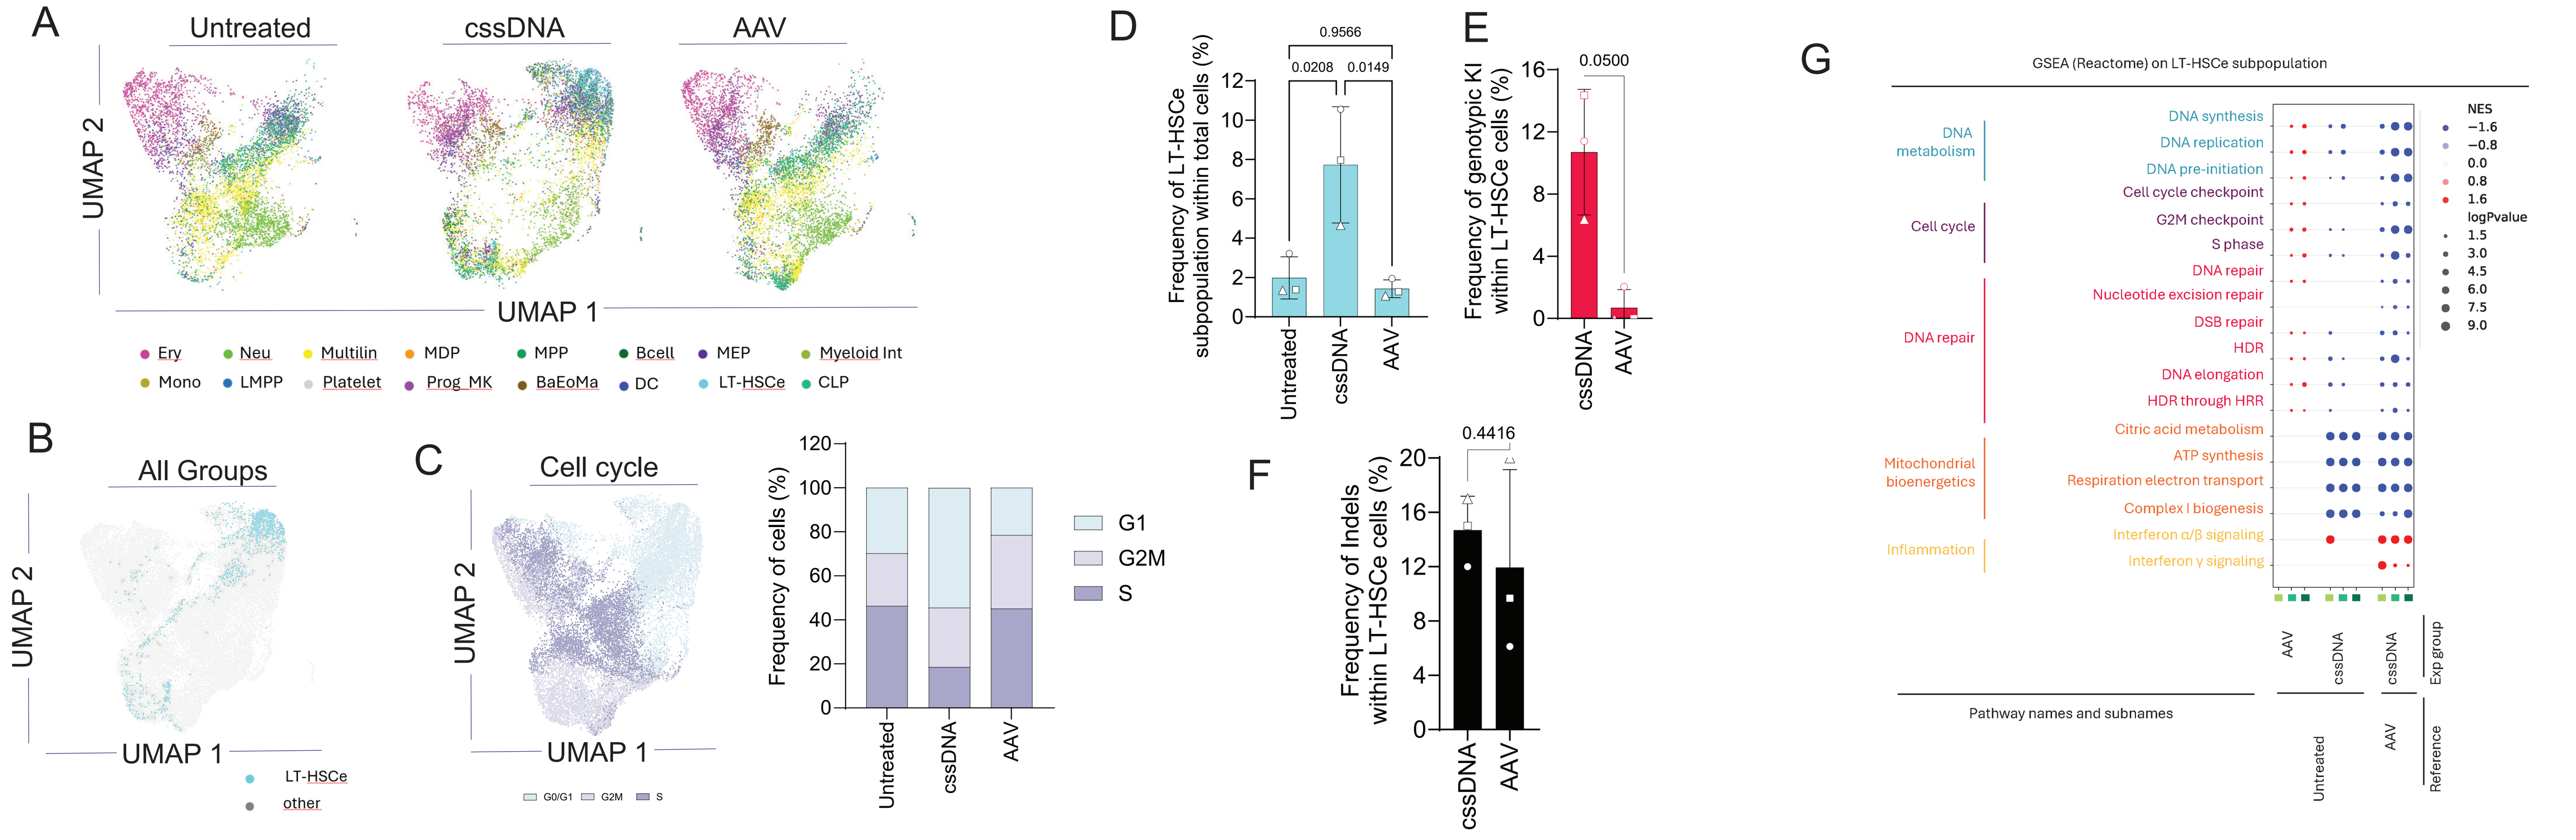
A
Untreated
cssDNA
AAV
UMAP 2
UMAP 1
Ery
Neu
Multilin
MDP
MPP
Bcell
MEP
Myeloid Int
Mono
LMPP
Platelet
Prog_MK
BaEoMa
DC
LT-HSCe
CLP
B
All Groups
LT-HSCe
other
C
Cell cycle
G0/G1
G2M
S
Frequency of cells (%)
G1
G2M
S
D
Frequency of LT-HSCe subpopulation within total cells (%)
0.9566
0.0208
0.0149
E
Frequency of genotypic KI within LT-HSCe cells (%)
0.0500
F
Frequency of Indels within LT-HSCe cells (%)
0.4416
G
GSEA (Reactome) on LT-HSCe subpopulation
DNA metabolism
Cell cycle
DNA repair
Mitochondrial bioenergetics
Inflammation
DNA synthesis
DNA replication
DNA pre-initiation
Cell cycle checkpoint
G2M checkpoint
S phase
DNA repair
Nucleotide excision repair
DSB repair
HDR
DNA elongation
HDR through HRR
Citric acid metabolism
ATP synthesis
Respiration electron transport
Complex I biogenesis
Interferon α/β signaling
Interferon γ signaling
NES
logPvalue
Pathway names and subnames
Exp group
Reference
Untreated

A. UMAP plots showing aggregated 5'scRNA CITE-Seq data obtained from untreated HSPCs either, edited with TALEN and AAV or cssDNA donor templates, 4 days post thawing and at the time of NCG mice transfer (n = 3 independent biological donors). **B.** UMAP plots aggregated from from all experimental groups showing the position of Long-term HSCs-enriched subpopulation (LTHSCe). **C**. UMAP plot showing the cell cycle phases (G0/G1, G2M and S) in all groups together and graph showing the distribution in all groups. **D**. LT-HSCe frequency within all subpopulations. Two-way ANOVA with Bonferroni post-tests. **E**. Frequency of KI and KO within the LT-HSCe subpopulation. Paired t-test.. Each dot represents data obtained from one HSPCs donor. **F**. Frequency of KO in LT-HSCs . **G**. Gene Set Enrichment Analysis (GSEA) obtained in LT-HSCe to compare the AAV and CssDNA experimental groups to the untreated reference group and to directly compare the cssDNA group to the AAV reference group. Normalized Enrichment Score (NES) as well as Log P value obtained for each donor (n=3 independent HSPC donors) are illustrated by a red/white/blue color code and size of the dots, respectively. Red: upregulated (red), Blue: downregulated (blue).

5 CssDNA eficiently edits T-cells

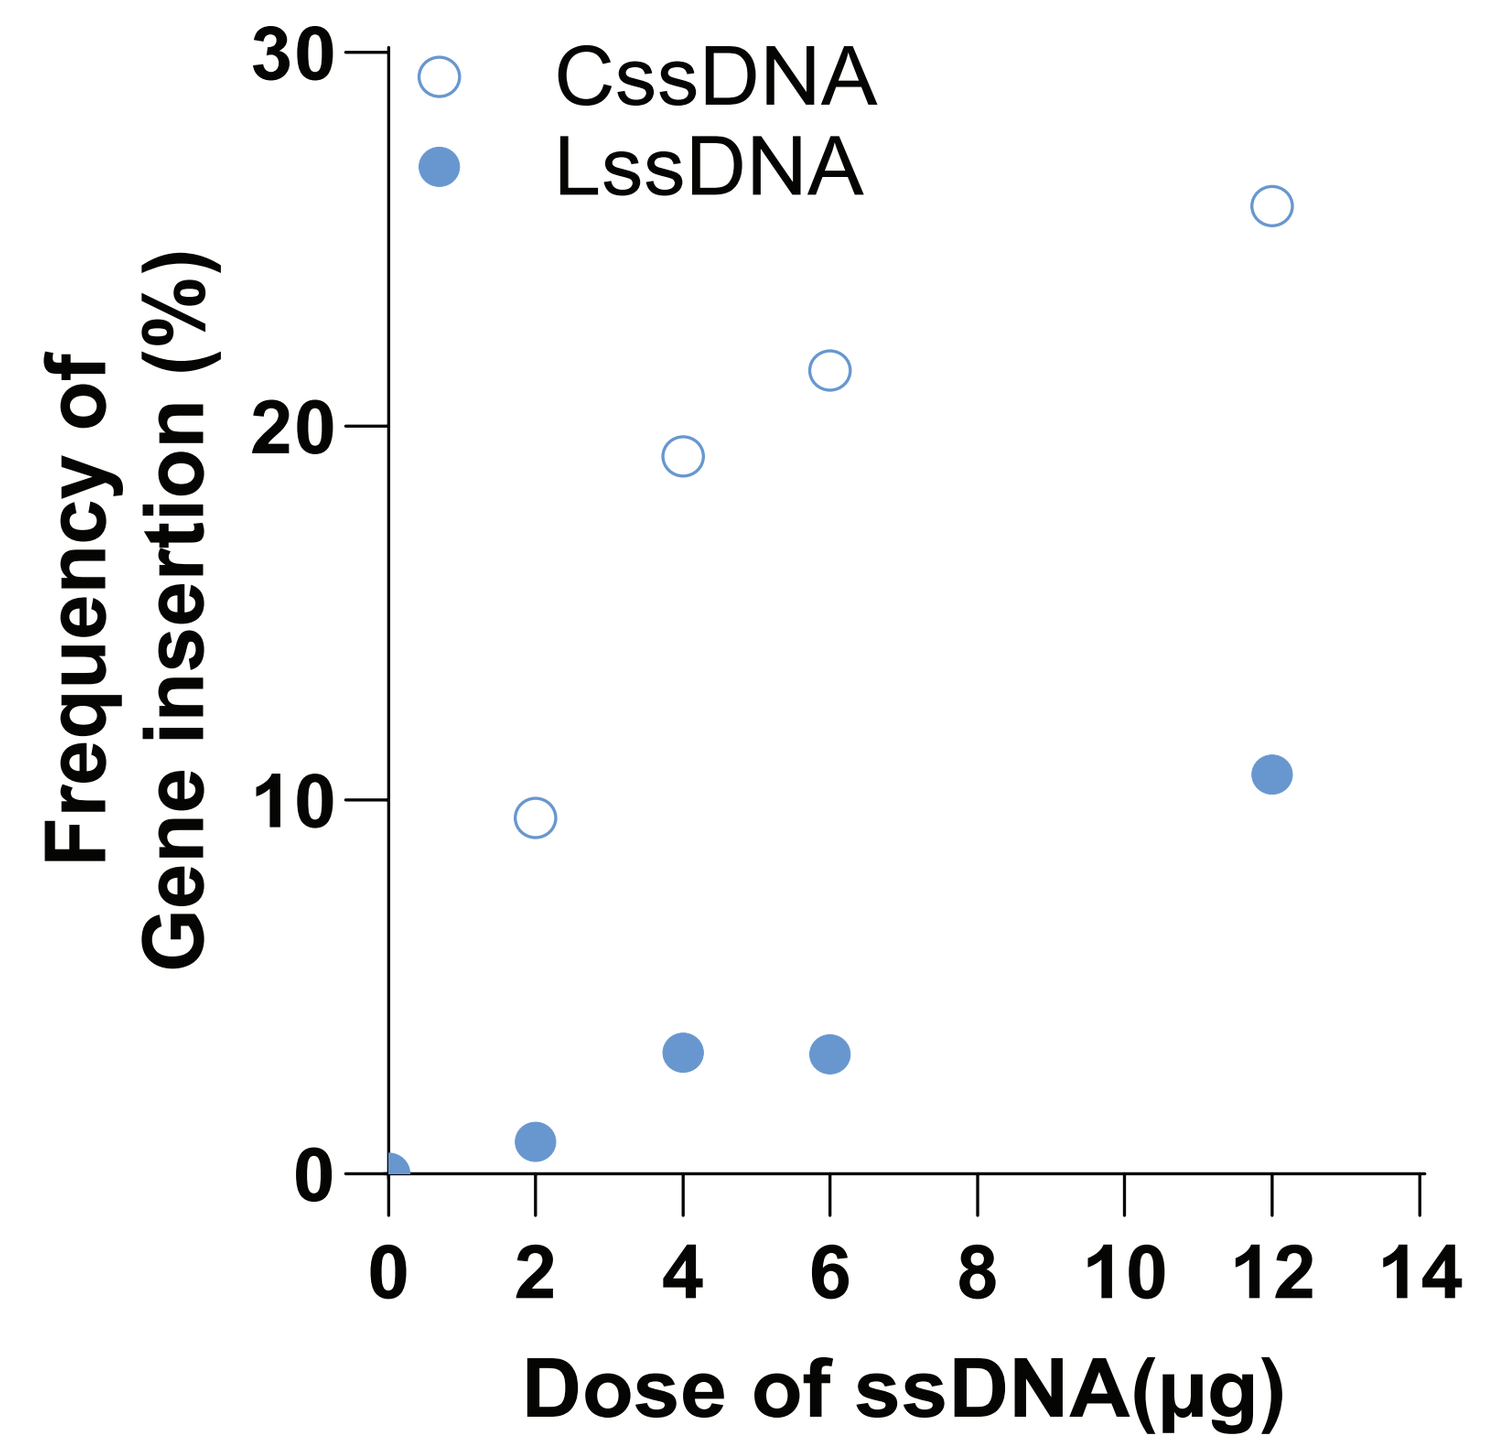
CssDNA
LssDNA
Frequency of Gene insertion (%)
Dose of ssDNA(µg)

T-cells were editied as shown in panel 1 with B2M KO and integration of 2.2Kb donor template comparing circular and linear ssDNA. Efficiencies at day 7 after gene editing are shown

6 Conclusions

Linear single-stranded DNA (lssDNA) or circular single-stranded DNA (cssDNA) can serve as donor templates for targeted gene insertion at the B2M locus in HSPCs and T-cells using TALEN technology.

Circular ssDNA yields a 3-5 fold higher gene insertion frequency compared to linear ssDNA, achieving up to 49% precise targeted gene insertion events in HSPCs.

cssDNA/TALEN-mediated HSPCs editing show efficient engraftment in the bone marrow of NCG mice, retaining 50% of the gene insertion events detected prior to injection. This technique generates HSPCs with superior *in vivo* engraftment capacity and editing stability compared to those edited via AAV/TALEN.

cssDNA-mediated transgene delivery coupled with TALEN editing promotes higher levels of correctly edited HSC-enriched subpopulation than AAV/TALEN-mediated editing process.

Some images were generated with Biorender.com

This communication expressly or implicitly contains certain forward-looking statements concerning Cellectis and its business. Cellectis is providing this communication as of this date and does not undertake to update any forward-looking statements contained herein as a result of new information, future events or otherwise. This communication contains Cellectis' proprietary information. TALEN® and Cellectis® are trademarks owned by the Cellectis Group.